Avapritinib Durably Improves Gastrointestinal Symptoms of Indolent Systemic Mastocytosis: Long-Term Outcomes From the PIONEER Study

Poster Number 518

Matthew P. Giannetti,[1] Mariana Castells,[1] Cem Akin,[2] Thanai Pongdee,[3] Saranya Venugopal,[4] Hui-Min Lin,[4] Rahul Kalathiya,[4] Kate Newberry,[4] Matthew J. Hamilton[5]

[1]Division of Allergy and Clinical Immunology, Department of Medicine, Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA; [2]Division of Allergy and Clinical Immunology, Department of Internal Medicine, University of Michigan, Ann Arbor, MI, USA; [3]Division of Allergic Diseases, Mayo Clinic, Rochester, MN, USA; [4]Blueprint Medicines Corporation, Cambridge, MA, USA; [5]Division of Gastroenterology, Hepatology, and Endoscopy, Department of Medicine, Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA

Introduction

- Indolent systemic mastocytosis (ISM) is a clonal mast cell disease driven by the *KIT* D816V mutation in ~95% of adult cases[1–4]
- Patients with ISM can have lifelong debilitating symptoms across multiple organ systems[5–9] (**Figure 1**)
 - Gastrointestinal (GI) symptoms, including nausea, vomiting, abdominal bloating, abdominal pain, and diarrhea, are frequent issues impacting quality of life in many patients with ISM[7]
- GI symptom management includes frequent use of mast-cell stabilizers (e.g., oral cromolyn sodium and ketotifen), H1 and H2 antihistamines, and proton pump inhibitors[9,10]
- PIONEER is the first and largest randomized, double-blind, placebo-controlled trial of a highly selective KIT D816V-targeting agent in patients with ISM and led to the approval of avapritinib by the FDA[11,12]
- In the PIONEER study (NCT03731260),[13] 24 weeks of avapritinib 25 mg once-daily (QD) treatment plus best supportive care (BSC) improved mean total symptom score (TSS) as measured by the ISM-Symptom Assessment Form (ISM-SAF; ©2018 Blueprint Medicines Corporation), a validated tool specifically developed for evaluation of ISM symptomology based on self-reported severity of 11 ISM symptoms (scores range from 0–110, with moderate to severe defined as a TSS ≥28)[14–16] *versus* placebo in patients with ISM, and these effects were durable over Years 1 through 4 of avapritinib treatment [see AAAAI Poster 509]
 - At 24 weeks, avapritinib 25 mg QD plus BSC also improved ISM-SAF–assessed GI symptoms *versus* placebo plus BSC[17]
- Here, we report GI outcomes with long-term avapritinib from PIONEER

Figure 1. ISM Symptoms

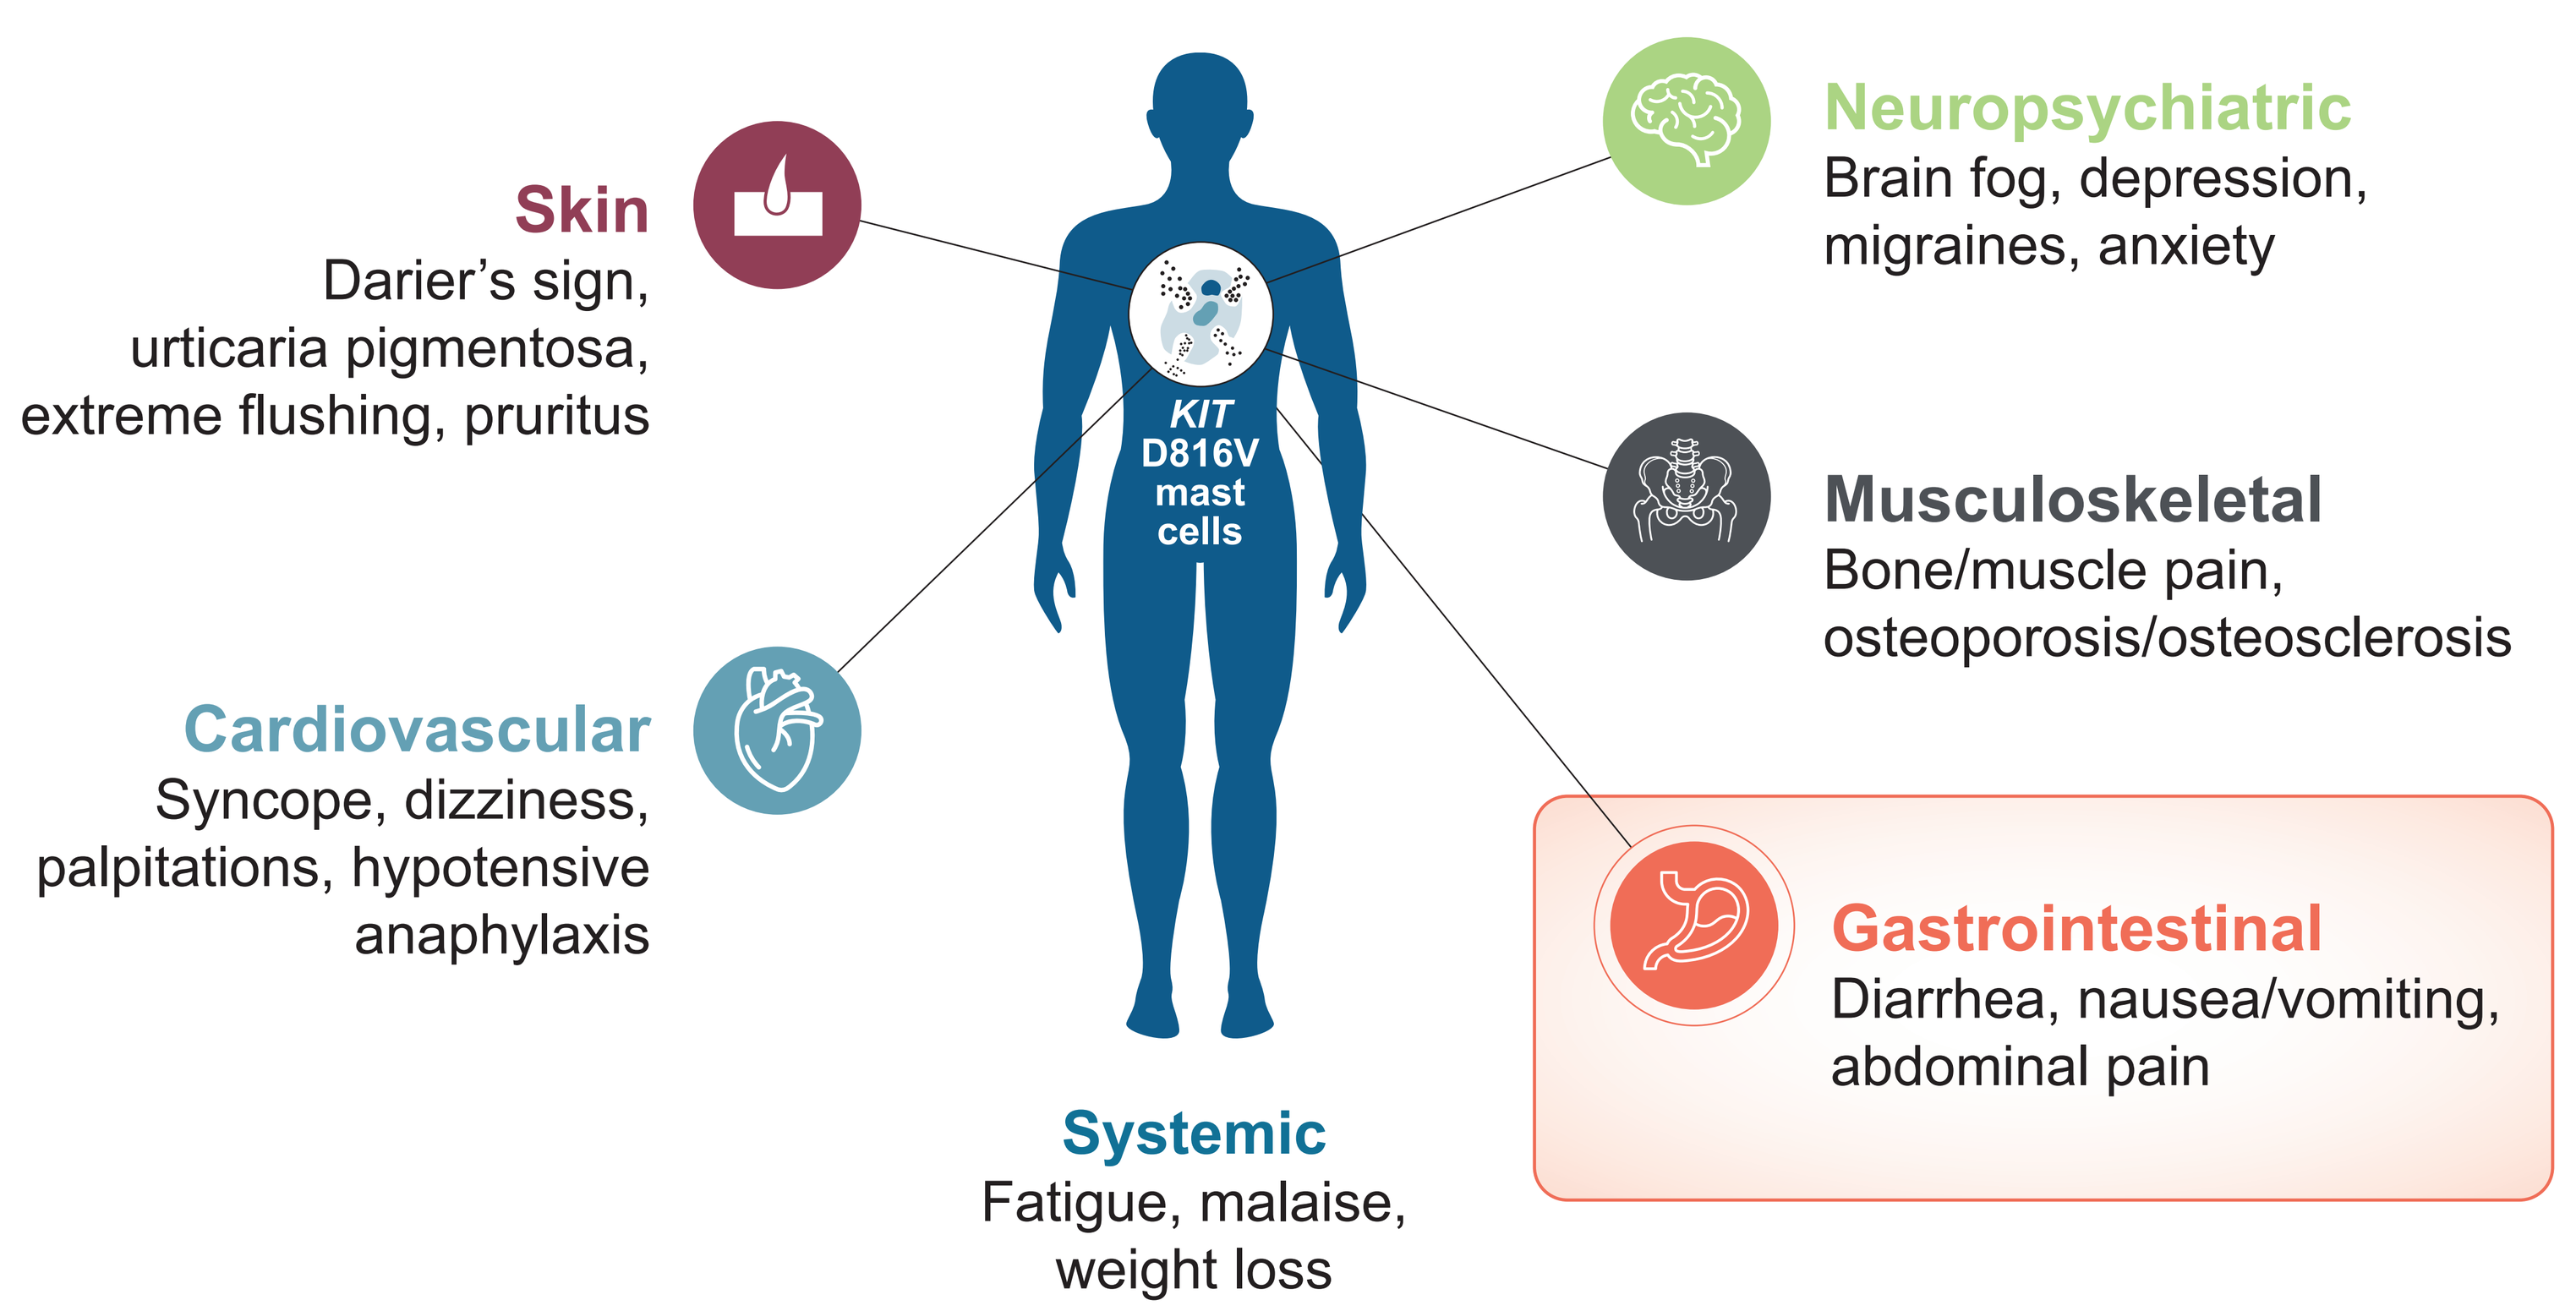

Skin
Darier's sign, urticaria pigmentosa, extreme flushing, pruritus
Cardiovascular
Syncope, dizziness, palpitations, hypotensive anaphylaxis
KIT D816V mast cells
Systemic
Fatigue, malaise, weight loss
Neuropsychiatric
Brain fog, depression, migraines, anxiety
Musculoskeletal
Bone/muscle pain, osteoporosis/osteosclerosis
Gastrointestinal
Diarrhea, nausea/vomiting, abdominal pain

ISM, indolent systemic mastocytosis.

Methods

- Patients with moderate to severe ISM symptoms who completed the randomized dose-finding (Part 1), or randomized, double-blind, placebo-controlled (Part 2) portions of PIONEER rolled over to the open-label, long-term extension (Part 3) with up to 5-year follow-up (**Figure 2**)
- Individual ISM-SAF GI symptom scores of diarrhea (0–10), abdominal pain (0–10), and nausea (0–10) severity, and diarrhea frequency (14-day diarrhea count average) were assessed
- 4-year long-term efficacy and cumulative safety from initiation of avapritinib at 25 mg QD are presented up to the data cut-off date of September 17, 2025
- Changes in the dose or frequency of BSC use for ISM-related GI symptoms (H1 and H2 antihistamines, and cromolyn sodium) at 3 years (156 Weeks) from the initiation of avapritinib 25 mg QD are also presented; as of the data cut-off date, all enrolled patients had reached this minimum three-year treatment duration (or discontinued the study)

Figure 2. PIONEER study design

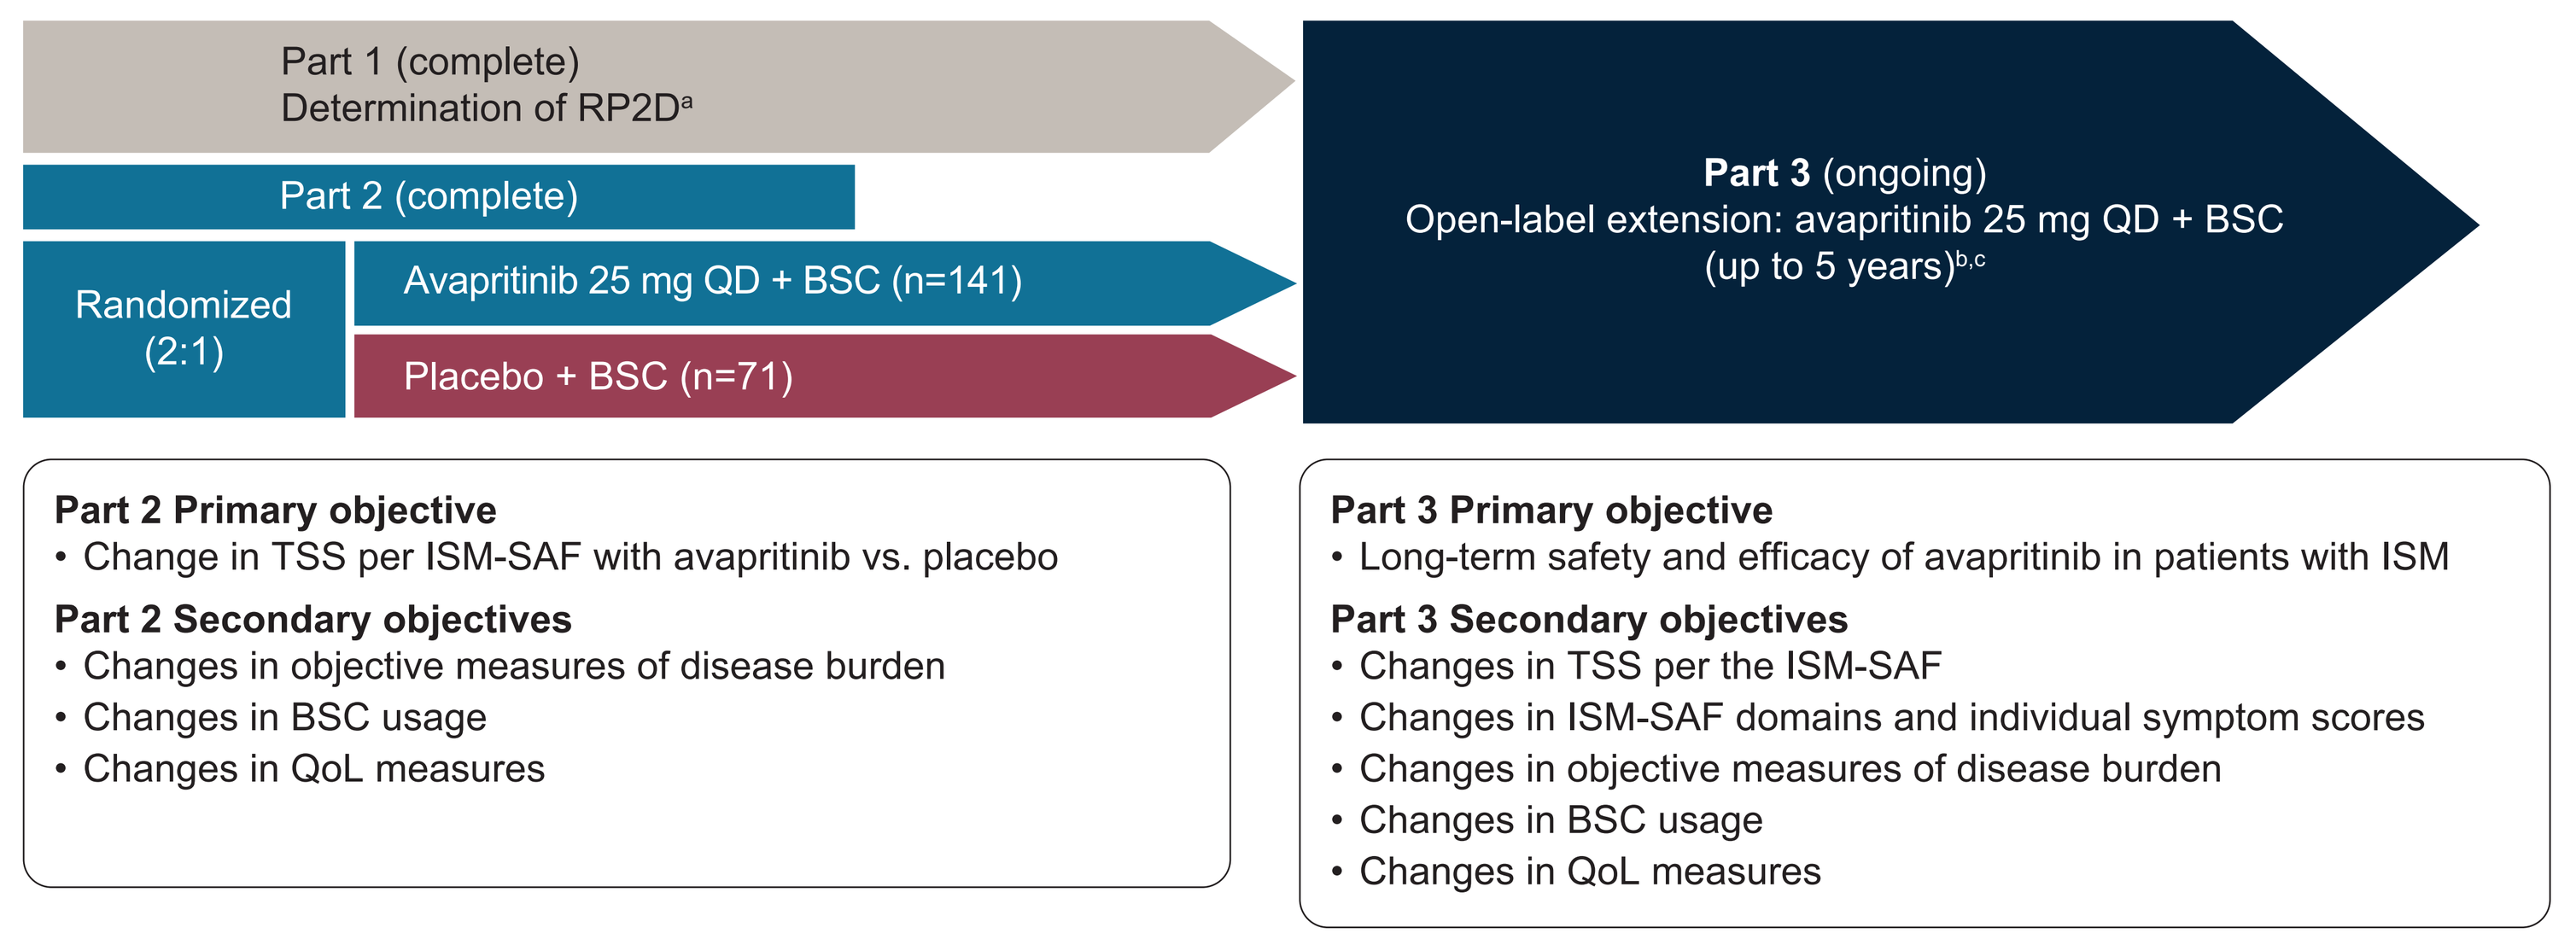

Part 1 (complete)
Determination of RP2D[a]
Part 2 (complete)
Randomized (2:1)
Avapritinib 25 mg QD + BSC (n=141)
Placebo + BSC (n=71)
Part 3 (ongoing)
Open-label extension: avapritinib 25 mg QD + BSC (up to 5 years)[b,c]
Part 2 Primary objective
• Change in TSS per ISM-SAF with avapritinib vs. placebo
Part 2 Secondary objectives
• Changes in objective measures of disease burden
• Changes in BSC usage
• Changes in QoL measures
Part 3 Primary objective
• Long-term safety and efficacy of avapritinib in patients with ISM
Part 3 Secondary objectives
• Changes in TSS per the ISM-SAF
• Changes in ISM-SAF domains and individual symptom scores
• Changes in objective measures of disease burden
• Changes in BSC usage
• Changes in QoL measures

[a]The RP2D of avapritinib was identified based on efficacy and safety results from Part 1 that included 4 cohorts: 25 mg avapritinib (n=10), 50 mg avapritinib (n=10), 100 mg avapritinib (n=10) and placebo (n=9). [b]N=226, includes patients from Part 1 who continued avapritinib 25 mg QD or crossed over from placebo to avapritinib 25 mg QD. This also includes patients from Part 2 who received avapritinib 25 mg QD or who crossed over from placebo to avapritinib 25 mg. [c]Patients could dose increase to 50 mg QD in Part 3.
BSC, best supportive care; ISM-SAF, Indolent Systemic Mastocytosis-Symptom Assessment Form; QD, once daily; QoL, quality of life; RP2D, recommended Part 2 dose; TSS, total symptom score.

Results

- Baseline patient demographics and characteristics across all patients treated with avapritinib 25 mg QD are presented in **Table 1**
- The mean change (standard deviation) in ISM-SAF TSS was –17.66 (19.32) at Week 48 (henceforth 1 year) and –17.66 (19.07) at Week 204 (henceforth 4 years), in all patients treated with avapritinib

Table 1. Baseline demographic and characteristics

Patient demographic	Avapritinib 25 mg QD (n=226)
Age (years), median (range)	51 (18–79)
Female, n (%)	166 (74)
ISM-SAF TSS score, mean (SD; n)	48.1 (19.5; 224)
ISM-SAF GI domain score, mean (SD; n)	9.1 (6.3; 224)
ISM-SAF abdominal pain severity, mean (SD; n)	3.8 (2.4; 224)
ISM-SAF nausea severity, mean (SD; n)	2.8 (2.5; 224)
ISM-SAF diarrhea severity, mean (SD; n)	2.7 (2.3; 224)
ISM-SAF diarrhea frequency, mean (SD; n)	1.3 (1.6; 224)
ISM-SAF diarrhea frequency in patients with ≥1 daily diarrhea episode, mean (SD; n)	2.4 (1.6; 105)
Prior cytoreductive therapy, n (%)[a]	29 (13)
Prior TKI therapy, n (%)	17 (8)
Number of BSC treatments, median (range)	3 (1–7)
BSC use at baseline, n (%)[b]	224 (99)
H1 antihistamines	221 (98)
H2 antihistamines	153 (68)
Leukotriene receptor antagonists	94 (42)
Cromolyn sodium	74 (33)
Proton pump inhibitors	43 (19)
Corticosteroids	26 (12)
Anti-IgE antibody (omalizumab)	29 (13)

[a]Cytoreductive therapies included dasatinib, imatinib, masitinib, nilotinib, midostaurin, brentuximab vedotin, cladribine, hydroxyurea, rapamycin, and interferon alfa. Includes treatments received by patients at baseline; patients may have received BSC treatments previously that had been discontinued at the time of enrollment/baseline. [b]All patients had ≥2 BSC prior to or at screening.
GI, gastrointestinal; IgE, immunoglobulin E; SD, standard deviation; TKI, tyrosine kinase inhibitor.

Figure 3. Median percent change from baseline in ISM-SAF GI individual symptom scores for avapritinib 25 mg QD

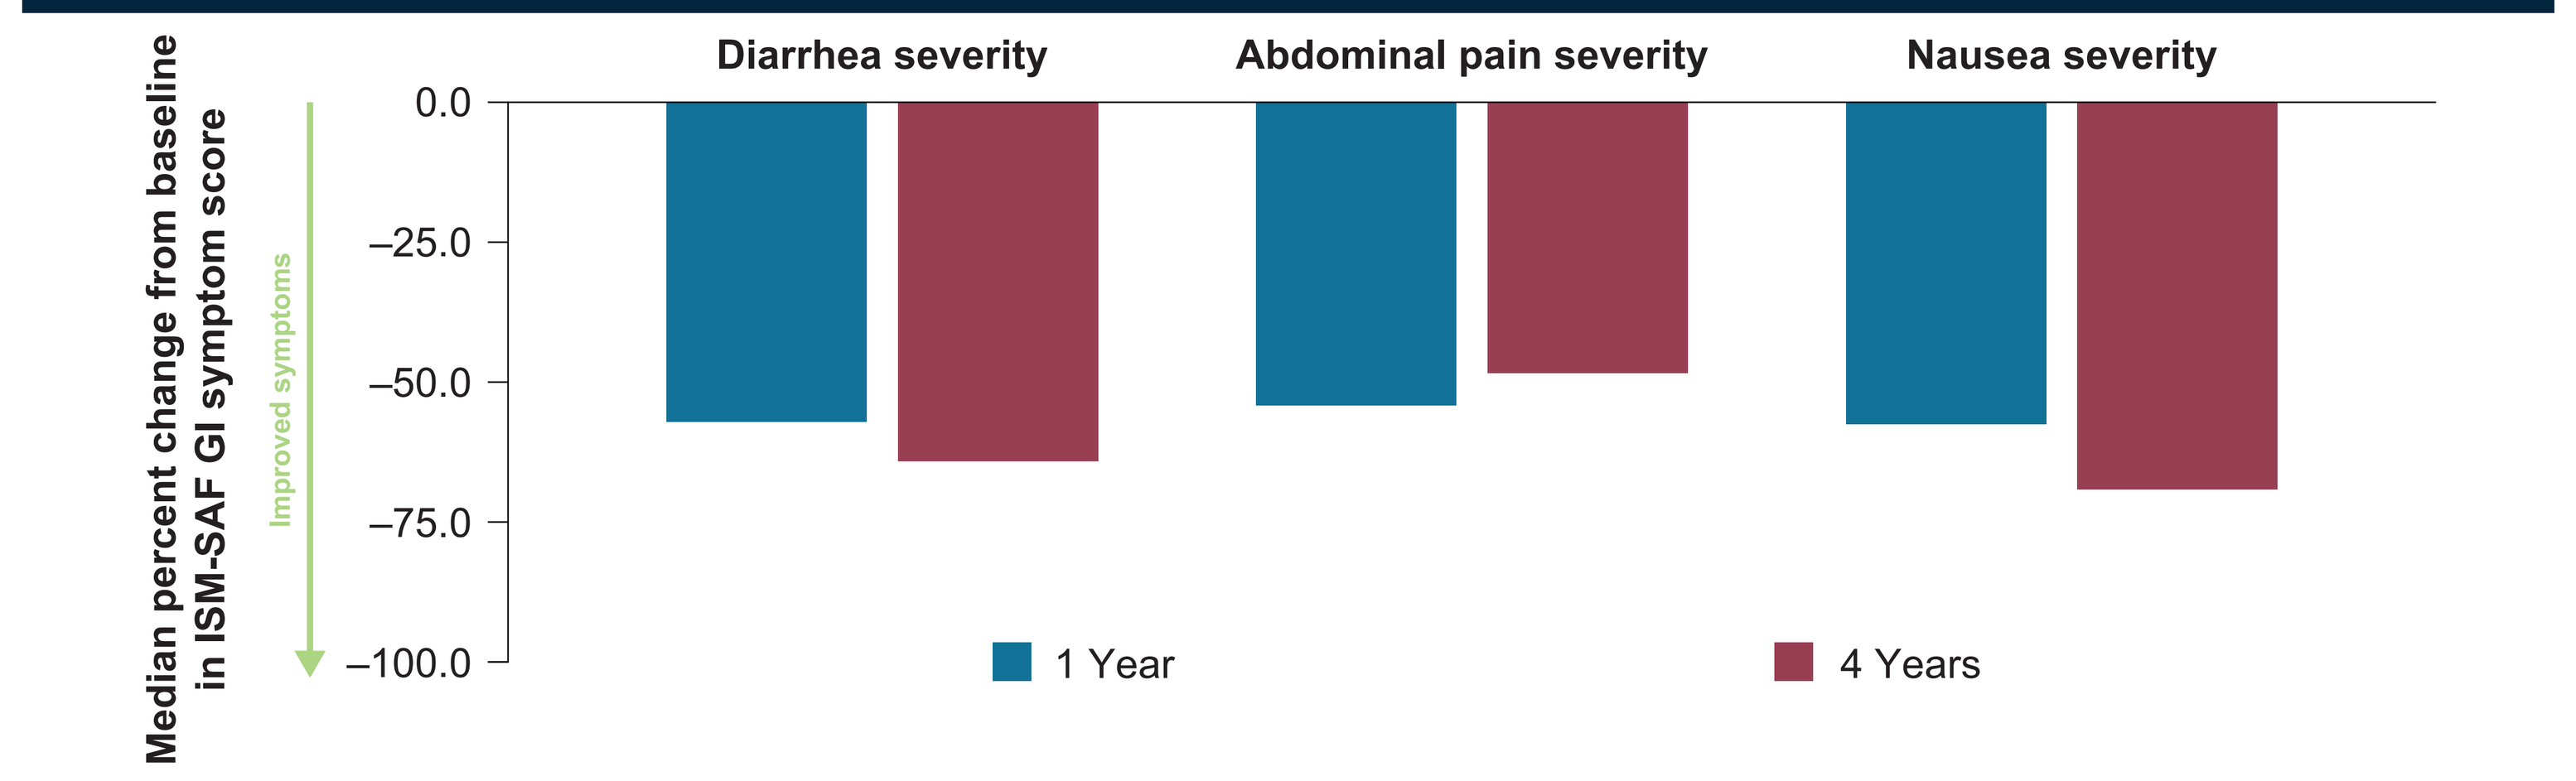

Diarrhea severity
Abdominal pain severity
Nausea severity
Median percent change from baseline in ISM-SAF GI symptom score
Improved symptoms
0.0
–25.0
–50.0
–75.0
–100.0
1 Year
4 Years

- All avapritinib-treated patients experienced durable improvements in ISM-SAF GI individual symptom scores (**Figure 3**)
 - Median percent change from baseline in ISM-SAF diarrhea severity was –57.1% at 1 year (n=167) and –64.2% at 4 years (n=74) in patients with diarrhea at baseline
 - Median percent change from baseline in ISM-SAF abdominal pain severity was –54.1% (n=184) and –48.4% (n=80) at 1 year and 4 years, respectively
 - For ISM-SAF nausea severity, the median percent change from baseline at the corresponding time points was –57.5% (n=169) and –69.1% (n=71)
- Median percent change from baseline in ISM-SAF diarrhea frequency in patients with ≥1 daily diarrhea episode at baseline was –57.8% at 1 year (n=86), and –65.6% at 4 years (n=44) (**Figure 4**)

Figure 4. Median percent change from baseline in ISM-SAF diarrhea frequency for avapritinib 25 mg QD in patients with a ≥1 daily diarrhea episode

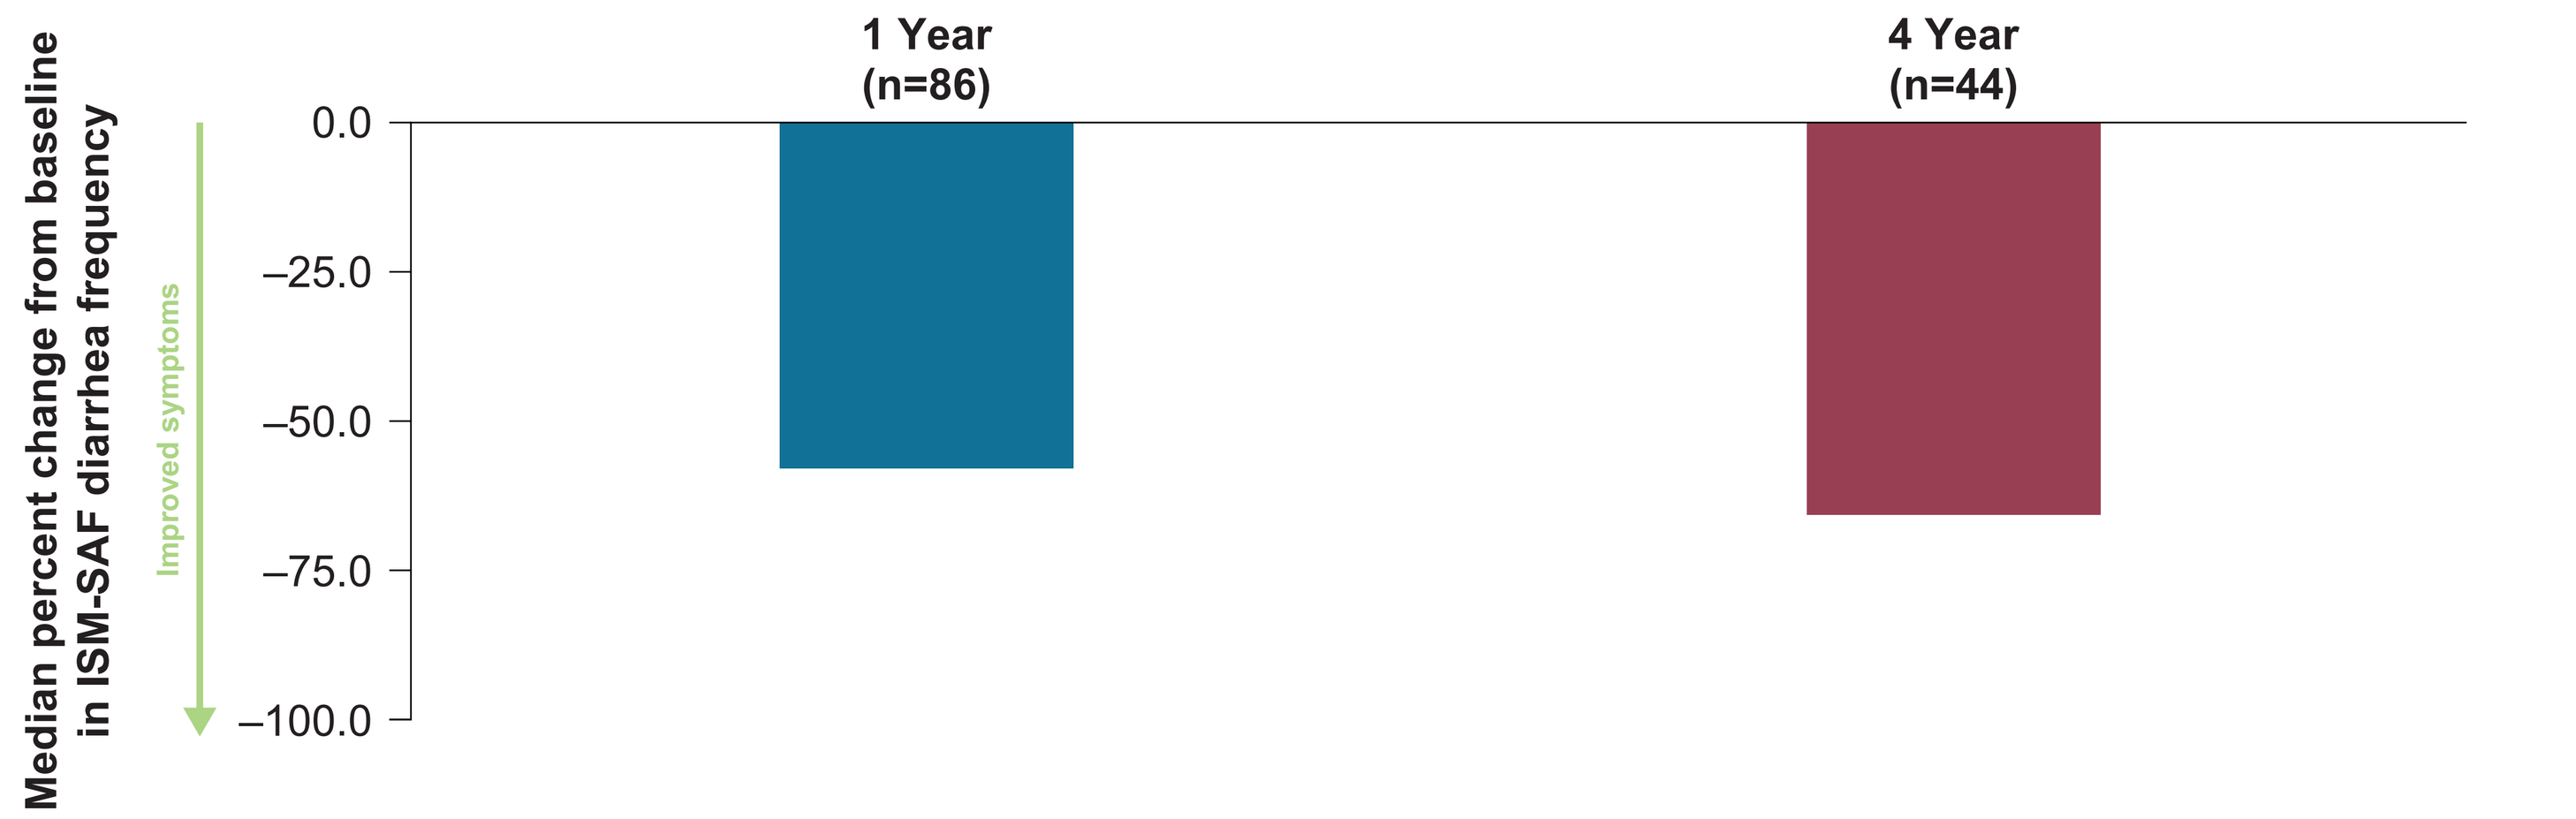

1 Year
(n=86)
4 Year
(n=44)
Median percent change from baseline in ISM-SAF diarrhea frequency
Improved symptoms
0.0
–25.0
–50.0
–75.0
–100.0

- After 3 years, 70 (32%) of the 221 patients using H1 antihistamines, 44 (28%) of the 153 patients using H2 antihistamines, and 27 (36%) of the 74 patients using cromolyn sodium at baseline had decreased use of these medications (**Figure 5**)

Figure 5. Percentage of patients with changes in BSC medications used to manage GI symptoms at 3 years

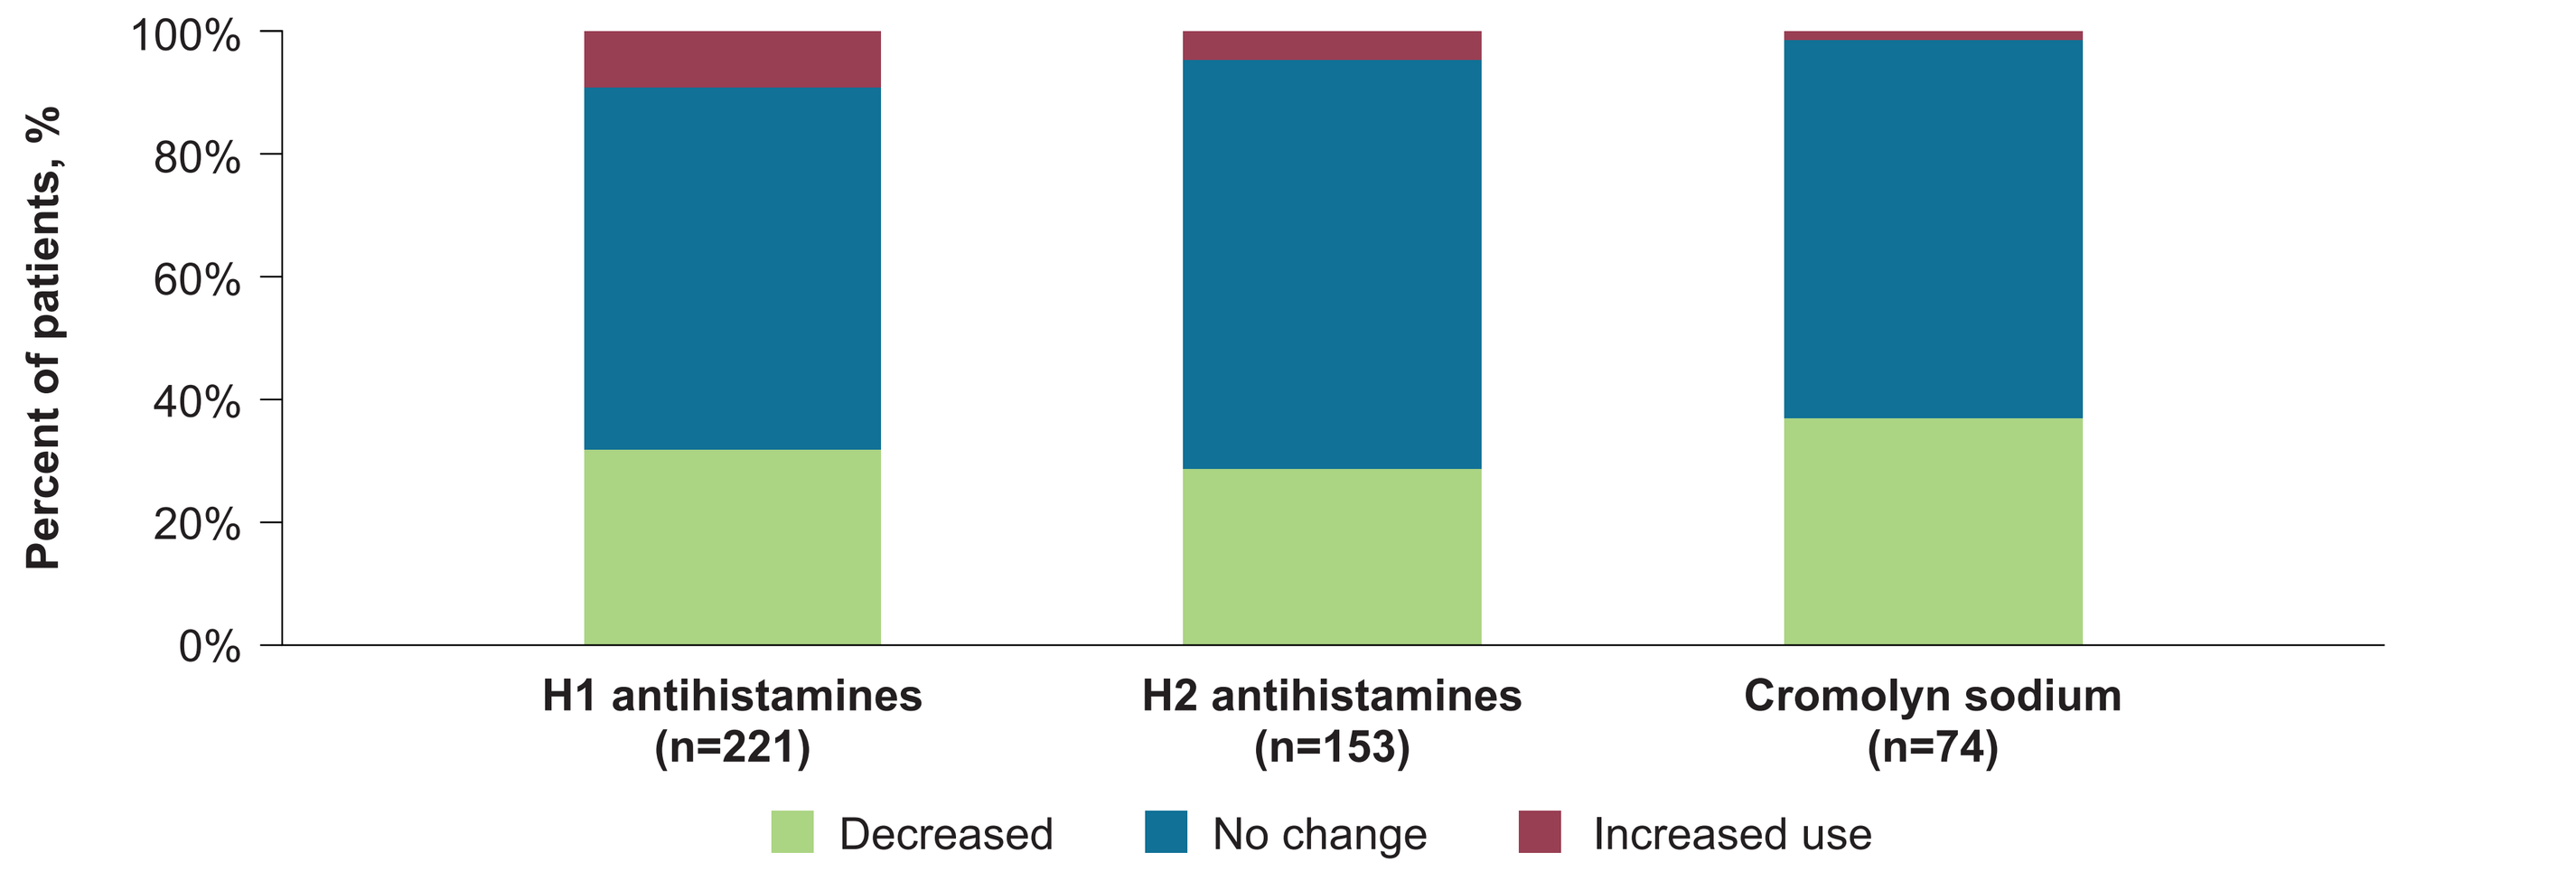

Percent of patients, %
100%
80%
60%
40%
20%
0%
H1 antihistamines
(n=221)
H2 antihistamines
(n=153)
Cromolyn sodium
(n=74)
Decreased
No change
Increased use

Table 2. Summary of AEs

	Part 2[a]		Parts 1, 2, 3 combined[b]
	Avapritinib 25 mg QD + BSC (n=141)	Placebo + BSC (n=71)	All patients who initiated avapritinib 25 mg QD + BSC (N=226)
Median length of follow-up (months)	5.6	5.6	46.5
Any AEs, n (%)	128 (91)	66 (93)	224 (99)
Any TRAEs, n (%)	77 (55)	32 (45)	168 (74)
Grade ≥3 AEs, n (%)	30 (21)	15 (21)	108 (48)[c]
Grade ≥3 TRAEs, n (%)	3 (2)	2 (3)	14 (6)
Serious adverse events	7 (5)	8 (11)	53 (24)
Serious TRAEs	0 (0)	0 (0)	3 (1)[d]
TRAEs leading to discontinuation, n (%)	2 (1)	1 (1)	6 (3)
Most common TRAEs (≥5% of patients), n (%)			
Peripheral edema	9 (6)	1 (1)	30 (13)
Periorbital edema	9 (6)	2 (3)	23 (10)
Headache	11 (8)	7 (10)	22 (10)
Nausea	9 (6)	6 (9)	18 (8)
Fatigue	6 (4)	2 (3)	17 (8)
Diarrhea	4 (3)	2 (3)	16 (7)
Alopecia	5 (4)	3 (4)	13 (6)

[a]Data cut: June 23, 2022. [b]Data cut: September 17, 2025. [c]One death (Grade 5 serious AE) occurred during the study and was unrelated to treatment; the patient had a medical history of anaphylaxis and atrial fibrillation, and the event was assessed as due to anaphylaxis in the context of atrial fibrillation. [d]Serious TRAEs included peripheral edema (1), gastric hemorrhage (1), and transient loss of vision (1). None of these events led to discontinuation. AEs, adverse events; BSC, best supportive care; TRAEs, treatment-related adverse events.

Safety

- The safety profile of avapritinib remained consistent with the previously reported placebo-controlled portion with longer-term median follow-up of 4 years (**Table 2**)
 - Grade ≥3 treatment-related adverse events (TRAEs) through 4 years remained low
 - Discontinuations due to TRAEs remained limited, occurring in six patients (3%)
 - The most frequently reported adverse events associated with treatment were edema events, with the majority being Grade 1

Conclusions

- Patients treated with avapritinib plus BSC showed meaningful improvements in GI symptoms of diarrhea, abdominal pain, and nausea that were durable through 4 years of follow-up
- Treatment with avapritinib reduced the use of GI-directed BSC medications in patients with ISM
- Avapritinib continues to be well tolerated with no new safety concerns after a median follow-up of 4 years

Acknowledgments

The authors thank the patients, their families, all other investigators, and all investigational site members involved in this study. This study was funded by Blueprint Medicines Corporation, a wholly-owned subsidiary of Sanofi. Medical writing support was provided by Francis Golder, BVSc, PhD, CMPP, and editorial support by Isobel Markham, MSc, of the Prime Group of Companies (Knutsford, UK) and was funded by Blueprint Medicines Corporation, a wholly-owned subsidiary of Sanofi. Responsibility for all opinions, conclusions, and data interpretation lies with the authors.

Scan the QR code to view our **Interactive Poster**

Conflicts of interest/disclosures

Dr Giannetti has received research funding from Blueprint Medicines Corporation, a wholly-owned subsidiary of Sanofi and consulting fees from Cogent Biosciences. Full disclosures for all authors are available upon request at medinfo@blueprintmedicines.com.

References

1. Pardanani A. *Am J Hematol*. 2023;98:1097–1116; 2. Kristensen T et al. *Am J Hematol*. 2014;89:493–498; 3. Ungerstedt J et al. *Cancers*. 2022;14:3942; 4. Garcia-Montero AC et al. *Blood*. 2006;108:2366–2372; 5. Mesa RA et al. *Cancer*. 2022;128:3691–3699; 6. Hermine O et al. *PLoS One*. 2008;3:e2266; 7. van Anrooij B et al. *Allergy*. 2016;71:1585–1593; 8. Hartmann K et al. *J Allergy Clin Immunol*. 2016;137:35–45; 9. Akin C et al. *J Allergy Clin Immunol*. 2022;149:1912–1918; 10. Pardanani A. *Blood*. 2013;121:3085–3094; 11. Evans EK et al. *Sci Transl Med*. 2017;9:eaao1690; 12. Ayvakit (avapritinib) Prescribing Information. Cambridge, MA: Blueprint Medicines Corporation; 2024; 13. Gotlib J et al. *NEJM Evidence*. 2023;2:EVIDoa2200339; 14. Shields AL et al. *Orphanet J Rare Dis*. 2023;18:69; 15. Taylor F et al. *Orphanet J Rare Dis*. 2021;16:414; 16. Padilla B et al. *Orphanet J Rare Dis*. 2021;16:434; 17. Ogbogu PU et al. AAAAI. 2023;131:S81

Presented at the American Academy of Allergy, Asthma & Immunology (AAAAI) 2026 Annual Meeting, February 27–March 2, 2026, in Philadelphia, PA , USA. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute.