PROSPECTOR: A Global, Prospective Study to Determine the Prevalence of the *KIT* D816V Mutation in Peripheral Blood From Patients With Evidence of Systemic Mast Cell Activation

Poster Number 161

Karin Hartmann,[1,2] Vito Sabato,[3] Jonathan A. Bernstein,[4] Cristina Bulai Livideanu,[5] Patricia Lugar,[6] Albert Hirdt,[7,8] Ivan Alvarez-Twose,[9] Aaron Zakharyan,[10] Tiffany Ashton,[10] Gerard Hoehn,[10] Cem Akin[11]

[1]Division of Allergy, Department of Dermatology, University Hospital Basel and University of Basel, Basel, Switzerland; [2]Department of Biomedicine, University Hospital Basel and University of Basel, Basel, Switzerland; [3]University of Antwerp and Antwerp University Hospital, Antwerp, Belgium; [4]Department of Internal Medicine, University of Cincinnati College of Medicine and Bernstein Clinical Research Center, Cincinnati, OH, USA; [5]CEREMAST CHU de Toulouse, Toulouse, France; [6]Department of Pediatrics, Duke University Medical Center, Durham, NC, USA; [7]Private Practice, New Paltz, NY, USA; [8]Clinical Mentor, NYITCOM, Old Westbury, NY, USA; [9]Institute of Mastocytosis Studies of Castilla-La Mancha, Spanish Reference Center of Mastocytosis, Toledo, Spain; [10]Blueprint Medicines Corporation, Cambridge, MA, USA; [11]University of Michigan, Ann Arbor, MI, USA.

Background

- Mast cell activation syndromes (MCAS) are a group of disorders characterized by systemic mast cell (MC) activation which can be caused by IgE-mediated and non-IgE-mediated triggers[1–3]
- Patients may experience debilitating symptoms associated with abnormal mast cell mediator release involving 2 or more organ systems (**Table 1**)[1,2,4]
- MCAS can be classified as clonal or non-clonal based on the presence or absence of the *KIT* D816V mutation and/or aberrant CD25 expression on MCs[2,5]
- Systemic mastocytosis (SM) is a rare clonal MC disease primarily driven by the *KIT* D816V mutation in approximately 95% of cases[6,7]
- Importantly, *KIT* D816V in bone marrow (BM) MCs can be detected in early stages of SM[5], however, the invasive nature of BM sampling required for diagnosis per World Health Organization (WHO)[8] criteria and limited molecular assay sensitivity can be barriers for diagnosis[7]
- The Spanish Network on Mastocytosis (Red Española de Mastocitosis [REMA]) developed a scoring system, that is not dependent on BM biopsies, for predicting MC clonality in patients presenting with MCAS symptoms[9,10]
- Further, high sensitivity real-time qPCR assays, such as allele-specific oligonucleotides-PCR[6] or droplet digital PCR[11], are now available to detect the presence of *KIT* D816V more reliably in peripheral blood (PB)[12]
- Testing for increased *TPSAB1* gene copy number (GCN) as an indicator of hereditary alpha-tryptasemia (HaT), which causes elevated serum tryptase, may also aid in the diagnosis of SM[13]
- Despite these efforts, the prevalence of clonal mast cell disease with evidence of systemic MCA is still unknown and warrants further investigation

Table 1. Summary of symptoms related to systemic mast cell activation

Organ system	Symptoms
Skin	Pruritus, urticaria, angioedema, and flushing
Gastrointestinal	Nausea, vomiting, diarrhea, and abdominal cramping
Cardiovascular	Hypotensive syncope or near syncope, and tachycardia
Respiratory/naso-ocular	Wheezing, conjunctival injection, and nasal congestion
Neurological	Difficulty concentrating, fatigue, headaches
Musculoskeletal	Bone pain, muscle pain, osteopenia

Study design and objectives

- PROSPECTOR (NCT04811365) is a global, multi-center screening study determining the prevalence of *KIT* D816V mutation in PB in patients with evidence of systemic MCA (**Figure 1**)
- The key eligibility criteria and key study endpoints are shown in **Table 2** and **Table 3**, respectively

