Effectiveness and tolerability of avapritinib in indolent systemic mastocytosis : first results from the French nationwide real-world AIMS study

Clément Gourguechon, Cristina Livideanu, Maël Heiblig, Edwige Le Mouel, Stéphane Barete, Yannick Chantran, Ludovic Lhermitte, Orianne Debeaupuis, Hassiba Bouktit, Olivier Hermine, Julien Rossignol and CEREMAST investigators

Systemic Mastocytosis

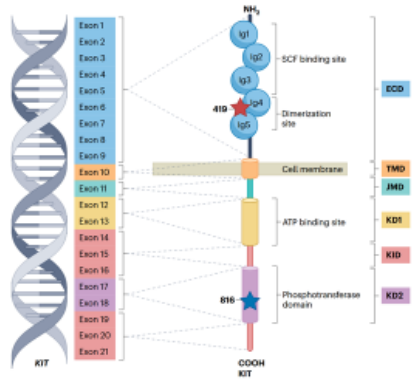

NH2
SCF binding site
Dimerization site
419
Cell membrane
ATP binding site
Phosphotransferase domain
816
COOH
KIT
ECD
TMD
JMD
KD1
KID
KD2

- Heterogenous rare disease (prevalence 1/3500-4000, incidence 1,21-1,56/100 000h/yrs*)
- Myeloproliferative disorder subcategory (WHO 2022)
- **Abnormal tissue accumulation** of clonal mast cell
 - Immunophenotypic (CD25, CD2, CD30)
 - Morphology : spindle shape, hypogranular cytoplasm, atypic nuclei
 - Molecular : **c-KIT D816V mutation**
- **Clinical Heterogeneity**
- **90 % are non advanced form**
 - **Indolent Systemic Mastocytosis (ISM)** is the most prevalent subtype
 - **Anaphylaxis** and **severe osteoporosis** can be inaugural
 - **Altered QoL** in most of patients related to **mast cell activation symptoms**
- **Avapritinib**
 - **First selective KIT inhibitor approved for ISM** with moderate to severe symptoms despiste best supportive cares
 - **Demonstrate clinical efficacy** on symptoms and biomarkers with good tolerability

. *Estimation. Jorgensen Eur J Epidemiol 2025 Bergström Acta Oncologica 2024 Schuler JACI 2021. Kibsgaard Int J Women Dermatol 2020. Cohen Bjh 2014. Van Doormaal JACI 2013 ; Khouri ;

Akin et al, Nat Rev Dis Prim. 2025
Gotlib et al, NEJM Evidence. 2023

AIMS study : a real world experience of avapritinib use in ISM

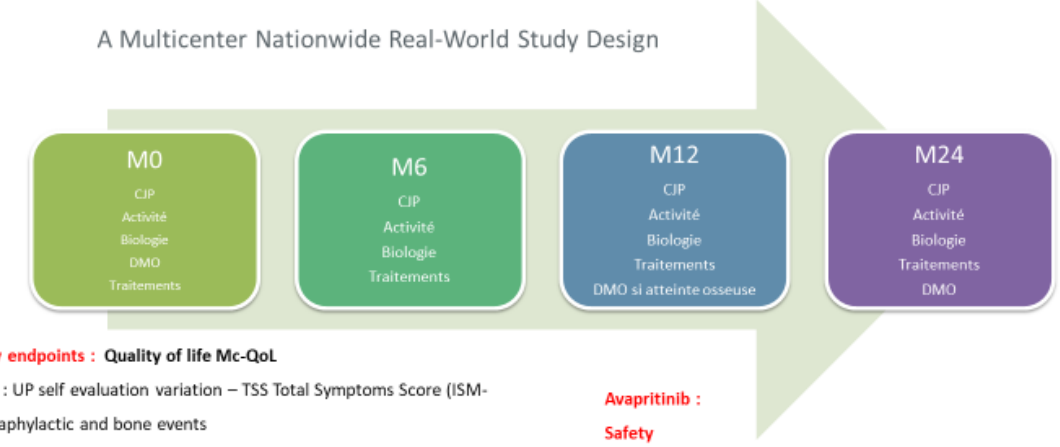

A Multicenter Nationwide Real-World Study Design
M0
CJP
Activité
Biologie
DMO
Traitements
M6
CJP
Activité
Biologie
Traitements
M12
CJP
Activité
Biologie
Traitements
DMO si atteinte osseuse
M24
CJP
Activité
Biologie
Traitements
DMO

Primary endpoints : Quality of life Mc-QoL

Activity : UP self evaluation variation – TSS Total Symptoms Score (ISM-SAF) anaphylactic and bone events

Biology : Tryptase –KIT VAF (Variant allelic Frequency) - CTX (Beta Crosslaps)

DXA : bone mineral density assessment

Traitements : Mediators cibling and symptomatics treatments

Avapritinib :

Safety

Discontinuation, dose reduction or increase

AIMS study : a French nationwide multicentric study

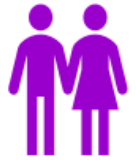

n= 162

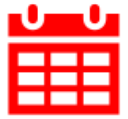

April 2024 to April 2026

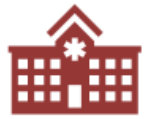

CEREMAST NETWORK = 21 centers

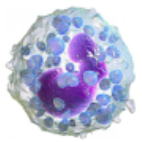

Indolent systemic mastocytosis according WHO 2016

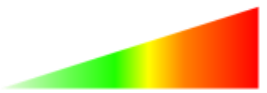

Debilating symptoms despite best supportive symptomatic cares

AIMS study : a real life population

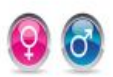

Women : 63,6 %

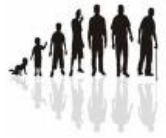

Median age : 54,4 years

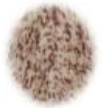

Skin involvement : 89 %

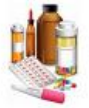

Previous Omalizumab treatment = 1/4

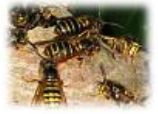

Anaphylaxis : 19,8 %

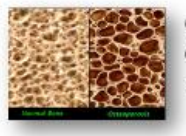

Osteopenia : 26,5 %
Osteoporosis : 22 %
Bone Fracture : 22 %

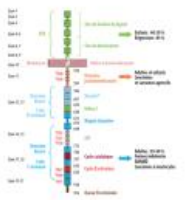

Mutation KITD816V : 97 %
VAF : 0,89 %

Tryptase : 47,8 µg/l

AIMS study : first 6 month results

MC-QoL n=90

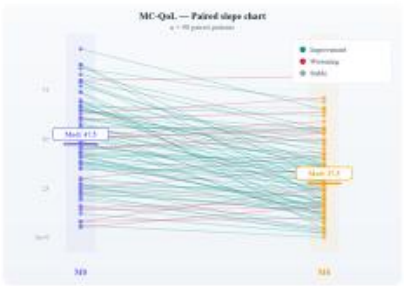

MC-QoL — Paired slope chart
M0
M6

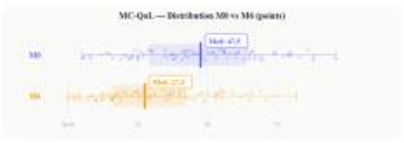

MC-QoL — Distribution M0 vs M6 (points)
M0
M6

ISM-SAF n=78

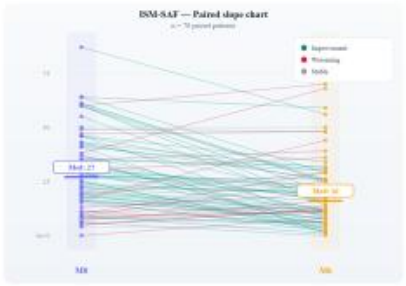

ISM-SAF — Paired slope chart
M0
M6

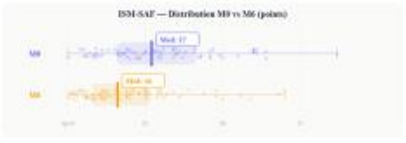

ISM-SAF — Distribution M0 vs M6 (points)
M0
M6

Tryptase n= 70

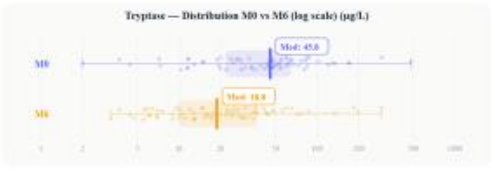

Tryptase — Distribution M0 vs M6 (log scale) (µg/L)
M0
M6

VAF KIT n= 45

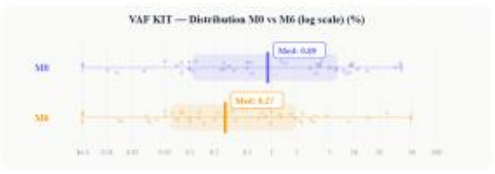

VAF KIT — Distribution M0 vs M6 (log scale) (%)
M0
M6

Symptomatic treatment at 6 month n= 90

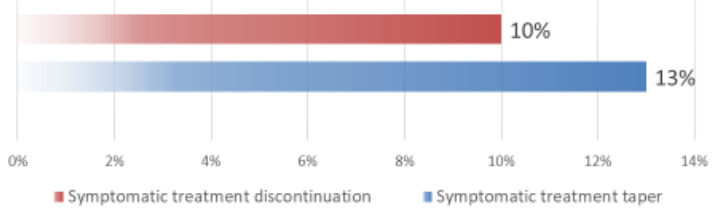

10%
13%
0%
2%
4%
6%
8%
10%
12%
14%
Symptomatic treatment discontinuation
Symptomatic treatment taper

23 % discontinued or tapered at least one symptomatic treatment at M6

Avapritinib Safety (n = 162)

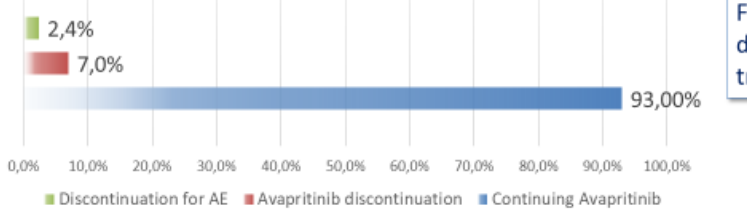

2,4%
7,0%
93,00%
0,0%
10,0%
20,0%
30,0%
40,0%
50,0%
60,0%
70,0%
80,0%
90,0%
100,0%
Discontinuation for AE
Avapritinib discontinuation
Continuing Avapritinib

Discontinuation n= 12 (2,6 month)
For no severe AE (n=4) : oedema, diffuse pain, asthenia, digestive troubles