Duration of Treatment and Reduction in Serum Tryptase Levels in Patients with Advanced Systemic Mastocytosis Treated with Avapritinib versus Best Available Therapy

Abstract P1015

Andreas Reiter, MD[1]; Jason Gotlib, MD, MS[2]; Iván Álvarez-Twose, MD, PhD[3]; Deepti H. Radia, MD[4]; Johannes Luebke, MD[1]; Priyanka J. Bobbili, ScD, MS[5]; Aolin Wang, PhD[5]; Chelsea Norregaard, MPH[6]; Saša Dimitrijević, PhD[7]; Erin Sullivan, PhD, MPH[6]; Melinda Louie-Gao, PhD[6]; Juliana Schwaab, MD[1]; Ilene A. Galinsky, MSN, ANP-C[8]; Cecelia Perkins, MPH[2]; Wolfgang R. Sperr, MD[9]; Priya Sriskandarajah, MBBS, MRCP, PhD[4]; Andi Chin[5]; Selvam R. Sendhil[5]; Mei Sheng Duh, ScD, MPH[5]; Peter Valent, MD[9]; Daniel J. DeAngelo, MD, PhD[8]

[1]Department of Hematology and Oncology, University Hospital Mannheim, Mannheim, Germany; [2]Stanford Cancer Institute/Stanford University School of Medicine, Stanford, California, United States; [3]Institute of Mastocytosis Studies of Castilla La Mancha (CLMast) — Spanish Reference Center (CSUR) for Mastocytosis and CIBERONC, Virgen del Valle Hospital, Toledo, Spain; [4]Guy's & St Thomas' NHS Foundation Trust, of Guy's Hospital, London, United Kingdom; [5]Analysis Group, Inc., Boston, Massachusetts, United States; [6]Blueprint Medicines Corporation, Cambridge, Massachusetts, United States; [7]Blueprint Medicines Corporation, Zug, Switzerland; [8]Department of Medical Oncology, Dana-Farber Cancer Institute, Boston, Massachusetts, United States; [9]Department of Internal Medicine I, Division of Hematology and Hemostaseology, and Ludwig Boltzmann Institute for Hematology and Oncology, Medical University of Vienna, Vienna, Austria

Background

- Advanced systemic mastocytosis is a rare and heterogenous disease, often characterized by the buildup of mast cells in various organs and tissues[1,2]
- The World Health Organization (WHO) classifies AdvSM in three subtypes: aggressive systemic mastocytosis (ASM), SM with an associated hematologic neoplasm (SM-AHN), and mast cell leukemia (MCL)[3]
- AdvSM patients frequently have poor prognosis[4] and median durations of therapy (DOT) for standards of care have ranged from 7.9−9 months in real-world practice[5,6]
- Recent research has shown that elevated serum tryptase level (≥125 ng/mL) is of prognostic significance for AdvSM patients[7]
- Avapritinib, a selective inhibitor of *KIT* D816V, has been approved for AdvSM patients in the United States (US)[8] and Europe (for patients treated with prior systemic therapy)[9] based on results from the Phase 1 EXPLORER (NCT02561988) and Phase 2 PATHFINDER (NCT03580655) single-arm trials[10,11]
- As of yet, no randomized controlled trials (RCTs) have been conducted comparing clinical outcomes for avapritinib versus best available therapy (BAT) for AdvSM

Aim

- This study (NCT04695431) compared DOT and maximum reduction in serum tryptase levels between patients treated with avapritinib versus BAT,[12] with this specific analysis including avapritinib patients who received a starting dose of ≤200mg once daily

Methods

Data sources

- Clinical trial data (*avapritinib cohort*)
 - Data from patients treated with avapritinib in the safety populations of the EXPLORER and PATHFINDER trials was used (data cut-off: April 20, 2021; data on file, Blueprint Medicines Corporation)
- Real-world data (*BAT cohort*)
 - A global, observational, retrospective chart review study was conducted at 6 study sites (4 European, 2 US) to identify and collect data from AdvSM patients who received BAT
 - De-identified data from eligible patients were abstracted from patient health records into standardized electronic case report forms from March 26, 2021 to October 4, 2021

Sample selection

- Real-world patients treated with BAT were identified based on the following key inclusion and exclusion criteria, similar to those from EXPLORER and PATHFINDER
- **Inclusion criteria:**
 - Adults (aged ≥18 years) with a diagnosis of AdvSM and documented subtype
 - Received ≥1 line of systemic therapy (not necessarily as first line) at a participating site on or after January 1, 2009
 - If a patient received multiple lines of therapy at a participating site, data on all available therapies were collected and analyzed
 - The date of initiation of each line of therapy at the participating site was defined as the index date
- **Exclusion criteria:**
 - History of another primary malignancy that was diagnosed or required therapy within 3 years before the index date, except for completely resected basal cell and squamous cell skin cancer, curatively treated localized prostate cancer, and completely resected carcinoma *in situ* in any site
 - Received avapritinib as the first therapy for AdvSM at a participating site

Study endpoints

- DOT was defined as the time interval between initiation of each line of therapy to discontinuation for any reason
- Maximum reduction in serum tryptase was defined as the maximum percentage change in serum tryptase levels during each line of therapy

Statistical analysis

- DOT was compared between cohorts using a Cox proportional hazards model, and maximum reduction in serum tryptase levels was compared using a generalized estimating equation linear model
- Both models were weighted by inverse probability of treatment weights (IPTW) to adjust for confounding by key variables, and were further adjusted for variables that remained unbalanced after weighting
- Subgroup analyses were conducted in patients who (1) received ≥1 prior line of therapy (2L+), and (2) a 200mg starting dose of avapritinib

Results

Baseline population characteristics

- The analysis included 136 avapritinib patients and 137 BAT patients, the latter contributing 213 lines of therapy
- Median (range) age for avapritinib and BAT patients was 68.0 (31.0−88.0) and 69.0 (20.9−87.5) years; 58.8% of patients were male in the avapritinib cohort versus 65.7% in the BAT cohort
- There were 68 (50.0%) lines of therapy contributed from European patients in the avapritinib cohort versus 179 (84.0%) in the BAT cohort; the remaining patients were treated at North American sites (**Table 1**)
- Serum tryptase levels ≥125 ng/mL were identified in 102 (75.0%) and 137 (64.3%) lines of therapy contributed from avapritinib and BAT cohorts (P<0.05)
- Patients in the avapritinib cohort were most frequently pre-treated with tyrosine kinase inhibitors (TKI) (55.9%), followed by cytoreductive therapies (16.2%) (**Table 2**)
- Patients in the BAT cohort were most frequently pretreated with cytoreductive therapies (26.8%), followed by TKIs (22.5%)

Table 1. Baseline demographics and clinical characteristics

Baseline characteristics, unweighted sample[1,2]	Avapritinib cohort	BAT cohort	*P* value[3]
Number of unique patients	**N = 136**	**N = 137**	
Number of lines of therapy	**N = 136**	**N = 213**	
Demographic characteristics			
Age (years)			0.710
Mean (SD)	66.8 (10.6)	65.8 (11.8)	--
Median (min, max)	68.0 (31.0, 88.0)	69.0 (20.9, 87.5)	--
Sex, n (%)			
Female	56 (41.2%)	73 (34.3%)	0.234
Male	80 (58.8%)	140 (65.7%)	0.234
Region, n (%)			
North America	68 (50.0%)	34 (16.0%)	<0.001*
Europe	68 (50.0%)	179 (84.0%)	<0.001*
Medical history			
Performance status			
ECOG			0.195
n (%)	136 (100.0%)	213 (100.0%)	--
Mean (SD)	1.1 (0.8)	1.0 (0.7)	--
Median (min, max)	1.0 (0.0, 3.0)	1.0 (0.0, 3.0)	--
ECOG category, n (%)			
0	28 (20.6%)	50 (23.5%)	0.618
1	73 (53.7%)	120 (56.3%)	0.706
≥2	35 (25.7%)	43 (20.2%)	0.280
Anemia, n (%)	74 (54.4%)	119 (55.9%)	0.876
Thrombocytopenia, n (%)	47 (34.6%)	112 (52.6%)	0.001*
Disease characteristics			
AdvSM subtype diagnosis, n (%)			
SM-AHN	91 (66.9%)	118 (55.4%)	0.043*
ASM	23 (16.9%)	62 (29.1%)	0.014*
MCL	22 (16.2%)	33 (15.5%)	0.984
Skin involvement			
Any skin involvement, n (%)	47 (34.6%)	68 (31.9%)	0.694
Leukocyte count			
≥16 x 10^9/L, n (%)	22 (16.2%)	52 (24.4%)	0.089
Serum tryptase (ng/mL)			
≥125 ng/mL, n (%)	102 (75.0%)	137 (64.3%)	0.048*
KIT mutation[2]			
Patients tested, n (%)	130 (95.6%)	136 (99.3%)	0.066
KIT D816V positive, n (%)	122 (93.8%)	124 (91.2%)	0.553
SRSF2/ASXL1/RUNX1 (S/A/R) mutation panel			
Patients that were tested for at least one mutation, n (%)	136 (100.0%)	105 (76.6%)	<0.001*
Number of mutated genes in *S/A/R* panel, n (%)			
0	73 (53.7%)	41 (39.0%)	0.034*
1	40 (29.4%)	42 (40.0%)	0.113
≥2	23 (16.9%)	22 (21.0%)	0.528

**P<0.05*

Abbreviations: AdvSM: advanced systemic mastocytosis; ASM: aggressive systemic mastocytosis; DOT: duration of treatment; ECOG: Eastern Cooperative Oncology Group; max: maximum; MCL: mast cell leukemia; min: minimum; S/A/R: SRSF2/ASXL1/RUNX1; SD: standard deviation; SM-AHN: systemic mastocytosis with an associated hematologic neoplasm.

Notes:

[1] The baseline period was defined as 8 weeks leading up to the index date for the avapritinib cohort and the 12 weeks leading up to the index date for the BAT cohort. Characteristics are reported for patients included in the DOT analysis.

[2] Descriptive statistics are reported at the line of therapy level for all variables except *KIT* and S/A/R mutations, which are reported at the patient level, since each patient in the BAT cohort could contribute more than one line of therapy to the analysis.

[3] Comparisons between cohorts were conducted using the Wilcoxon rank-sum test for continuous variables and chi-squared test for categorical variables. For categorical variables with expected counts <5, Fisher's exact tests were used instead of Chi-squared.

Table 2. Prior systemic therapy used to treat AdvSM patients

Prior systemic therapy	Avapritinib cohort	BAT cohort	*P* value[1]
Number of unique patients	**N = 136**	**N = 137**	
Number of lines of therapy	**N = 136**	**N = 213**	
Number with prior systemic therapy, n (%)	85 (62.5%)	97 (45.5%)	0.003*
Number of prior lines of systemic therapy received, n (%)			<0.001*
Mean (SD)	1.0 (1.1)	0.1 (0.3)	--
Median (min, max)	1.0 (0.0, 6.0)	0.0 (0.0, 2.0)	--
0	51 (37.5%)	116 (54.5%)	0.003*
1	54 (39.7%)	63 (29.6%)	0.066
2	20 (14.7%)	24 (11.3%)	0.436
≥3	11 (8.1%)	10 (4.7%)	0.285
Prior treatments received, n (%)			
TKI therapy	76 (55.9%)	48 (22.5%)	<0.001*
Cytoreductive therapy	22 (16.2%)	57 (26.8%)	0.030*
Biologic therapy	18 (13.2%)	26 (12.2%)	0.907

**P<0.05*

Abbreviations: AdvSM: advanced systemic mastocytosis; max: maximum; min: minimum; SD: standard deviation; TKI: tyrosine kinase inhibitor.

Notes:

[1] Comparisons between cohorts were conducted using the Wilcoxon rank-sum test for continuous variables and chi-squared test for categorical variables. For categorical variables with expected counts <5, Fisher's exact tests were used instead of Chi-squared.

Duration of treatment

- Among BAT patients with agent-level data available (188/213 lines of therapy), most common treatments were midostaurin (52.7%) and cladribine (23.9%) (**Table 3**)
- In 1L, BAT patients were most commonly treated with tyrosine kinase inhibitors; in 2L and 3L+, BAT patients were most commonly treated with cytoreductive agents
- In IPTW-weighted Cox analysis, DOT was significantly longer for avapritinib versus BAT (hazard ratio [95% CI]: 0.35 [0.23, 0.51]; P<0.001) (**Table 4**)
- Results were similar in subgroups (**Table 4**)

Table 3. Summary of best available therapies received by real-world patients

	Overall[1]	1L	2L	3L+
Number of unique patients	**N = 137**	**N = 134**	**N = 52**	**N = 17**
Number of lines of therapy	**N = 213**	**N = 134**	**N = 52**	**N = 27**
Total number of lines of therapy contributed by patient				
Mean (SD)	1.6 (0.9)	1.6 (0.9)	1.0 (-)	1.0 (0.0)
Median (min, max)	1.0 (1.0, 7.0)	1.0 (1.0, 7.0)	1.0 (1.0, 1.0)	1.0 (1.0, 1.0)
Number of lines of therapy contributed, n (%)				
1	86 (62.8%)	83 (61.9%)	1 (100.0%)	2 (100.0%)
2	36 (26.3%)	36 (26.9%)	0 (0.0%)	0 (0.0%)
≥3	15 (10.9%)	15 (11.2%)	0 (0.0%)	0 (0.0%)
Year of line of therapy start date, n (%)				
2009–2013	62 (29.1%)	45 (33.6%)	12 (23.1%)	5 (18.5%)
2014–2017	94 (44.1%)	53 (39.6%)	27 (51.9%)	14 (51.9%)
2018–2021	57 (26.8%)	36 (26.9%)	13 (25.0%)	8 (29.6%)
Agents used in each included line of therapy, n (%)[1]				
TKI therapy	120 (56.3%)	85 (63.4%)	24 (46.2%)	11 (40.7%)
Cytoreductive therapy	82 (38.5%)	40 (29.9%)	27 (51.9%)	15 (55.6%)
Biologic therapy	22 (10.3%)	14 (10.4%)	6 (11.5%)	2 (7.4%)
Agent-level information available[2]	**N = 188**	**N = 122**	**N = 42**	**N = 24**
TKI				
Midostaurin	99 (52.7%)	69 (56.6%)	21 (50.0%)	9 (37.5%)
Ripretinib	4 (2.1%)	3 (2.5%)	0 (0.0%)	1 (4.2%)
Ibrutinib	3 (1.6%)	3 (2.5%)	0 (0.0%)	0 (0.0%)
Dasatinib	2 (1.1%)	2 (1.6%)	0 (0.0%)	0 (0.0%)
Imatinib	2 (1.1%)	1 (0.8%)	0 (0.0%)	1 (4.2%)
Cytoreductive therapy				
Cladribine	45 (23.9%)	23 (18.9%)	13 (31.0%)	9 (37.5%)
Hydroxyurea	14 (7.4%)	8 (6.6%)	4 (9.5%)	2 (8.3%)
Azacitidine	2 (1.1%)	0 (0.0%)	1 (2.4%)	1 (4.2%)
Decitabine	2 (1.1%)	2 (1.6%)	0 (0.0%)	0 (0.0%)
Biologic therapy				
Interferon-alfa	10 (5.3%)	8 (6.6%)	2 (4.8%)	0 (0.0%)
Pegylated interferon	6 (3.2%)	3 (2.5%)	3 (7.1%)	0 (0.0%)
Brentuximab vedotin	4 (2.1%)	3 (2.5%)	1 (2.4%)	0 (0.0%)
Gemtuzumab ozogamicin	1 (0.5%)	0 (0.0%)	0 (0.0%)	1 (4.2%)

Abbreviations: 1L: first line of therapy; 2L: second line of therapy; 3L+: third or later line of therapy; max: maximum; min: minimum; SD: standard deviation; TKI: tyrosine kinase inhibitor.

Notes:

[1] The sum of individual agent class counts across each group (i.e., overall, 1L, 2L, 3L+) may differ from the number of lines of therapy in each corresponding group because some patients were treated with multiple agents in each line of therapy.

[2] Agent-level information for prior treatments was reported among patients from all study sites except Medical University of Vienna (Austria) (N=25 lines of therapy), where only treatment class information was collected per local regulations.

Figure 1. KM curve for DOT among AdvSM patients treated with avapritinib (≤200mg) versus BAT

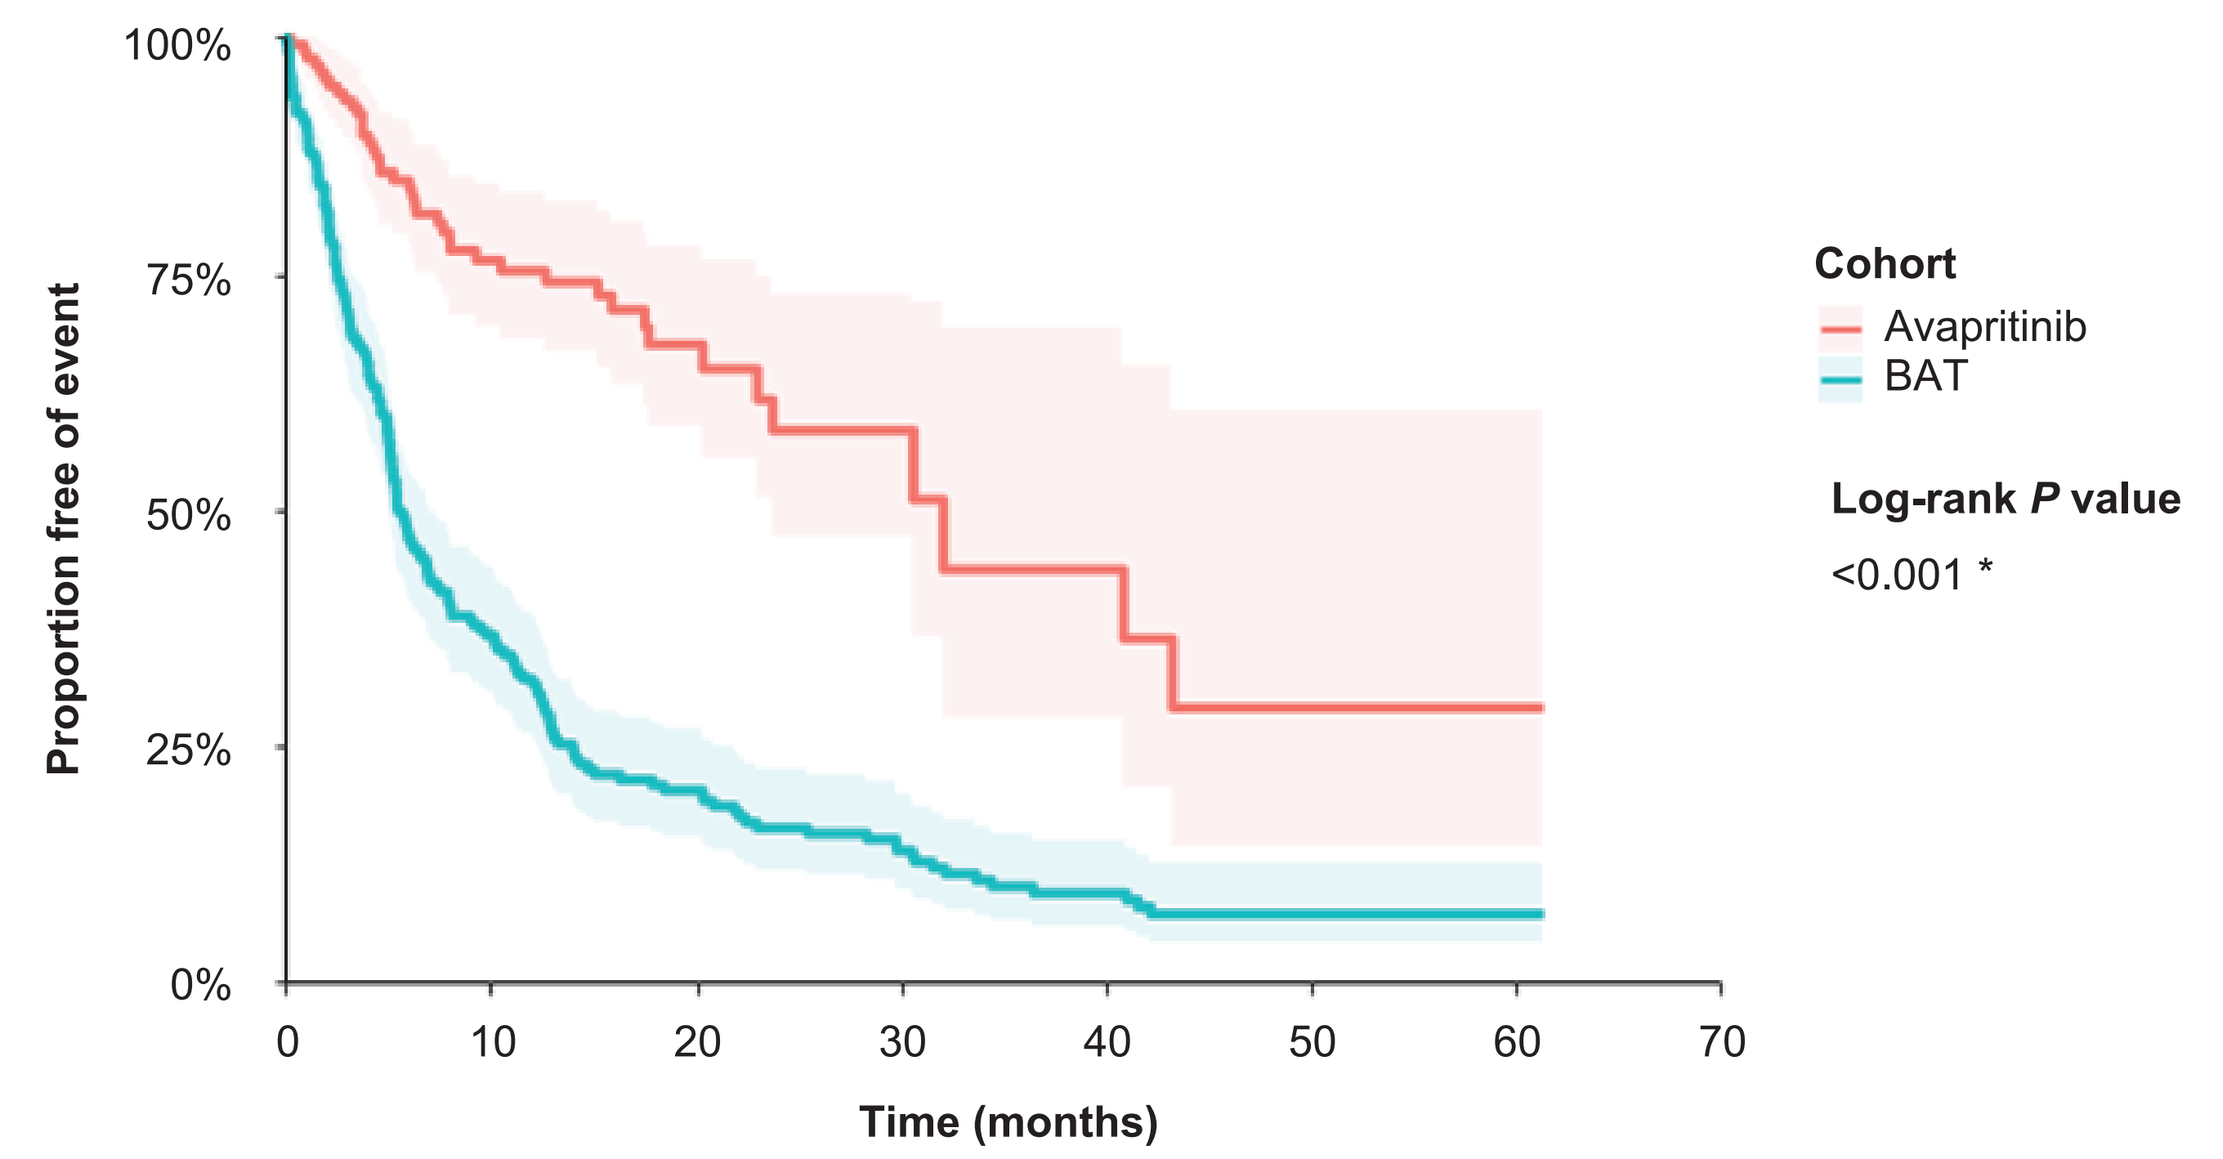
100%
75%
50%
25%
0%
Proportion free of event
0
10
20
30
40
50
60
70
Time (months)
Cohort
Avapritinib
BAT
Log-rank *P* value
<0.001 *

Number at risk	0	10	20	30	40	50	60	70
Avapritinib	136	70	28	8	6	4	1	0
BAT	213	71	36	23	13	10	9	0

**P<0.05*

Abbreviations: AdvSM: advanced systemic mastocytosis; BAT: best available therapy.

Note: The follow-up time for the BAT cohort was truncated to match the maximum follow-up time of the avapritinib cohort. In the BAT cohort, 137 patients contributed 213 lines of therapy to the analysis.

Table 4. Duration of treatment among subgroups of patients with AdvSM treated with avapritinib versus BAT

	Avapritinib	BAT
Avapritinib (≤200mg) vs BAT, all lines of therapy		
Number of unique patients	136	137
Number of lines of therapy	136	213
Median DOT, months (95% CI)	32.1 (23.8, NE)	5.5 (5.1, 7.0)
Adjusted HR (95% CI)[1]	0.35 (0.23, 0.51); *P*<0.001*	
Avapritinib (≤200mg) vs BAT, 2L+		
Number of unique patients	85	67
Number of lines of therapy	85	97
Median DOT, months (95% CI)	NR (43.3, NE)	5.2 (3.1, 8.1)
Adjusted HR (95% CI)	0.34 (0.21, 0.56); *P*<0.001*	
Avapritinib (200mg) vs BAT, 2L+		
Number of unique patients	79	67
Number of lines of therapy	79	97
Median DOT, months (95% CI)	43.3 (17.7, NE)	5.2 (3.1, 8.1)
Adjusted HR (95% CI)	0.36 (0.22, 0.57); *P*<0.001*	

**P<0.05*

Abbreviations: 2L+: second or later lines of therapy; AdvSM: advanced systemic mastocytosis; BAT: best available therapy; CI: confidence interval; DOT: duration of treatment; HR: hazard ratio.

[1] Stabilized IPTW weights were generated using the following baseline characteristics: age, sex, region, ECOG score, anemia (hemoglobin less than 10 g/dL), thrombocytopenia (platelet count less than 100 x 109/L), AdvSM subtype, skin involvement, leukocyte count of 16 × 109 per L or higher, serum tryptase level of 125 ng/mL or higher, number of mutated genes within the SRSF2/ASXL1/RUNX1 gene panel, number of prior lines of therapy, and prior use of tyrosine kinase inhibitor, cytoreductive, biologic or other systemic therapy.

Figure 2. Maximum percentage reduction in serum tryptase levels among subgroups of patients with AdvSM treated with avapritinib ≤200mg versus BAT

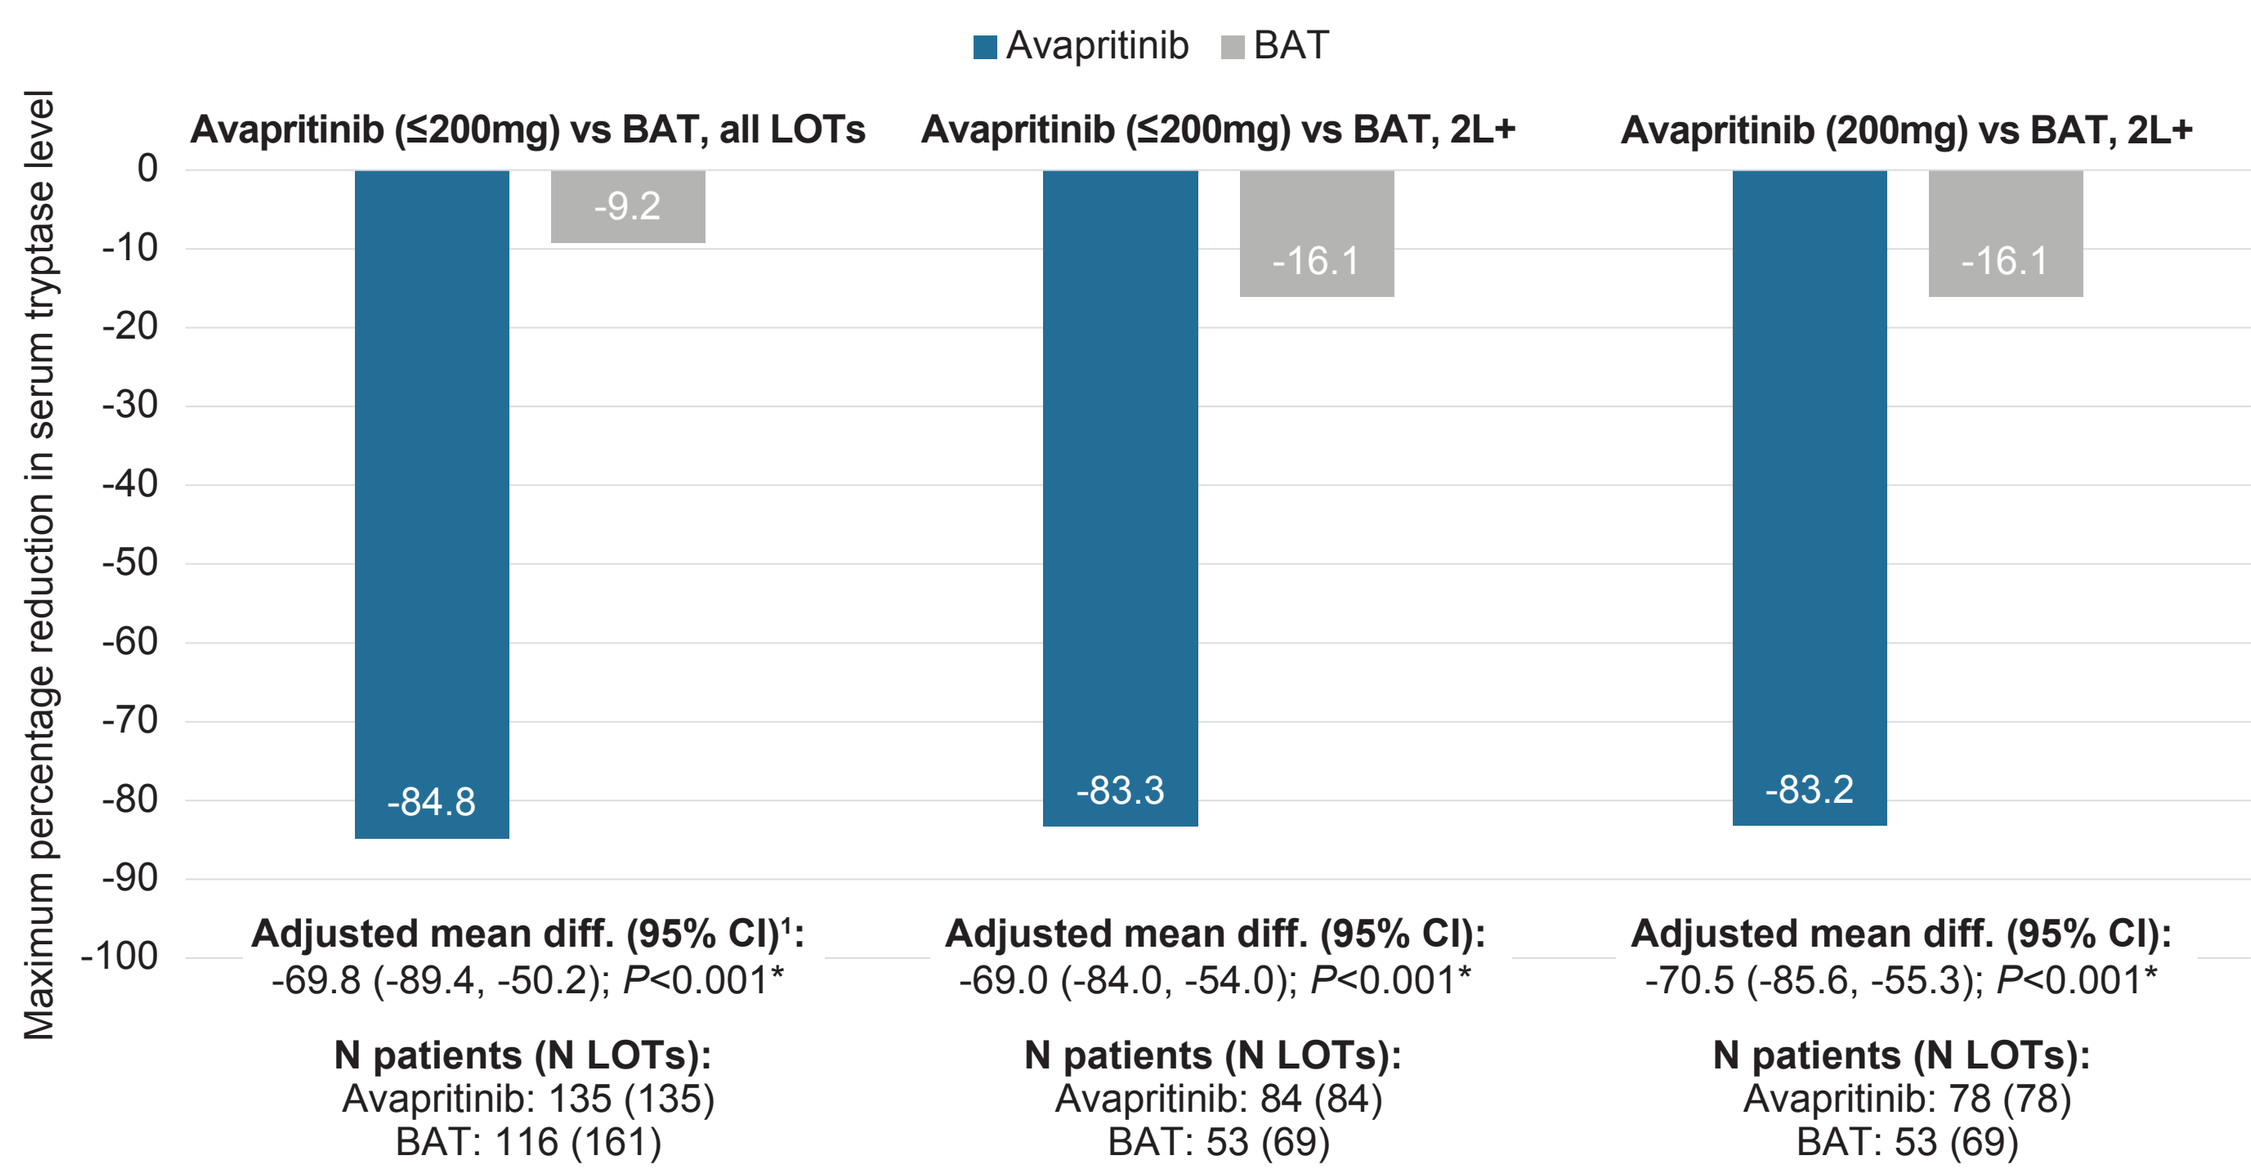
Avapritinib
BAT
Maximum percentage reduction in serum tryptase level
0
-10
-20
-30
-40
-50
-60
-70
-80
-90
-100
Avapritinib (≤200mg) vs BAT, all LOTs
-84.8
-9.2
Adjusted mean diff. (95% CI)[1]:
-69.8 (-89.4, -50.2); *P*<0.001*
N patients (N LOTs):
Avapritinib: 135 (135)
BAT: 116 (161)
Avapritinib (≤200mg) vs BAT, 2L+
-83.3
-16.1
Adjusted mean diff. (95% CI):
-69.0 (-84.0, -54.0); *P*<0.001*
N patients (N LOTs):
Avapritinib: 84 (84)
BAT: 53 (69)
Avapritinib (200mg) vs BAT, 2L+
-83.2
-16.1
Adjusted mean diff. (95% CI):
-70.5 (-85.6, -55.3); *P*<0.001*
N patients (N LOTs):
Avapritinib: 78 (78)
BAT: 53 (69)

**P<0.05*

Abbreviations: 2L+: second or later lines of therapy; AdvSM: advanced systemic mastocytosis; BAT: best available therapy; CI: confidence interval; max: maximum; SD: standard deviation.

[1] Stabilized weights were generated using the following baseline characteristics: age, sex, region, ECOG score, anemia (hemoglobin less than 10 g/dL), thrombocytopenia (platelet count less than 100 x 10^9/L), AdvSM subtype, skin involvement, leukocyte count of 16 × 10^9 per L or higher, serum tryptase level of 125 ng/mL or higher, number of mutated genes within the SRSF2/ASXL1/RUNX1 gene panel, number of prior LOTs, and prior use of tyrosine kinase inhibitor therapy.

Maximum reduction in serum tryptase levels

- The analysis of maximum reduction in serum tryptase included 135 avapritinib patients and 116 BAT patients, the latter contributing data on 161 lines of therapy (**Figure 2**)
- The unadjusted maximum percentage reduction in serum tryptase levels was -84.8% (standard deviation [SD]: 19.9%) in the avapritinib cohort versus -9.2% (SD: 161.4%) in the BAT cohort
- After accounting for differences in baseline key covariates using IPTW, the adjusted mean difference in maximum percentage reduction in serum tryptase comparing avapritinib to BAT was -69.8% (95% CI: -89.4%, -50.2%; *P*<0.001)
 - This indicates an approximately 70% greater maximum reduction in serum tryptase levels, within a line of therapy, for patients in the avapritinib cohort compared to patients in the BAT cohort
- Results were similar in subgroups (**Figure 2**)
 - Among patients treated with ≤200mg avapritinib in the 2L+ setting (i.e., pre-treated patients), the adjusted mean difference in maximum percentage reduction in serum tryptase comparing avapritinib to BAT was -69.0% (95% CI: -84.0, -54.0%; *P*<0.001)

Limitations

- AdvSM diagnoses for BAT cohort patients were based on local clinician-assessed evaluation using the 2016 revision to the WHO diagnostic criteria; thus, it is possible an incorrect subtype diagnosis was made prior to the substantial increases in disease knowledge that have occurred over the last decade. Diagnoses for the avapritinib cohort patients were confirmed using central adjudication.
 - The impact of this limitation is mitigated since all sites that contributed patients to the BAT cohort are Centers of Excellence in the treatment of AdvSM
- Since data collection for BAT cohort patients was conducted retrospectively, results may have been impacted by incomplete reporting for key prognostic characteristics, such as performance status
 - A sensitivity analysis assessing the impact of missing performance status indicated that this is not expected to impact results

Conclusions

- The results of this observational, retrospective, study indicate that patients with AdvSM treated with avapritinib in clinical trials experienced significantly longer DOT and greater reductions in serum tryptase levels compared to patients treated with BAT in real-world settings
- As no RCTs have been conducted, these data offer meaningful insights into the treatment benefit of avapritinib versus other therapies for AdvSM

Acknowledgements

This study was funded by Blueprint Medicines Corporation.

References

1. Valent P. Mastocytosis: A paradigmatic example of a rare disease with complex biology and pathology. *Am J Cancer Res.* 2013;3(2):159-172.
2. Ustun C, Arock M, Kluin-Nelemans HC, et al. Advanced systemic mastocytosis: From molecular and genetic progress to clinical practice. *Haematologica.* 2016;101(10):1133-1143.
3. Swerdlow S, Campo E, Harris N, et al. *WHO classification of tumours of haematopoietic and lymphoid tissues (revised 4th edition).* Lyon, France: IARC; 2017.
4. Lim K-H, Tefferi A, Lasho TL, et al. Systemic mastocytosis in 342 consecutive adults: survival studies and prognostic factors. *Blood.* 2009;113(23):5727-5736.
5. Singh A, Al-Kali A, Begna KH, et al. Midostaurin therapy for advanced systemic mastocytosis: Mayo Clinic experience in 33 consecutive cases. *Am J Hematol.* 2022;97(5):630-637.
6. Szudy-Szczyrek A, Bachanek-Mitura O, Gromek T, et al. Real-World Efficacy of Midostaurin in Aggressive Systemic Mastocytosis. *J Clin Med.* 2021;10(5).
7. Sperr WR, Kundi M, Alvarez-Twose I, et al. International prognostic scoring system for mastocytosis (IPSM): a retrospective cohort study. *Lancet Haematol.* 2019;6(12):e638-e649.
8. U.S. Food and Drug Administration. FDA approves avapritinib for advanced systemic mastocytosis. In:2021. https://www.fda.gov/drugs/resources-information-approved-drugs/fda-approves-avapritinib-advanced-systemic-mastocytosis
9. Blueprint Medicines Corporation. Blueprint Medicines' AYVAKYT® (avapritinib) Receives European Commission Approval for the Treatment of Adults with Advanced Systemic Mastocytosis. In:2022. https://ir.blueprintmedicines.com/news-releases/news-release-details/blueprint-medicines-ayvakytr-avapritinib-receives-european?utm_source=LinkedIn&utm_medium=social&utm_content=Image&utm_campaign=Corporate_data_milestones
10. DeAngelo DJ, Radia DH, George TI, et al. Safety and efficacy of avapritinib in advanced systemic mastocytosis: The phase 1 EXPLORER trial. *Nat Med.* 2021;27:2183-2191.
11. Gotlib J, Reiter A, Radia DH, et al. Efficacy and safety of avapritinib in advanced systemic mastocytosis: Interim analysis of the phase 2 PATHFINDER trial. *Nat Med.* 2021;27:2192-2199.
12. Medicine USNLo. Retrospective study assessing the effect of avapritinib versus best available therapy in patients with AdvSM. In:2021. https://www.clinicaltrials.gov/ct2/show/NCT04695431?term=NCT04695431&draw=2&rank=1

Presented at **EHA 2022 Hybrid Congress**, June 9-12, 2022. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute