A Systematic Literature Review of Avapritinib Outcomes in Patients with Indolent Systemic Mastocytosis

Poster # 529

Scan the QR code to view our **Interactive Poster**

Robyn Scherber,[1] Teresa Green,[1] Priyanka Bobbili,[2] Deepshekhar Gupta,[3] Sydney Ng,[4] Rachel Meade,[5] Olivia Reese,[3] Mei Sheng Duh,[2] Dakota Powell,[1] Erin Sullivan[1]

[1] Blueprint Medicines Corporation, Cambridge, Massachusetts, United States; [2] Analysis Group, Inc., Boston, Massachusetts, United States; [3] Analysis Group, Inc., Menlo Park, California, United States; [4] Analysis Group, Inc., Los Angeles, California, United States; [5] Analysis Group, Inc., San Francisco, California, United States

Rationale

- Indolent systemic mastocytosis (ISM) is a clonal mast cell disease characterized by abnormal proliferation and accumulation of mast cells, most commonly driven by the *KIT* D816V mutation.[1]
- As the majority (~95%) of patients with systemic mastocytosis carry a *KIT* D816V mutation, recent therapeutic advances have focused on *KIT* inhibitors.[2-4]
- Avapritinib is a selective inhibitor of D816V-mutated *KIT* approved for patients with ISM in the United States (US) and Europe based on findings from PIONEER (Phase 2; NCT03580655), a randomized, double-blind, placebo-controlled trial.
- This marks an opportunity to assess and review the increasing body of published evidence reporting the efficacy and safety of avapritinib for the treatment of ISM.

Objective

- This systematic literature review (SLR) was conducted to synthesize available published evidence documenting efficacy, safety, and quality of life (QoL) outcomes in patients with ISM treated with avapritinib.

Methods

Data Sources and Search Strategy

- An electronic search of article titles and abstracts was performed using pre-defined search terms, including Medical Subject Heading (MeSH) terms, to ensure a comprehensive capture of all relevant studies related to ISM.
- Databases, including MEDLINE, MEDLINE In-Progress, Embase, Cochrane Central Register of Controlled Trials, Cochrane Database of Systematic Reviews, and Northern Light Discovery Conference Database, were searched via OvidSP through April 23, 2025.
- Supplementary hand searches were conducted to capture studies published between 2022 and June 30, 2025, including key conference abstracts.

Study Selection and Data Extraction

- All studies identified via the electronic database and supplementary hand searches were screened for relevance to the study objectives based on pre-specified PICOS elements (**Table 1**).
- Screening and study selection at Level 1 (title and abstract) and Level 2 (full text) were conducted independently by two researchers, with adjudication of discrepancies by a third researcher.
- Relevant data points from included studies were independently extracted by two researchers, with any discrepancies adjudicated by a third researcher.

Table 1. PICOS Elements

Element	Inclusion Criteria
Population	Adult patients (≥18 years of age) with ISM
Intervention	Avapritinib
Comparator	No restrictions on comparator
Outcomes	Efficacy outcomes, including: reduction in serum tryptase and *KIT* MAF/VAF, DOT, PROs related to symptom reduction or QoL, polypharmacy reduction, and outcomes related to bone health or anaphylaxis Safety outcomes assessed via: TEAE/TRAEs, SAEs, and treatment discontinuation or dose modification due to AEs
Study type	All study types excluding letters, editorials, meta-analyses, case reports, systematic and targeted literature reviews, narrative reviews, and guidelines
Other	• Human studies • English only • Full-text articles published • Conference proceedings and abstracts published

Abbreviations: AE: adverse event; DOT: duration of treatment; ISM: indolent systemic mastocytosis; *KIT*: *KIT* gene; MAF: mutant allele fraction; PICOS: population intervention comparator outcomes study type; PRO: patient-reported outcome; QoL: quality of life; SAE: serious adverse event; TEAE: treatment-emergent adverse event; TRAE: treatment-related adverse event; VAF: variant allele fraction.

Results

Overview of Included Studies

- The search identified 79 studies (71 through electronic searches and 8 through hand searches).
- 17 studies met PICOS eligibility criteria after Level 1 and 2 screening and were included for data extraction (**Figure 1**).[1,5-20]
 - 16 studies reported outcomes assessed in PIONEER[1,5-19]
 - The PIONEER trial has 3 parts.[1] In Part 1, 25 mg once daily (QD) was identified as the recommended Phase 2 dose (RP2D). In Part 2, the efficacy and safety of avapritinib were evaluated in patients with ISM. Part 3 is an ongoing open-label extension of the trial evaluating safety and efficacy of avapritinib for up to 5 years.
 - 5 retrospective analyses of PIONEER were captured, including analyses of bone health outcomes,[11,18] avapritinib responses in patients with *KIT* mutations that required enhanced sensitivity for identification,[17,19] and a study of the safety and efficacy of avapritinib in patients receiving concomitant omalizumab.[6]
 - 1 study (AVATAR) assessed the impact of avapritinib on disease control and QoL for patients with ISM in a real-world setting.[20]

Figure 1. PRISMA Diagram

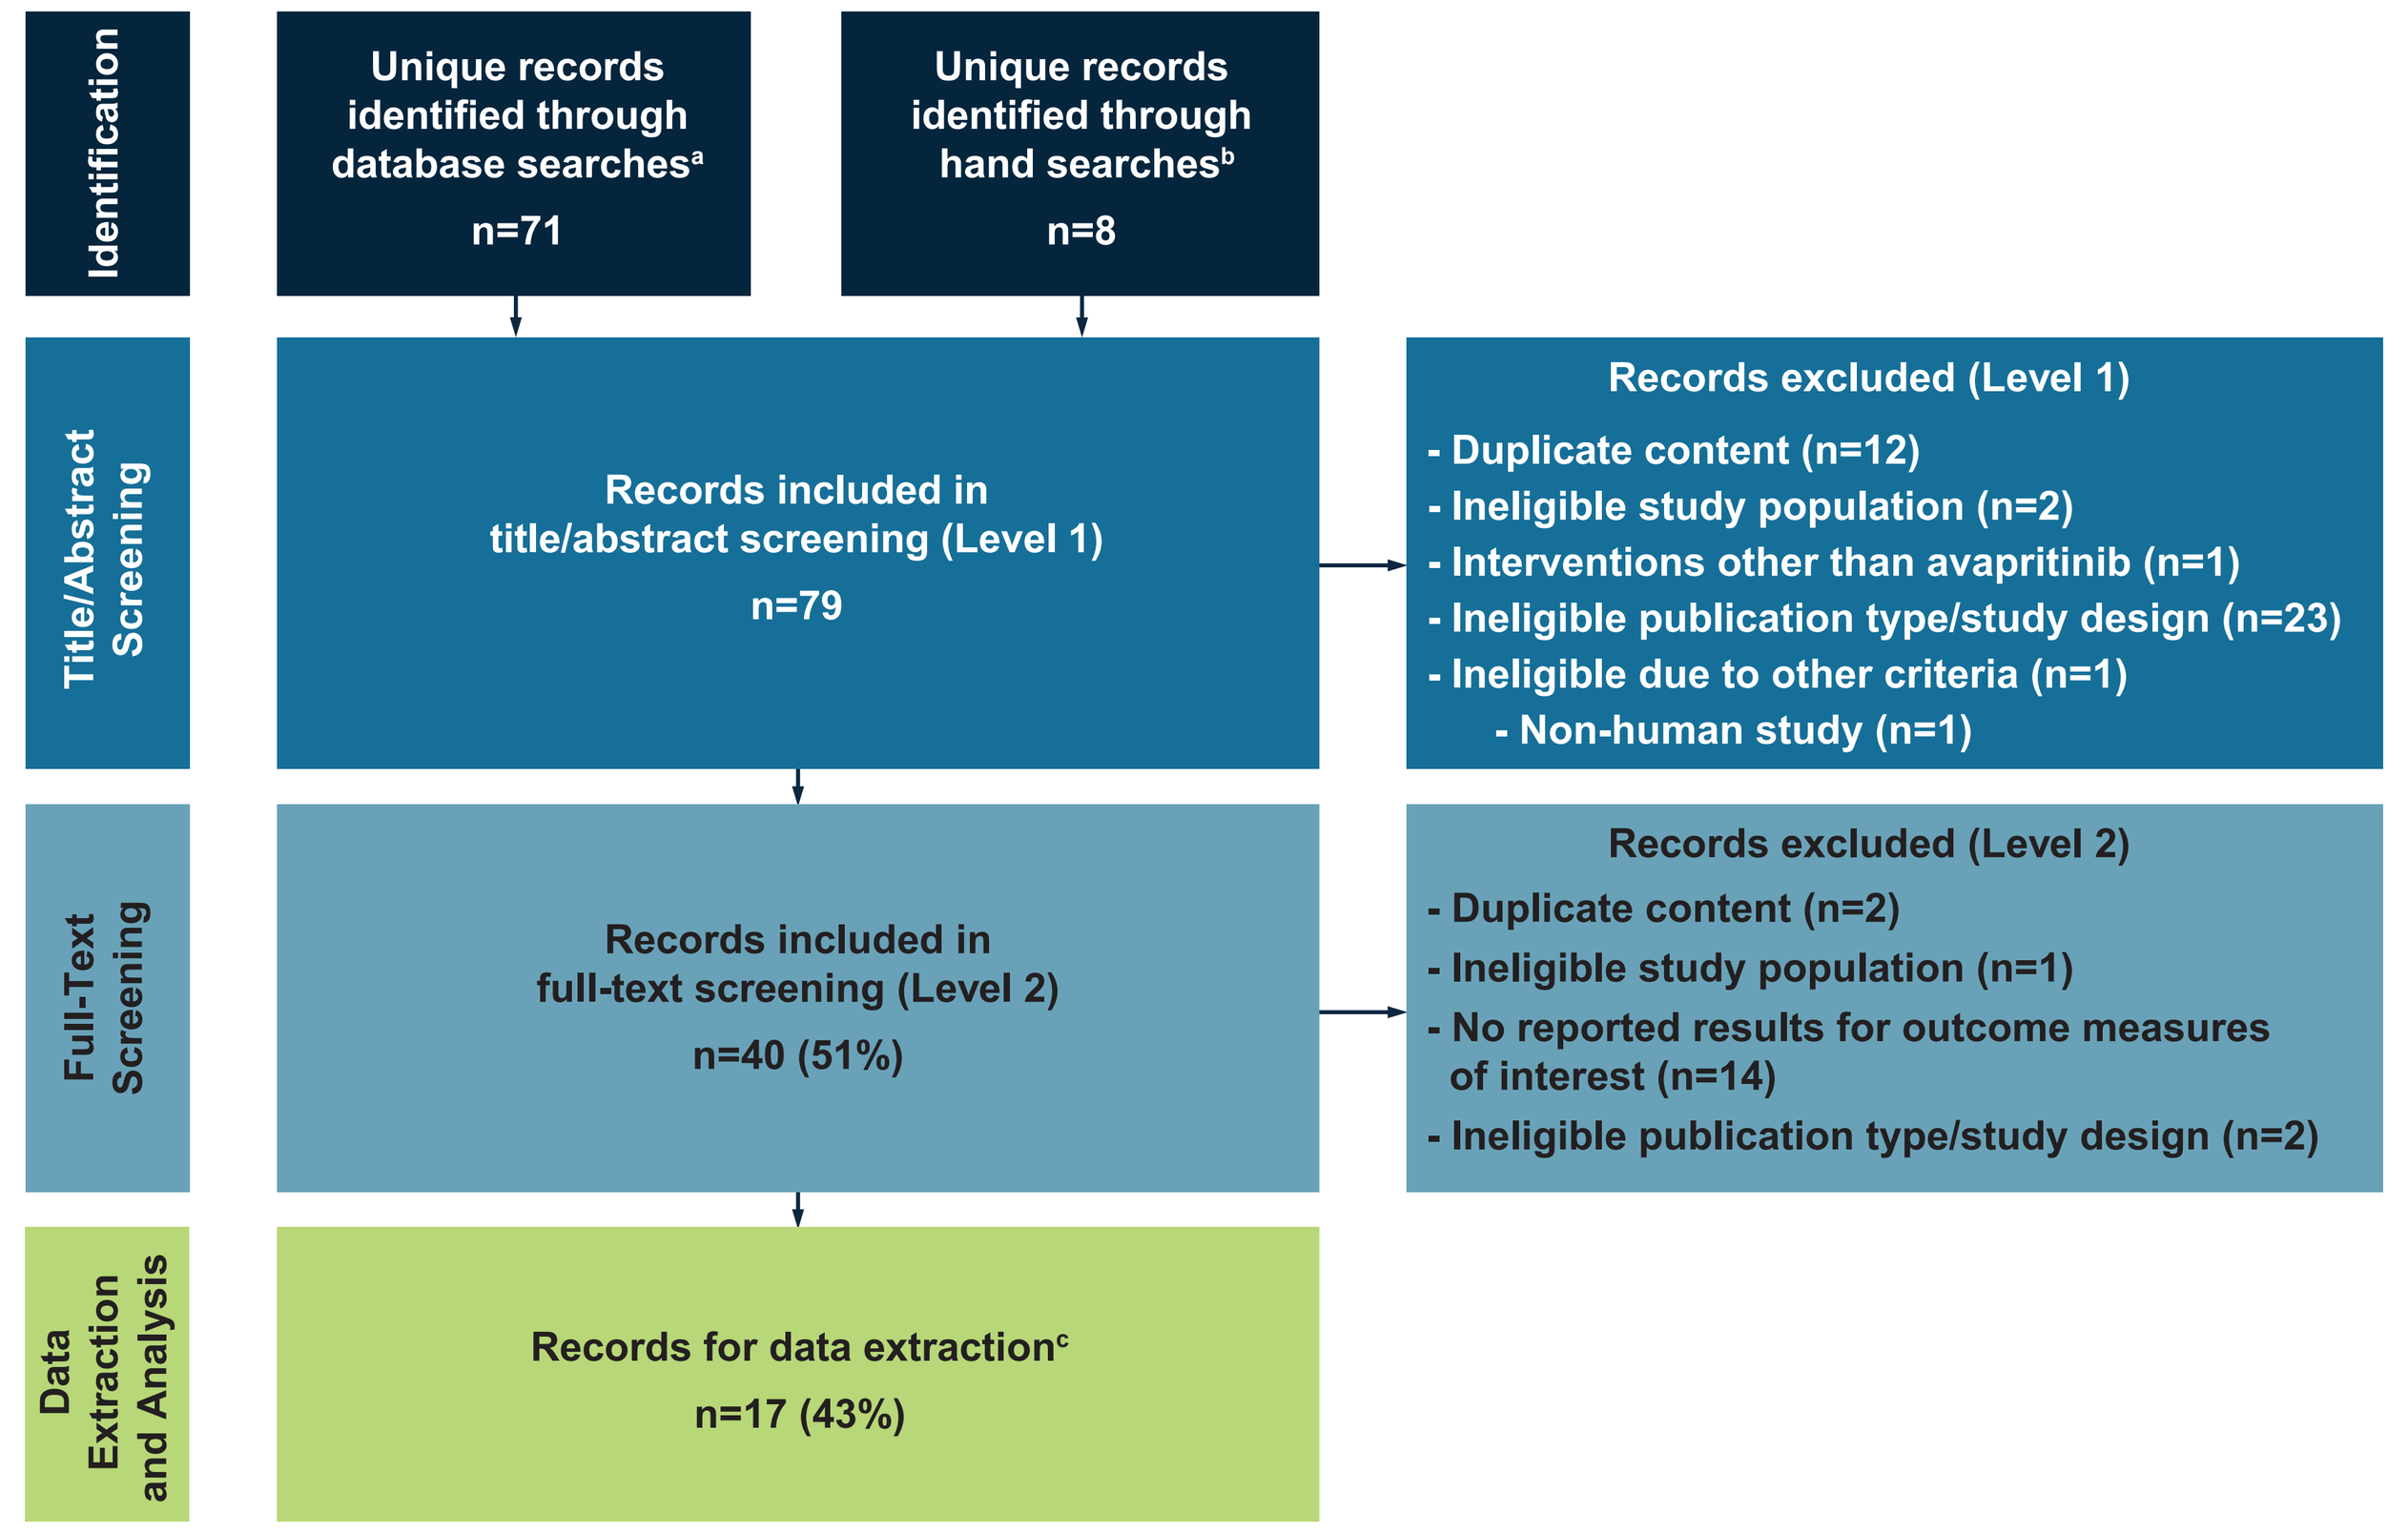
Identification
Unique records identified through database searches[a] n=71
Unique records identified through hand searches[b] n=8
Title/Abstract Screening
Records included in title/abstract screening (Level 1) n=79
Records excluded (Level 1)
- Duplicate content (n=12)
- Ineligible study population (n=2)
- Interventions other than avapritinib (n=1)
- Ineligible publication type/study design (n=23)
- Ineligible due to other criteria (n=1)
- Non-human study (n=1)
Full-Text Screening
Records included in full-text screening (Level 2) n=40 (51%)
Records excluded (Level 2)
- Duplicate content (n=2)
- Ineligible study population (n=1)
- No reported results for outcome measures of interest (n=14)
- Ineligible publication type/study design (n=2)
Data Extraction and Analysis
Records for data extraction[c] n=17 (43%)

Notes:
[a] Electronic databases were searched via OvidSP through April 23, 2025. Unique records were identified using OvidSP and EndNote de-duplication functionalities.
[b] Hand search cutoff date was June 30, 2025.
[c] This value excludes 4 records that met inclusion criteria but were deemed superseded by other captured records prior to data extraction.

Patient Clinical Characteristics

- Patients enrolled in both PIONEER (Part 1: N=39, Part 2: N=212, Part 3: N=226) and AVATAR (N=17) were required to have inadequately controlled ISM disease.[1,20]
- **Patient Demographics:**
 - The median patient ages reported across these studies ranged between 47–58 years, with the proportion of female patients ranging from 59–86%.[1,5-17,19,20]
- **Clinical Characteristics:**
 - Across all studies, baseline osteopenia and osteoporosis were identified in 19–71% and 22–47% of patients, respectively, with a history of spontaneous fractures reported in up to 29% of patients.[11,18,20]
 - In PIONEER Part 2, baseline anaphylaxis requiring treatment with epinephrine was reported in 7 (5%) patients receiving avapritinib and in none of the patients receiving placebo.[1] In the real-world AVATAR study, at baseline, anaphylaxis was observed in 35% of patients and hepato/splenomegaly in 12%.[20]
- **Molecular Profile:**
 - *KIT* D816V mutations were detected in >90% of patients across PIONEER, with median variant allele fractions (VAFs) around 0.3–0.4% in PIONEER and 1.1% in AVATAR.[5,8,12-14,20]
 - Median serum tryptase levels at baseline spanned 20–119 ng/mL.[1,5,6,8-10,12-17,19,20]

Treatment Information

- **Dosing and Duration:**
 - In PIONEER Part 1, avapritinib was tested at 25, 50, and 100 mg QD, with 25 mg QD selected as the RP2D and used in PIONEER Part 2;[1] the same dosage was also reported in AVATAR.[20]
 - In PIONEER Part 3 (open-label extension), patients from Parts 1 and 2 continued to receive avapritinib 25 mg QD with optional increase to 50 mg QD;[10,18] median treatment duration at 25 mg QD was 18 months, with 91% treated for ≥12 months.[10] Over 80% of patients receiving avapritinib remained on treatment during Part 3 after a median follow-up of 25.2 months (range: 0.7-53.5).
 - Retrospective PIONEER analyses included median treatment durations up to ~3 years.[18]
- **Prior Therapies:**
 - Across PIONEER studies, 12–16% of patients had received prior cytoreductive or tyrosine kinase inhibitor (TKI) therapy (e.g., midostaurin, imatinib, dasatinib, masitinib, interferon alfa).[1,14]
- **Best Supportive Care (BSC):**
 - Nearly all patients receiving avapritinib in PIONEER used ≥1 BSC medication at baseline (median: 3), most commonly antihistamines, leukotriene inhibitors, and cromolyn sodium.[1,5,7,8,12,13,16]
 - Opioid use at baseline was reported in up to 18% of patients receiving avapritinib.[7]

Efficacy Outcomes

- **ISM Symptom Assessment Form (ISM-SAF):**
 - After 24 weeks of treatment in PIONEER Part 2, avapritinib led to meaningful QoL gains, with avapritinib-treated patients experiencing a 15.6-point decrease in Total Symptom Score (TSS) on the ISM-SAF on average compared with an average 9.2-point decrease among placebo-treated patients (*P*=0.003) (**Table 2**).[1]
 - In PIONEER Part 3, the mean decrease in TSS among avapritinib-treated patients was 17.66 (standard deviation [SD]: 19.32) by Week 48 and 18.80 (SD: 22.46) by Week 96, highlighting a durable improvement in QoL with avapritinib.[10]

Table 2. Avapritinib Efficacy Outcomes

	PIONEER			AVATAR[20]
	Part 1[14] (Week 24)	Part 2[1] (Week 24)	Part 3[10,a] (Weeks 48 & 96)	(Week 24)
Reduction in ISM-SAF TSS, mean	—	Avapritinib: -15.6 Placebo: -9.2 *P*=0.003	Avapritinib: Week 48: -17.66 Week 96: -18.80	—
Proportion with ≥50% reduction in serum tryptase	Avapritinib: 70% Placebo: 0%	Avapritinib: 54% Placebo: 0% *P*<0.001	—	—
Reduction in serum tryptase, mean %	—	—	—	Avapritinib: -67.1%
Proportion with ≥50% reduction in *KIT* D816V VAF	—	Avapritinib: 68% Placebo: 6% *P*<0.001	—	—
Reduction in *KIT* D816V VAF, mean %	—	—	—	Avapritinib: -48.1%

Abbreviations: ISM: indolent systemic mastocytosis; ISM SAF: Indolent Systemic Mastocytosis-Symptom Assessment Form; *KIT*: *KIT* gene; TSS: Total Symptom Score; VAF: variant allele fraction.
Note:
[a] On-study patients who received avapritinib 25 mg QD were included for analysis.

- **Mastocytosis-QoL (MC-QoL):**
 - After 24 weeks of treatment, MC-QoL scores, reflective of mastocytosis-related impairment, decreased by 34% among patients receiving avapritinib in PIONEER Part 2 versus 18% decrease among patients in the placebo arm (**Figure 2**),[1] underscoring substantial improvements in mastocytosis-related burden for patients receiving avapritinib over placebo. MC-QoL improvements were consistent across symptom severity subgroups[9] and were sustained to 96 weeks of treatment.[10]
 - Domain-specific reductions in mastocytosis-related impairment, as measured by MC-QoL, were also observed by Week 24 in PIONEER Part 2, with the strongest reductions observed in Emotions and Social Life/Functioning (**Figure 3**).[1]
 - At Week 24 in the real-world AVATAR study, the median MC-QoL score decreased from 61 at baseline to 17, reflecting a 76% reduction in mastocytosis-related impairment (**Figure 4**).[20] In the AVATAR study, the greatest domain-specific improvements in MC-QoL were Social Life/Functioning and Emotions.

Figure 2. PIONEER: Improvement in Mastocytosis-Related Impairment (MC-QoL) Overall and across Subgroup Analyses by Week 24[1,6,9,a]

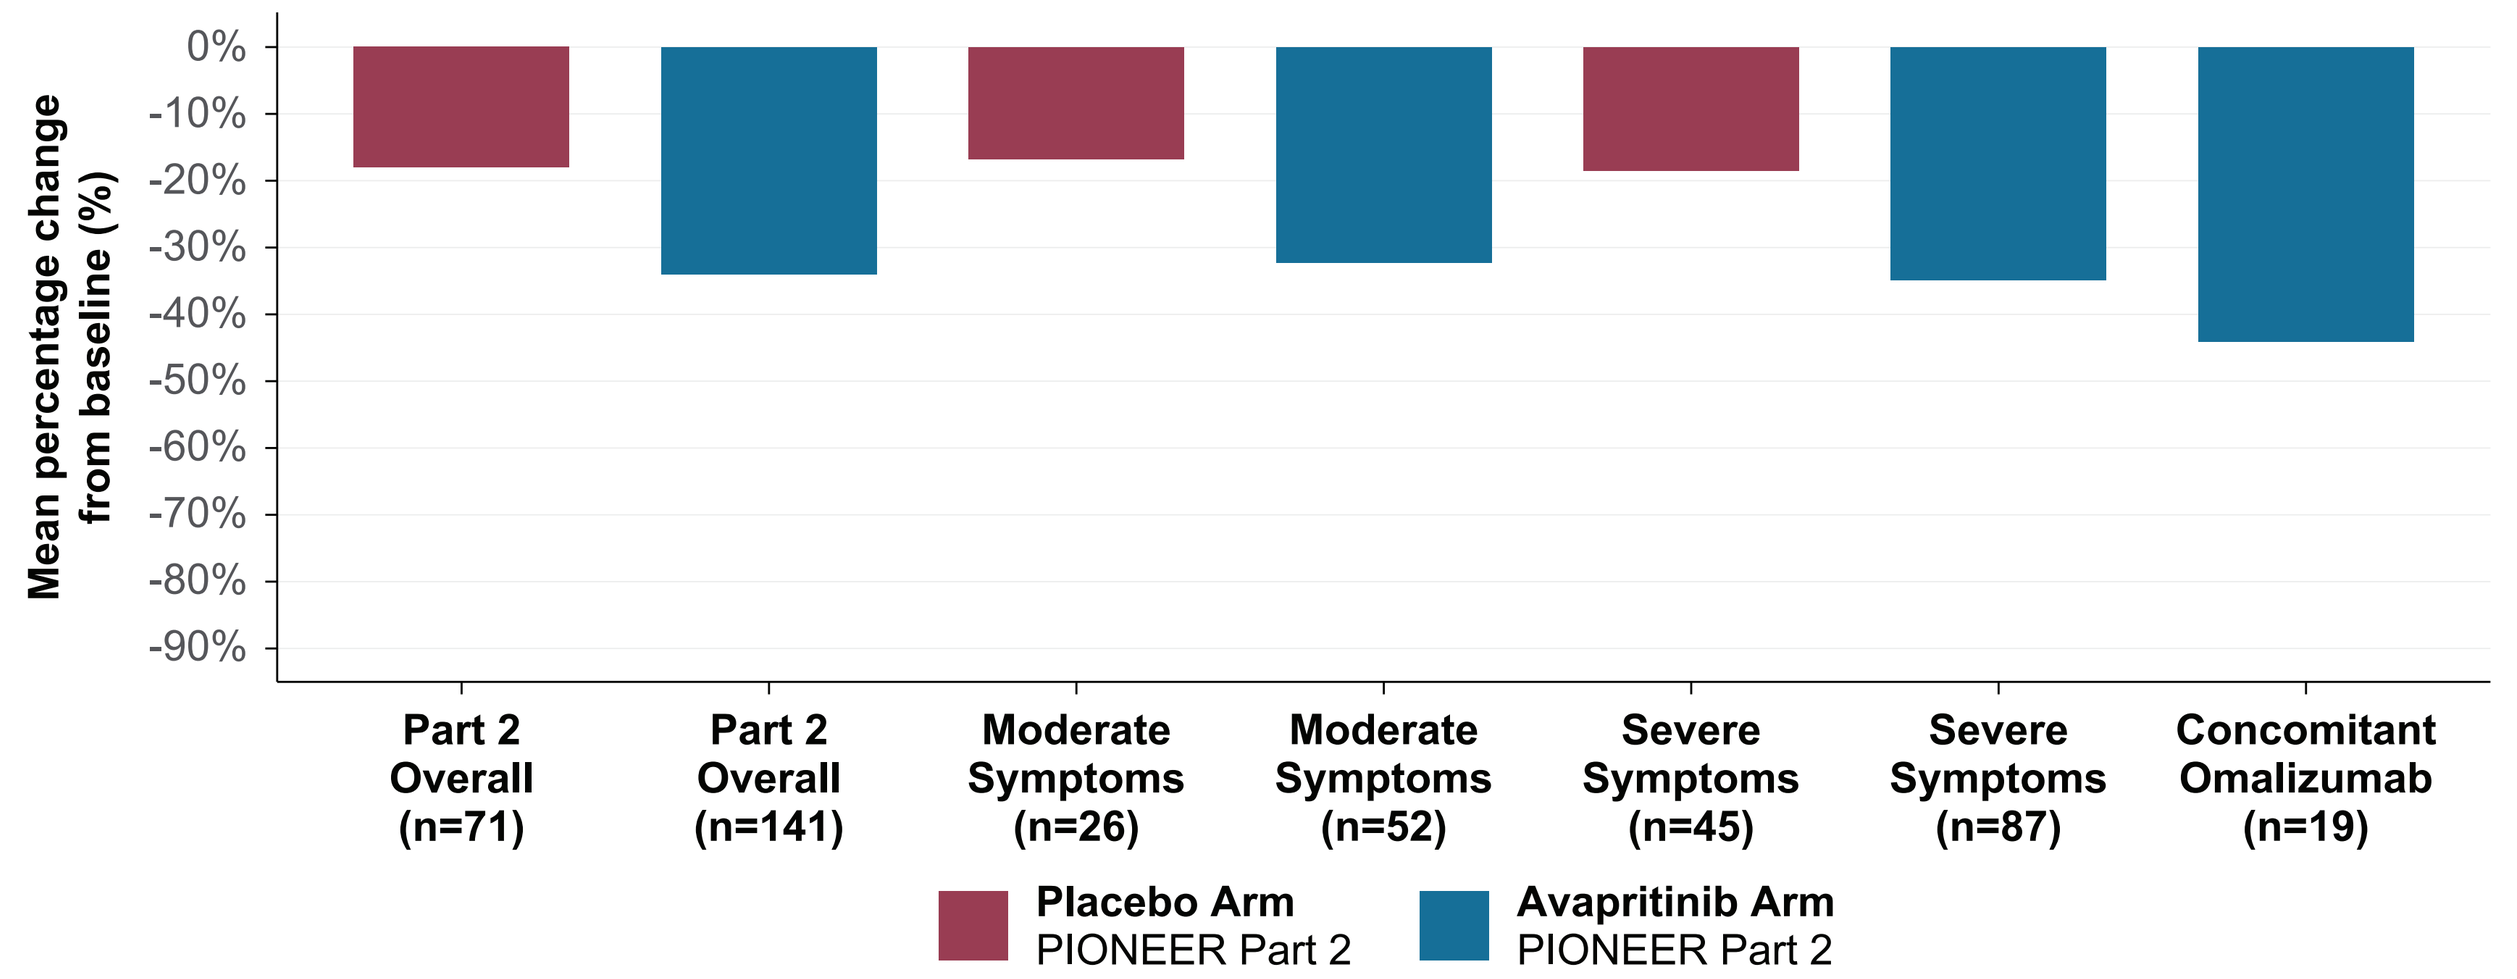
Mean percentage change from baseline (%)
0%
-10%
-20%
-30%
-40%
-50%
-60%
-70%
-80%
-90%
Part 2 Overall (n=71)
Part 2 Overall (n=141)
Moderate Symptoms (n=26)
Moderate Symptoms (n=52)
Severe Symptoms (n=45)
Severe Symptoms (n=87)
Concomitant Omalizumab (n=19)
Placebo Arm PIONEER Part 2
Avapritinib Arm PIONEER Part 2

Abbreviations: ISM SAF: Indolent Systemic Mastocytosis-Symptom Assessment Form; MC-QoL: Mastocytosis-Quality of Life; TSS: Total Symptom Score.
Note:
[a] MC-QoL scores range from 0 to 100, with higher scores indicating worse impairment and severity levels defined as severe (>60.0), moderate (>40.0 to ≤60.0), mild (>20.0 to ≤40.0) and none (≤20.0) as previously reported.[1] Symptom and concomitant omalizumab subgroups include participants in PIONEER Part 2 only.[6,9] Moderate and Severe Symptom subgroups in PIONEER Part 2 were defined by the ISM-SAF TSS, with moderate symptoms corresponding to TSS of 28-42 and severe symptoms corresponding to ≥42 at baseline.[9]

Figure 3. PIONEER: Overall and Domain-Specific Improvements in Mastocytosis-Related Impairment (MC-QoL) by Week 24[1,a]

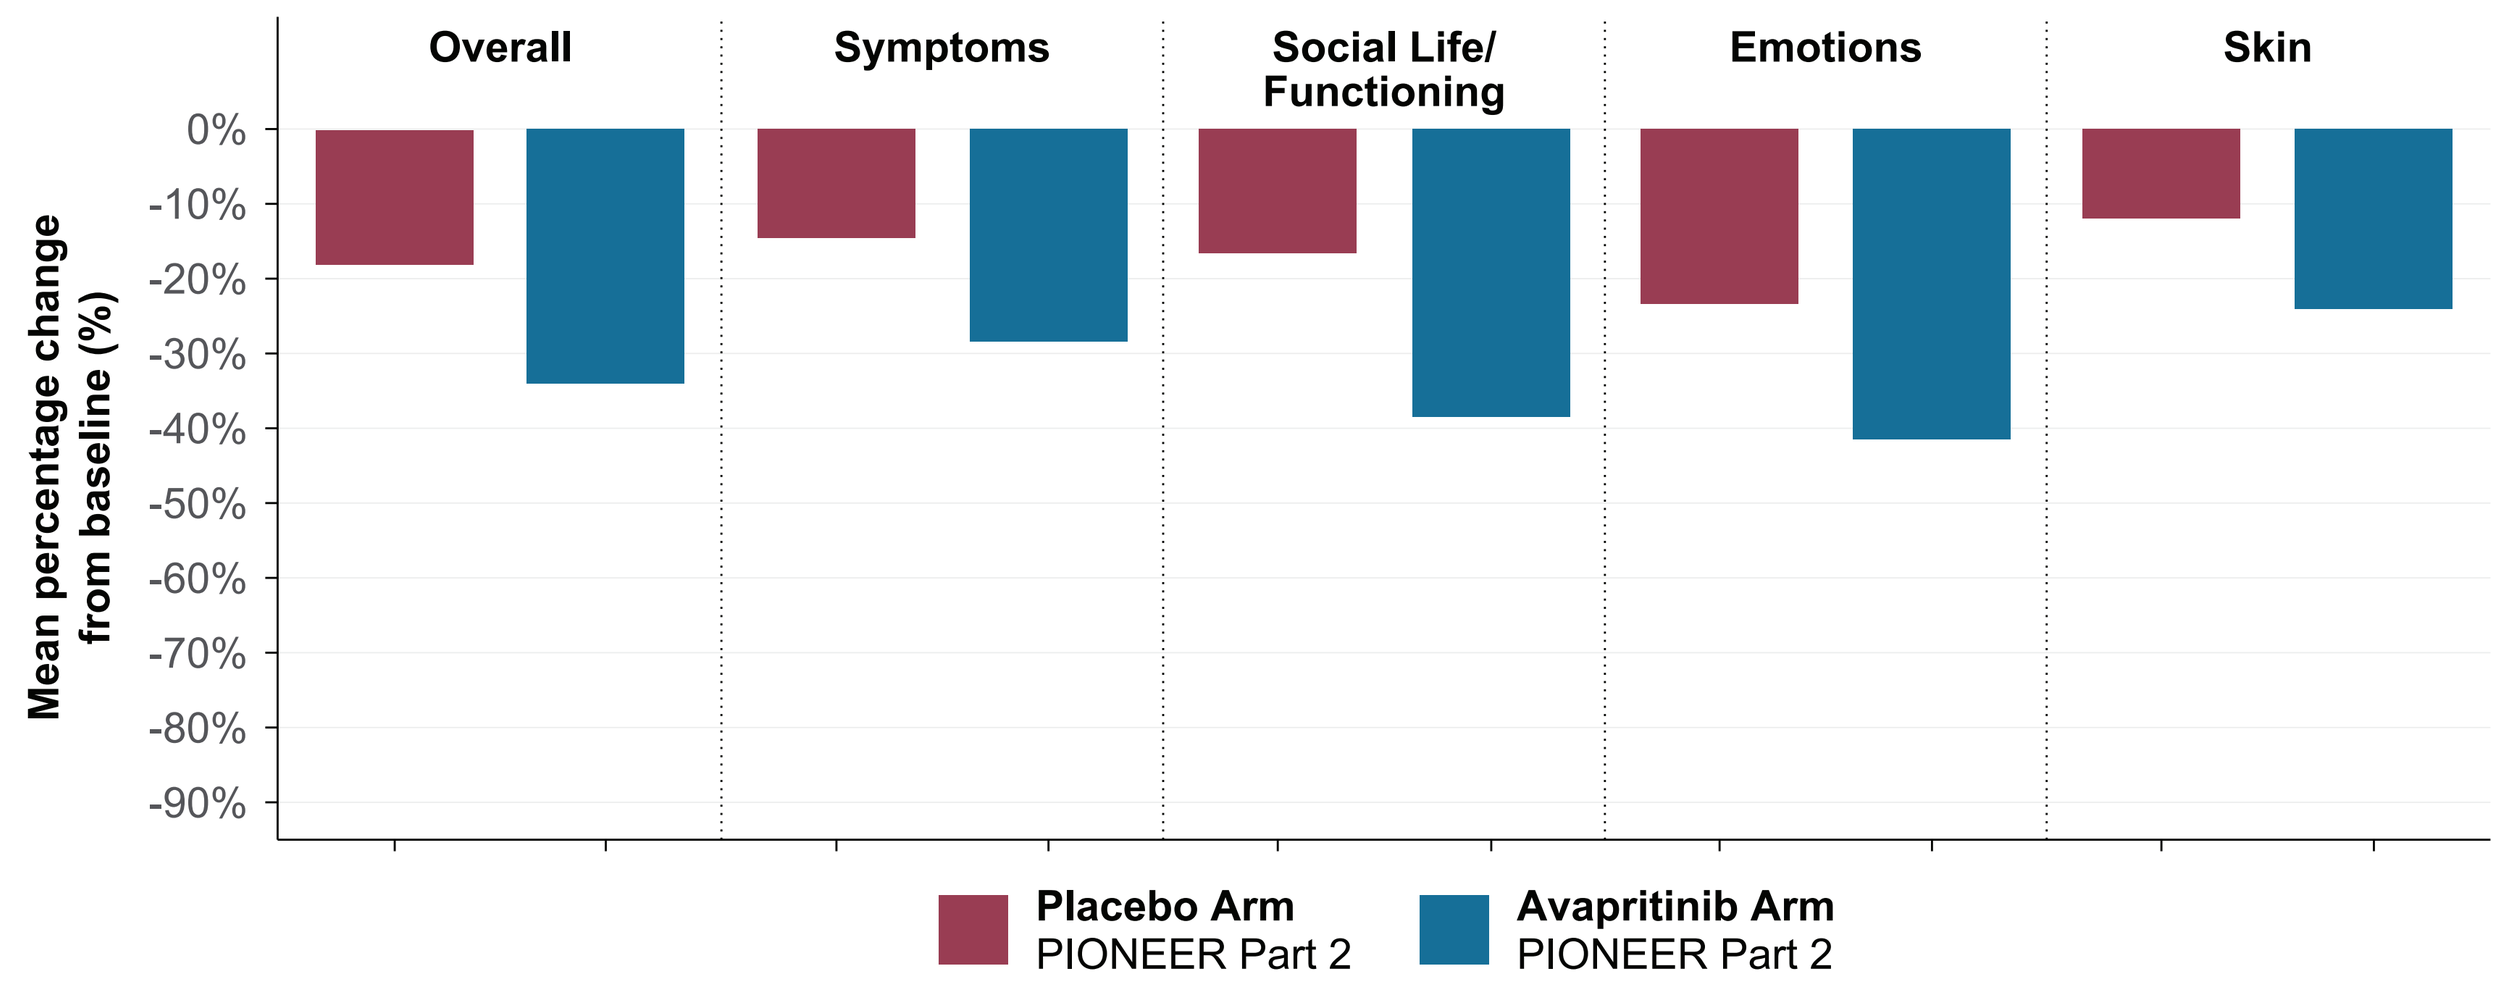
Overall
Symptoms
Social Life/ Functioning
Emotions
Skin
Mean percentage change from baseline (%)
0%
-10%
-20%
-30%
-40%
-50%
-60%
-70%
-80%
-90%
Placebo Arm PIONEER Part 2
Avapritinib Arm PIONEER Part 2

Abbreviations: MC-QoL: Mastocytosis-Quality of Life.
Note:
[a] MC-QoL scores range from 0 to 100, with higher scores indicating worse impairment and severity levels defined as severe (>60.0), moderate (>40.0 to ≤60.0), mild (>20.0 to ≤40.0) and none (≤20.0) as previously reported.[1]

Figure 4. AVATAR: Overall and Domain-Specific Improvements in Mastocytosis-Related Impairment (MC-QoL) by Week 24[20,a]

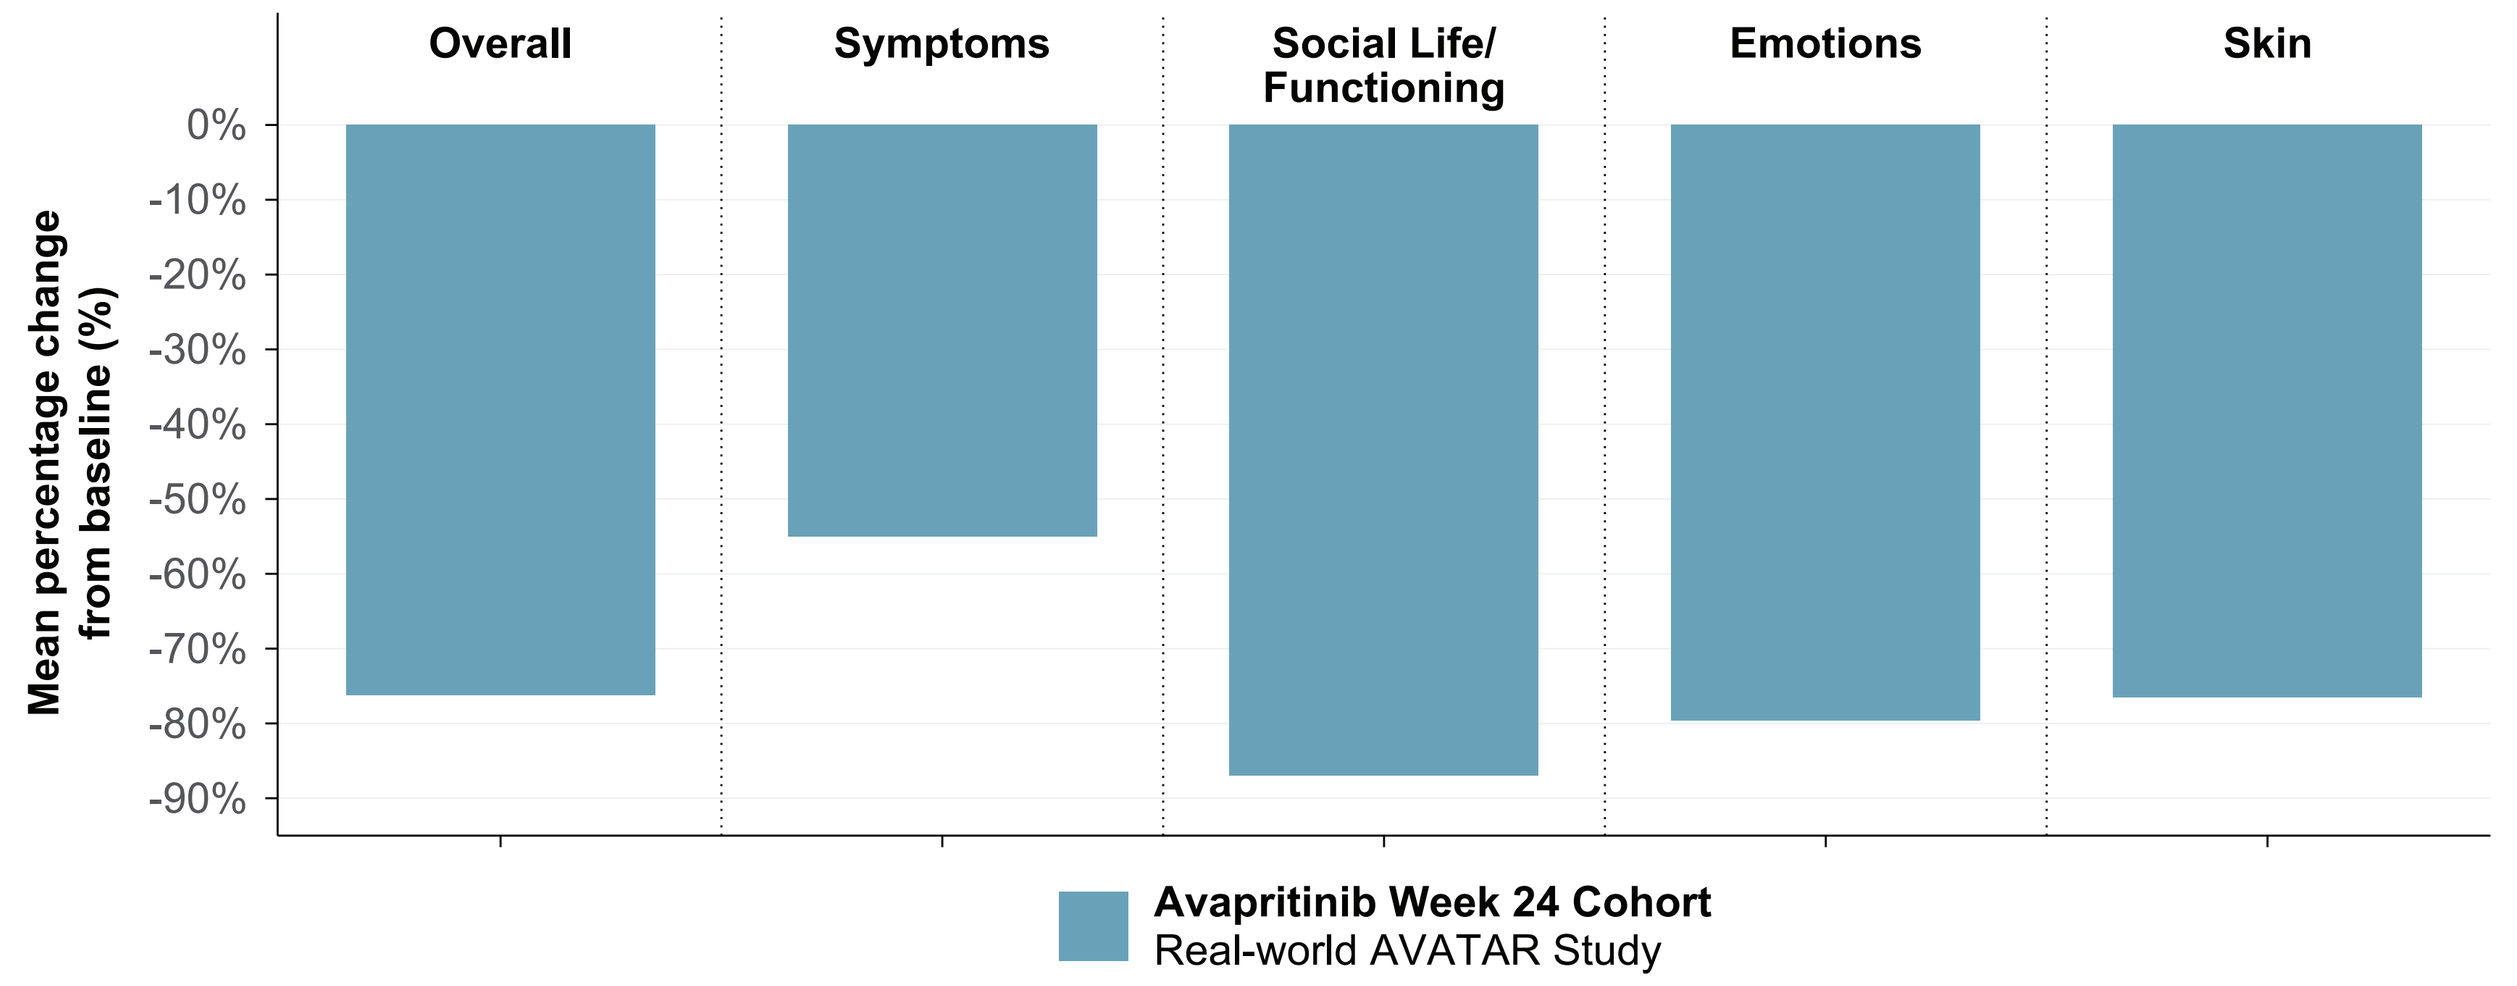
Overall
Symptoms
Social Life/ Functioning
Emotions
Skin
Mean percentage change from baseline (%)
0%
-10%
-20%
-30%
-40%
-50%
-60%
-70%
-80%
-90%
Avapritinib Week 24 Cohort Real-world AVATAR Study

Abbreviations: MC-QoL: Mastocytosis-Quality of Life.
Note:
[a] MC-QoL scores range from 0 to 100, with higher scores indicating worse impairment and severity levels defined as severe (>60.0), moderate (>40.0 to ≤60.0), mild (>20.0 to ≤40.0) and none (≤20.0) as previously reported.[1]

- **Efficacy and Molecular Outcomes:**
 - Across PIONEER and AVATAR, avapritinib produced substantial molecular responses among patients (**Table 2**), with ≥50% reductions in serum tryptase in 54–70% of patients.[1,8,13,14,16,20]
 - In PIONEER Part 2, 68% of patients experienced ≥50% reductions in *KIT* D816V VAF compared with 6% on placebo.[1,8,13,16] KIT mutations became undetectable in 11% of avapritinib-treated patients after 24 weeks of treatment.[13]
- **Other Clinical Improvements:**
 - Improvements in bone health and disease manifestations were observed in patients receiving avapritinib, including increased bone mineral density (up to +4.8% in the lumbar spine after 2 years), and lower mast cell burden, with notable improvements in cutaneous lesions.[1,11,13,14,16,18]
 - Anaphylactic episodes requiring epinephrine treatment were rare; in PIONEER Part 2, 1% (n=2) and 4% (n=3) reported anaphylaxis with avapritinib and placebo, respectively, by Week 24.[1]
- **Polypharmacy Reduction:**
 - Patients in PIONEER Part 2 also noted reduced need for supportive medications, with 31% lowering usage of BSC and 4% discontinuing BSC by Week 48.[1,5]

Safety Outcomes

- **Safety Profile:**
 - Across PIONEER studies, avapritinib demonstrated a safety profile comparable to placebo. In PIONEER Part 2, treatment-related adverse events (TRAEs) were reported in 55% of the avapritinib arm and 45% of the placebo arm, with Grade ≥3 TRAEs occurring in ≤3% for both arms.[1,5,8,10,12,13,16] By 96 weeks of treatment during the open-label extension, 70% (n=158) of avapritinib-treated patients experienced TRAEs of any grade and only 6% (n=13) experienced Grade ≥3 TRAEs, supporting a favorable safety profile for avapritinib over long-term therapy.
 - The most frequent TRAEs (≥5%) for patients receiving 25 mg QD avapritinib in all parts of PIONEER (median follow-up: 25.2 months) were headache, nausea, peripheral and periorbital edema, diarrhea, fatigue, and alopecia.[10]
 - By Week 24 in PIONEER Part 2, serious AEs (SAEs) occurred in 5% of avapritinib recipients and 11.3% of placebo recipients and were not treatment-related.[1] For patients receiving 25 mg QD avapritinib in all parts of PIONEER, 3 (1%) treatment-related SAEs were observed over 96 weeks of treatment.[10]

Conclusions

- Across 17 included studies, avapritinib 25 mg QD produced meaningful reductions in disease burden and durable improvements in symptoms and QoL for patients with ISM.
- Consistent biomarker and symptom improvements were observed, with ≥50% reductions in serum tryptase and *KIT* D816V VAF and improvements across all individual symptoms measured by ISM-SAF (i.e., skin, gastrointestinal, neurocognitive, systemic bone pain and fatigue).
- Avapritinib was well tolerated and demonstrated a comparable safety profile to placebo in PIONEER Part 2. Results from the open-label extension suggest that the safety profile for avapritinib remains positive over long-term treatment.
- This SLR provides the most current and comprehensive synthesis of outcomes data for patients with ISM treated with avapritinib, which can inform evidence-based clinical decision-making for this population.
- The evidence base for avapritinib is primarily derived from clinical trials. Additional real-world studies are ongoing to further inform the use of avapritinib for the treatment of ISM in routine clinical practice.

Acknowledgements

This study was funded by Blueprint Medicines Corporation.

References

1. Gotlib J, Castells M, Elberink HO, et al. *NEJM Evid.* 2023;2(6):EVIDoa2200339.
2. Garcia-Montero AC, Jara-Acevedo M, Teodosio C, et al. *Blood.* 2006;108(7):2366-2372.
3. Kristensen T, Vestergaard H, Bindslev-Jensen C, Moller MB, Broesby-Olsen S, Mastocytosis Centre OUH. *Am J Hematol.* 2014;89(5):493-498.
4. Ungerstedt J, Ljung C, Klimkowska M, Gulen T. *Cancers (Basel).* 2022;14(16).
5. Akin C, Gotlib J, Alvarez-Twose I, et al. AAAAI; February 23-26, 2024; Washington, DC.
6. Akin C, Hartmann K, Broesby-Olsen S, et al. ACAAI; October 24-28, 2024; Boston, MA.
7. Akin C, Siebenhaar F, Gotlib J, et al. AAAAI; February 24-27, 2023; San Antonio, TX.
8. Castells M, Gotlib J, Elberink HO, et al. AAAAI; February 24-27, 2023; San Antonio, TX.
9. Castells M, Maurer M, Hartmann K, et al. ACAAI; October 24-28, 2024; Boston, MA.
10. Sabato V, Gotlib J, Castells M, et al. EAACI; May 31-June 3, 2024; Valencia, Spain.
11. Siebenhaar F, Alvarez-Twose I, Sundaresh V, et al. AAAAI/WAO Joint Congress; February 28-March 3, 2025; San Diego, CA.
12. Giannetti M, Akin C, Livideanu C, et al. AIM; May 18-19, 2024; Boston, MA.
13. Gotlib J, Castells M, Elberink HO, et al. SOHO; September 6–9, 2023; Houston, TX.
14. Hartmann K, Broesby-Olsen S, Siebenhaar F, et al. EAACI; June 6-8, 2020; Remote.
15. Maurer M, Akin C, Broesby-Olsen S, et al. AAAAI; February 23-26, 2024; Washington, DC.
16. Maurer M, Siebenhaar F, Broesby-Olsen S, et al. AAAAI; February 24-27, 2023; San Antonio, TX.
17. Pongdee T, Vachhani P, Tashi T, et al. AAAAI/WAO Joint Congress; February 28-March 3, 2025; San Diego, CA.
18. Akin C, Hartmann K, Broesby-Olsen S, George T, Lin H, Siebenhaar F. EAC; May 29-June 1, 2025; Palm Beach, FL.
19. George T, Akin C, Pongdee T, et al. EAC; May 29-June 1, 2025; Palm Beach, FL.
20. Pyatilova P, le Coutre P, Herzog L, et al. AAAAI/WAO Joint Congress; February 28-March 3, 2025; San Diego, CA.

Presented at the **American Academy of Allergy, Asthma & Immunology Annual Meeting**, Philadelphia, PA , February 27–March 2, 2026. Please contact Robyn Scherber, rscherber@blueprintmedicines.com for permission to reprint and/or distribute.