Figure 1. PROSPECTOR study design

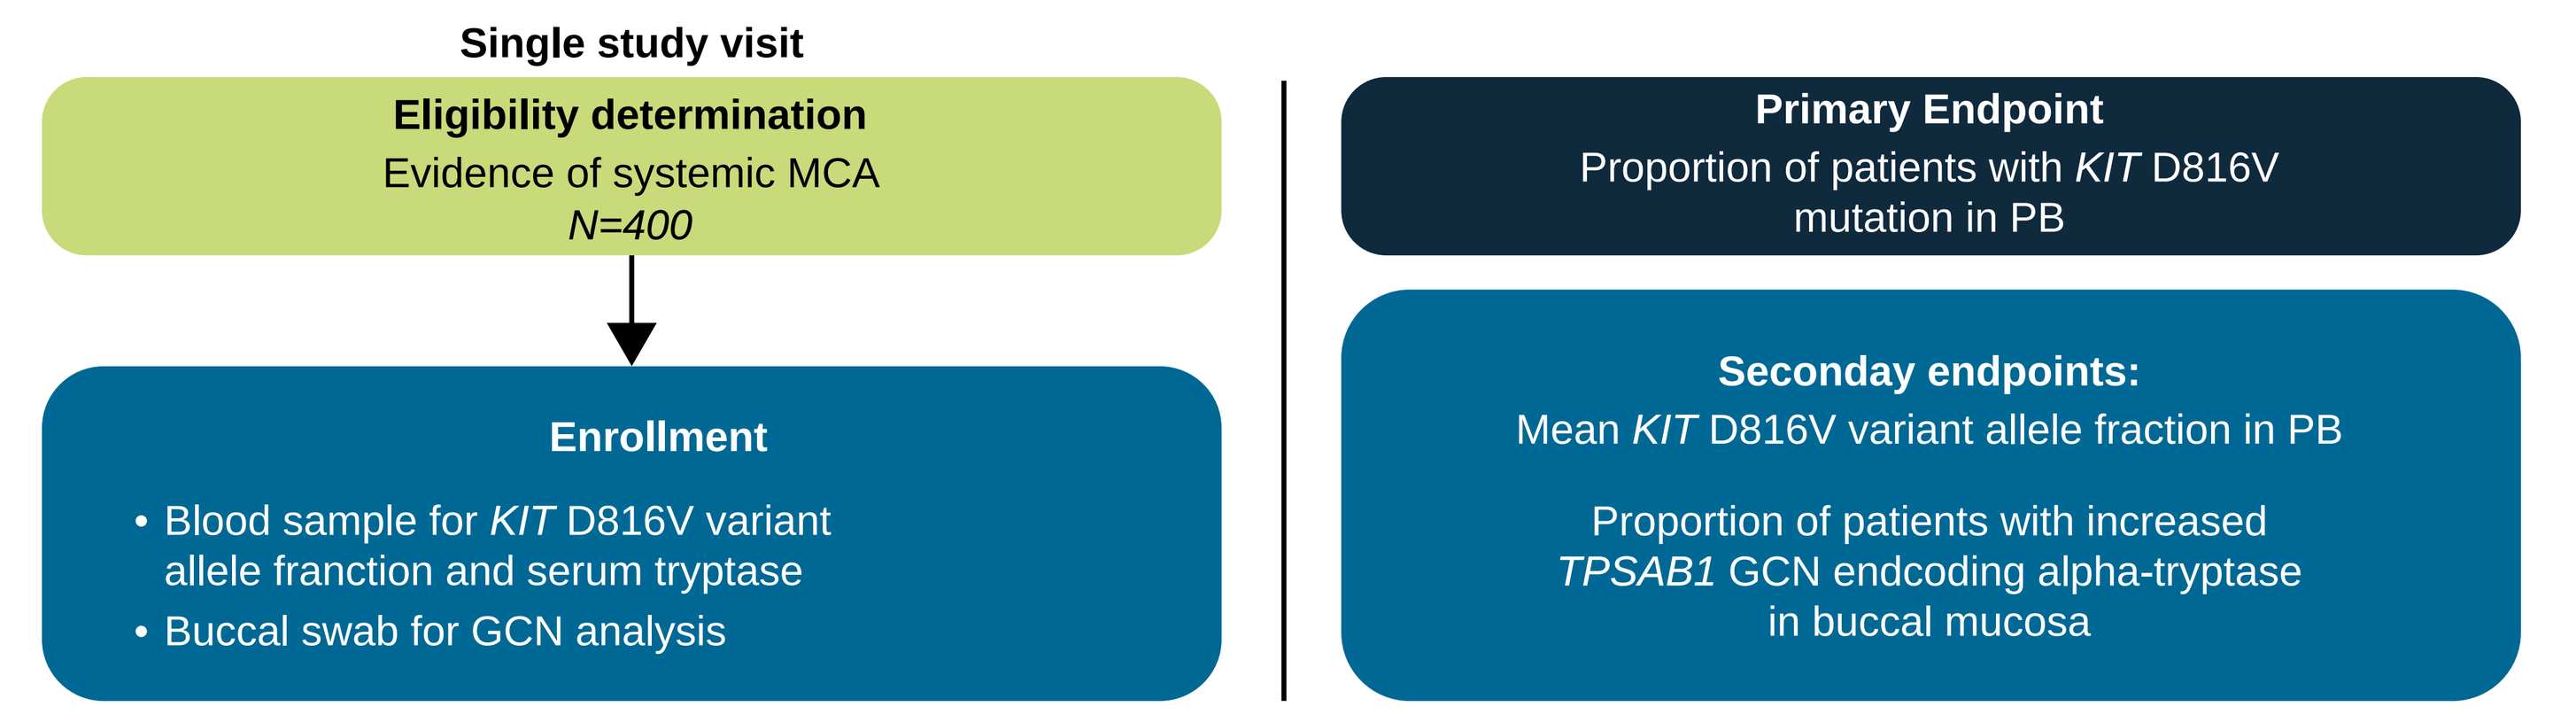
Single study visit
Eligibility determination
Evidence of systemic MCA
N=400
Enrollment
• Blood sample for KIT D816V variant allele franction and serum tryptase
• Buccal swab for GCN analysis
Primary Endpoint
Proportion of patients with KIT D816V mutation in PB
Seconday endpoints:
Mean KIT D816V variant allele fraction in PB
Proportion of patients with increased TPSAB1 GCN endcoding alpha-tryptase in buccal mucosa

GCN, gene copy number; MCA, mast cell activation; PB, peripheral blood; TPSAB1, tryptase Alpha/Beta 1.

Table 2. Key eligibility criteria

Inclusion criteria

- Aged ≥18 years
- Evidence of systemic mast cell activation, based on meeting ≥1 of 3 criteria:
 - Involvement of ≥2 organ systems (cardiovascular involvement necessary) and serum basal tryptase ≥8 ng/mL
 - Severe anaphylaxis from hymenoptera sting
 - Severe anaphylaxis with cardiovascular involvement and event-related tryptase elevation fitting the formula 20% of baseline plus 2 ng/mL in ≥1 event

Exclusion criteria

- Patients with known diagnoses of mastocytosis, including:
 - Mastocytosis in the skin
 - Indolent systemic mastocytosis
 - Smoldering systemic mastocytosis
 - Systemic mastocytosis with associated hematologic neoplasm
 - Aggressive systemic mastocytosis
 - Mast cell leukemia
 - Mast cell sarcoma

Table 3. Key study endpoints

Primary endpoint	Secondary endpoints
• The prevalence of *KIT* D816V mutation in PB by droplet digital PCR defined as the proportion of patients with evidence of systemic MCA and the *KIT* D816V mutation in PB	• *KIT* D816V variant allele fraction in PB • The prevalence of HaT defined as the proportion of patients with an increased *TPSAB1* GCN encoding alpha-tryptase on a single allele in buccal mucosa • The relationship between *KIT* D816V in PB and REMA score, other MCA clinical parameters or HaT diagnosis

HaT, hereditary alpha-tryptasemia; REMA, Red Española de Mastocitosis.

Enrollment and status

- Enrollment in PROSPECTOR is ongoing with 24 planned centers participating in the United States, the United Kingdom, and Europe (**Figure 2**)
- As of August 15, 2022, there were 105 patients enrolled and evaluated for *KIT* D816V mutation (**Table 4**) in PB using droplet digital PCR
 - 3/105 patients (2.9%) had detectable *KIT* D816V mutation
 - 15/105 patients (14.3%) had tryptase levels ≥20 ng/ml and 23.8% (25/105) of patients had increased *TPSAB1* GCN

Figure 2. Countries with active enrollment sites

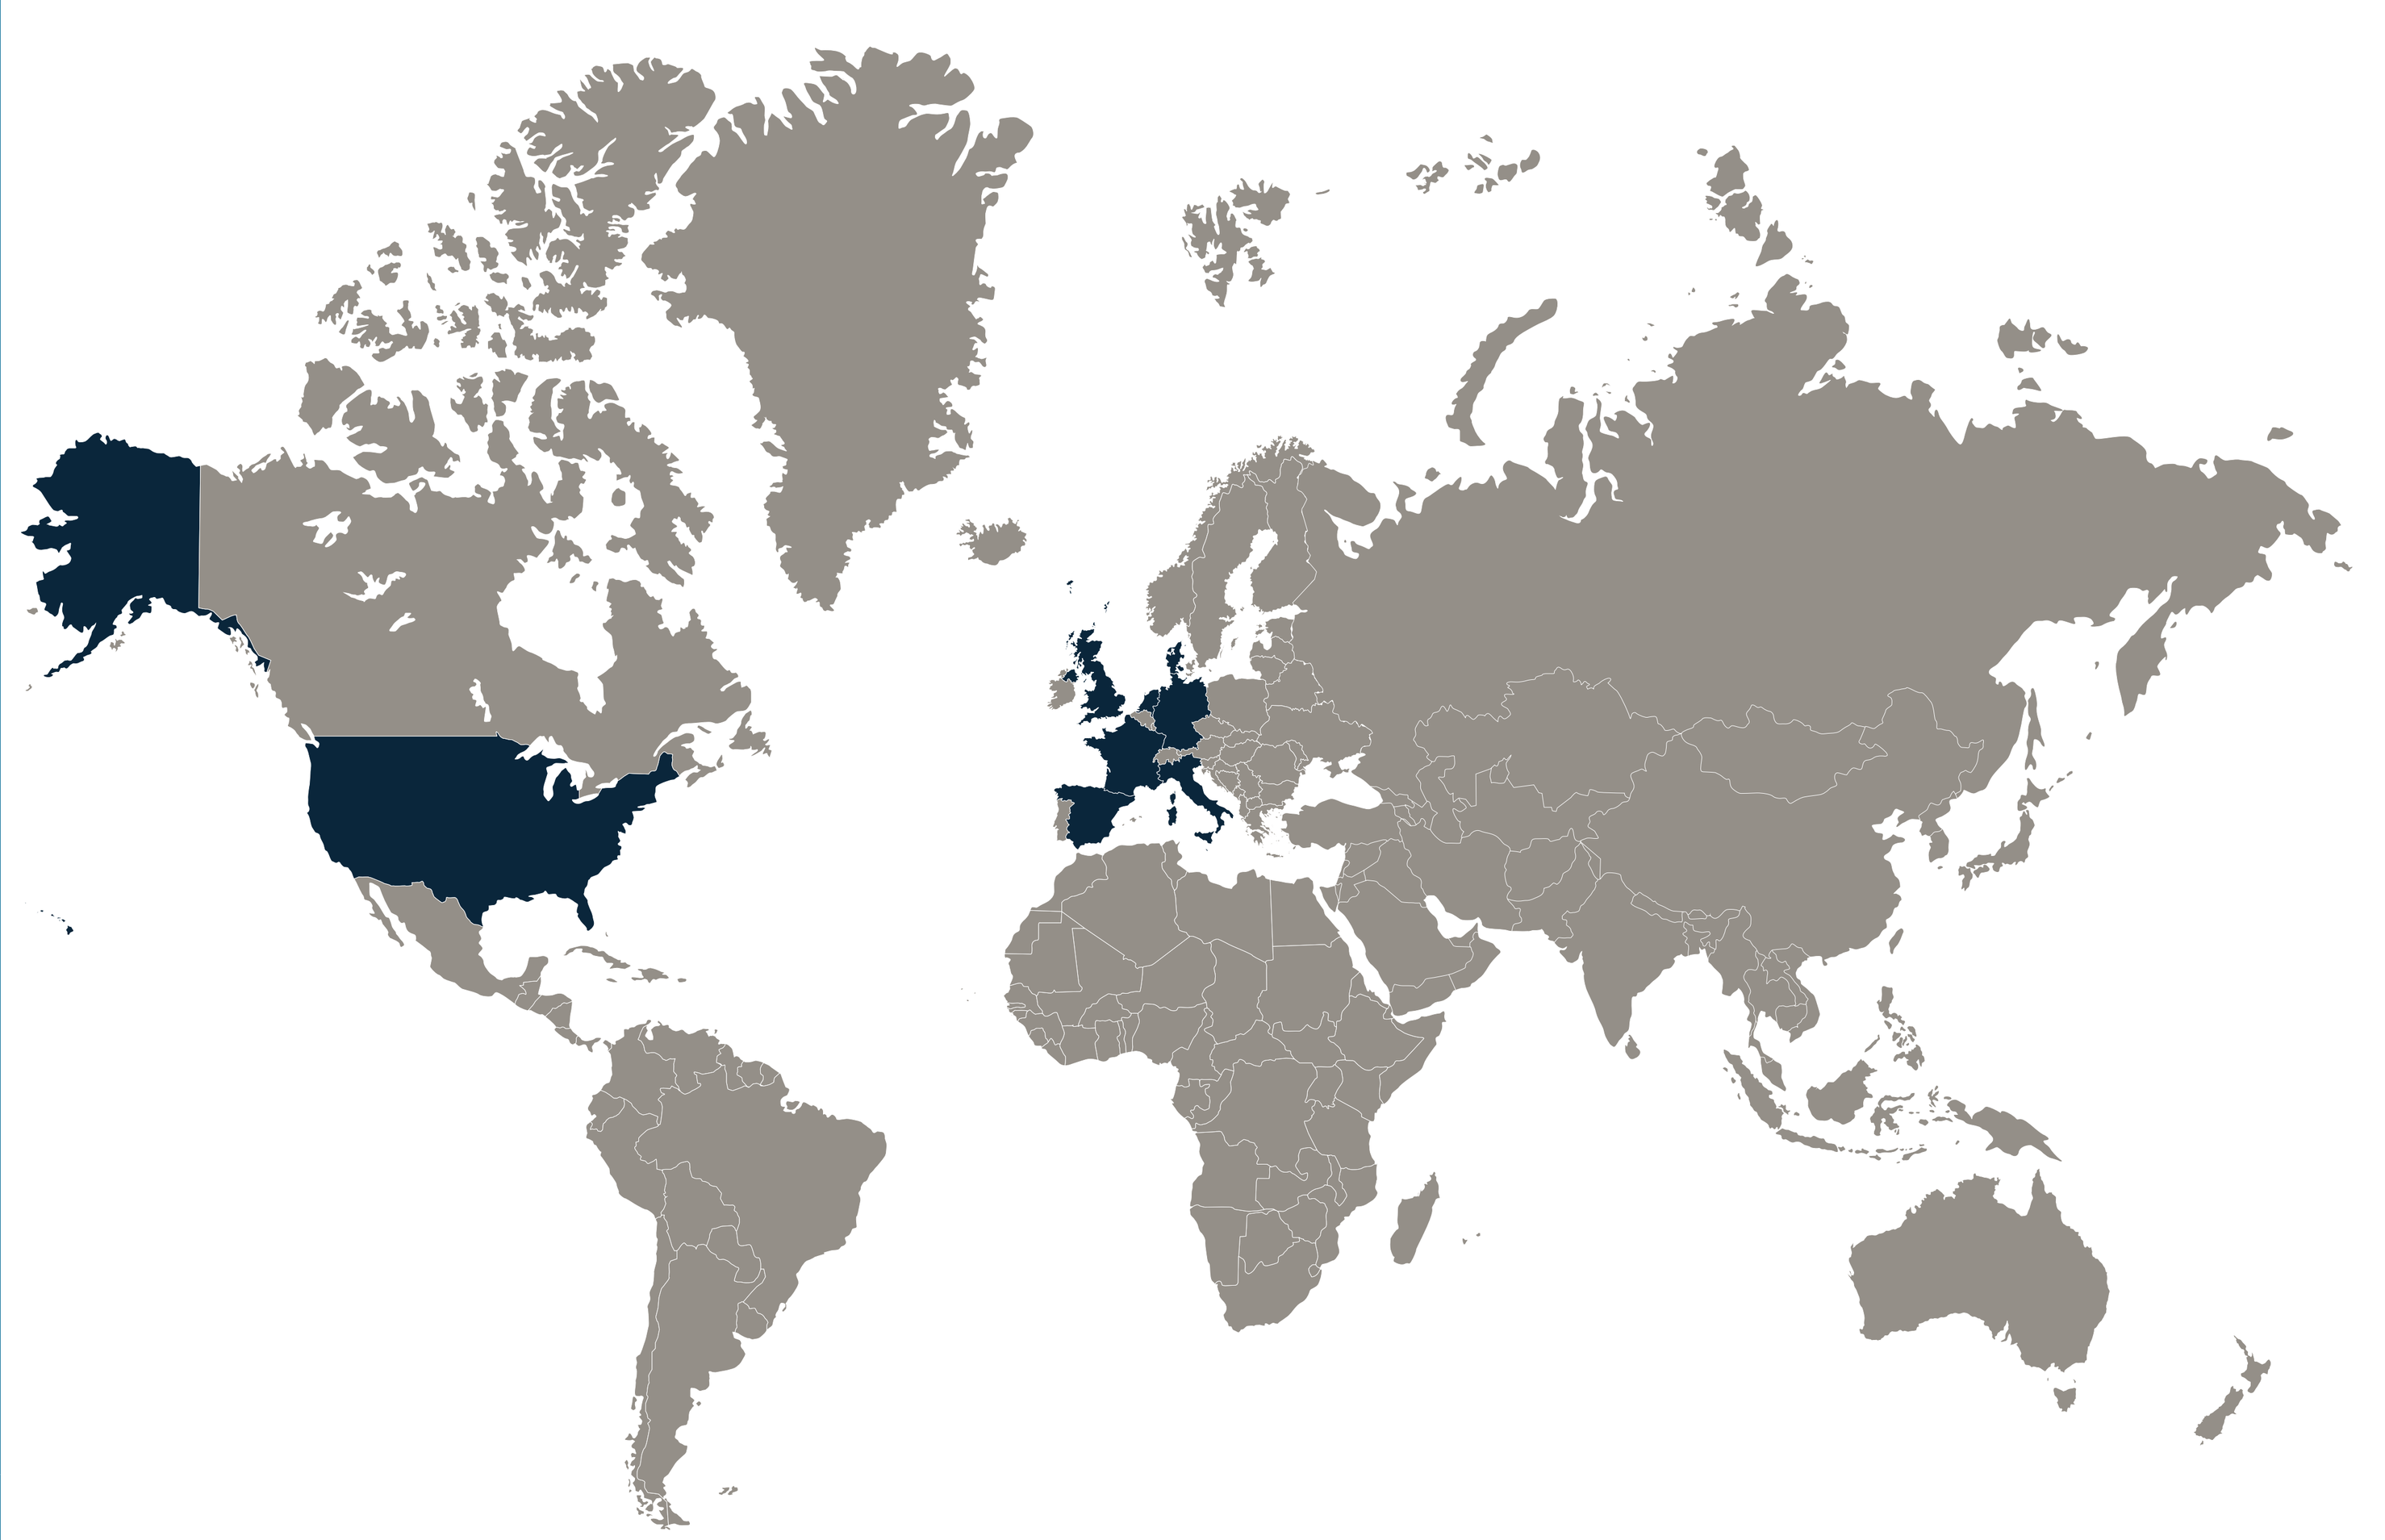

Table 4. Preliminary baseline demographics

Characteristic	N=105		
Age, mean (range)	55.7 (19–86)		
Gender, n (%)			
Female	59 (56.2)		
Male	46 (43.8)		
Ethnicity, n (%)			
Hispanic or Latino	2 (1.9)		
Not Hispanic or Latino	64 (61.0)		
Not reported	36 (34.3)		
Unknown	3 (2.9)		
KIT D816V positive, n (%)	3 (2.9)[a]		
Increased *TPSAB1* GCN, n (%)	25 (23.8)		
Serum tryptase (ng/mL), n (%)	All patients	*KIT* D816V positive	Increased *TPSAB1* GCN
0–<8	42 (40.0)	1 (1.0)	0 (0)
8–<20	44 (41.9)	2 (1.9)	16 (15.2)
≥20	15 (14.3)	0 (0)	8 (7.6)
Unknown	4 (3.8)	0 (0)	1 (1.0)
REMA score, mean (range)	-1.3 (-4–7)		
Inclusion criteria, n (%)			
Involvement of ≥2 organ systems (cardiovascular involvement necessary) and serum basal tryptase ≥8 ng/mL	45 (42.9)		
Severe anaphylaxis from hymenoptera sting	44 (41.9)		
Severe anaphylaxis with cardiovascular involvement and event-related tryptase elevation fitting the formula 20% of baseline plus 2 ng/mL in ≥1 event	16 (15.2)		

[a]The number of *KIT* D816V positive patients was initially reported to be 7 (6.7%) but subsequently was amended to 3 (2.9%).

Summary

- PROSPECTOR is a multicenter prospective screening study that aims to determine the prevalence of clonal mast cell disease in patients with evidence of systemic mast cell activation and the study is currently ongoing
- The frequency of patients positive for *KIT* D816V in this population suggests patients with MCA symptoms may have higher risk of clonal mast cell disease such as SM, which has a general prevalence of 1:10,000[14]
- Despite the high sensitivity of droplet digital PCR, the detection of low *KIT* D816V mutant allele frequency levels (<0.02%) in PB can be problematic; notably, some patients had increased serum tryptase without HaT, but were negative for *KIT* D816V
- Enrichment strategies and/or higher sensitive assays may be required to more accurately detect the *KIT* D816V mutation in patients with clonal mast cell disorders
- For more information, visit

https://clinicaltrials.gov/ct2/show/NCT04811365

- To learn more about our clinical trials, visit blueprintclinicaltrials.com or contact us in the USA at medinfo@blueprintmedicines.com or 1-888-BLU-PRNT (1-888-258-7768), and in Europe at medinfoeurope@blueprintmedicines.com or +31 85 064 4001

References

1. Jackson CW et al. *Int J Mol Sci*. 2021;22(20). 2. Akin C et al. *J Allergy Clin Immunol*. 2010;126(6):1099–1104 e1094. 3. González-de-Olano D et al. *Front Immunol*. 2017;8:792. 4. Pardanani A. *Am J Hematol*. 2021;96(4):508–525. 5. Valent P et al. *Int Arch Allergy Immunol*. 2012;157(3):215–225. 6. Garcia-Montero AC et al. *Blood*. 2006;108:2366–2372. 7. Kristensen T et al. *J Mol Diagn*. 2011;13:180–188. 8. Valent P et al. *Blood*. 2017:16;129(11):1420–1427. 9. Alvarez-Twose I et al. J Allergy Clin Immunol. 2010;125(6):1269–1278.e2. 10. Alvarez-Twose I et al. *Int Arch Allergy Immunol*. 2012;157(3):275–280. 11. Greiner G et al. *Clin Chem*. 2018;64(3):547–555. 12. Kristensen T et al. *Eur J Haematol*. 2016;96(4):381–388. 13. Lyons JJ et al. *J Allergy Clin Immunol*. 2021;147(2):622–632. 14. Cohen SS et al. *Br J Haematol*. 2014;166:521–528.

Acknowledgements

Medical writing support was provided by Danielle Scheunemann, MS, and editorial support was provided by Travis Taylor, BA, all of Paragon, Knutsford, UK, supported by Blueprint Medicines Corporation, Cambridge, MA, according to Good Publication Practice guidelines.

Disclosures

This research was funded by Blueprint Medicines Corporation. Blueprint Medicines Corporation reviewed and provided feedback on the poster. The authors had full editorial control of the poster and provided their final approval of all content. Dr Hartmann has received research support from Thermo Fisher, and advisory board or speaker fees and travel support from ALK-Abelló, Allergopharma, Blueprint Medicines Corporation, Deciphera, Leo, Menarini, Novartis, Takeda, and Thermo Fisher. Full disclosures for all authors are available upon request at medinfo@blueprintmedicines.com.

SCAN ME

Presented at AAAAI Annual Meeting, San Antonio, TX, February 24–27, 2023. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute.