Activity and Tolerability of BLU-667, a Highly Potent and Selective RET Inhibitor, in Patients with Advanced RET-altered Thyroid Cancers

6018

Matthew H. Taylor[1], Justin F. Gainor[2], Mimi I-Nan Hu[3], Viola Weijia Zhu[4], Gilberto Lopes[5], Sophie Leboulleux[6], Marcia S. Brose[7], Martin H. Schuler[8], Daniel W. Bowles[9], Dong-Wan Kim[10], Christina S. Baik[11], Elena Garralda[12], Chia-Chi Lin[13], Douglas Adkins[14], Debashis Sarker[15], Giuseppe Curigliano[16], Hui Zhang[17], Corinne Clifford[17], Michael R. Palmer[17], Christopher D. Turner[17], Vivek Subbiah[3]

[1]Oregon Health & Science University, Portland, OR; [2]Massachusetts General Hospital, Boston, MA; [3]The University of Texas MD Anderson Cancer Center, Houston, TX; [4]Chao Family Comprehensive Cancer Center, University of California Irvine School of Medicine, Orange, CA; [5]Sylvester Comprehensive Cancer Center, University of Miami Health System, Miami, FL; [6]Institut Gustave Roussy, Villejuif, France; [7]Department of Otorhinolaryngology: Head and Neck Surgery, Abramson Cancer Center of the University of Pennsylvania, Philadelphia, PA; [8]West German Cancer Center, University Hospital Essen, Essen, Germany; [9]University of Colorado, Aurora, CO; [10]Seoul National University Hospital, Seoul, Korea, Republic of (South); [11]Fred Hutchinson Cancer Research Center, Seattle, WA; [12]Hospital Universitari Vall d'Hebron, Vall d'Hebron Institute of Oncology (VHIO), Barcelona, Spain; [13]Department of Oncology, National Taiwan University Hospital, Taipei, Taiwan; [14]Washington University School of Medicine, St. Louis, MO; [15]King's College Hospital, Institute of Liver Studies, London, United Kingdom; [16]University of Milano, European Institute of Oncology, Division of Early Drug Development, Milan, Italy; [17]Blueprint Medicines Inc, Cambridge, MA

BACKGROUND

- RET alterations are targetable oncogenic drivers in multiple tumor types, including ~90% of advanced medullary thyroid cancer (MTC)[1] and ~20% of papillary thyroid cancer (PTC)[2]
- No selective RET inhibitors are approved

BLU-667: Designed to Treat RET-Altered Cancers

BLU-667 potently and selectively inhibits RET alterations, including those that confer resistance to MKI, while sparing VEGFR.[3]

BLU-667: High kinome selectivity for RET[a]

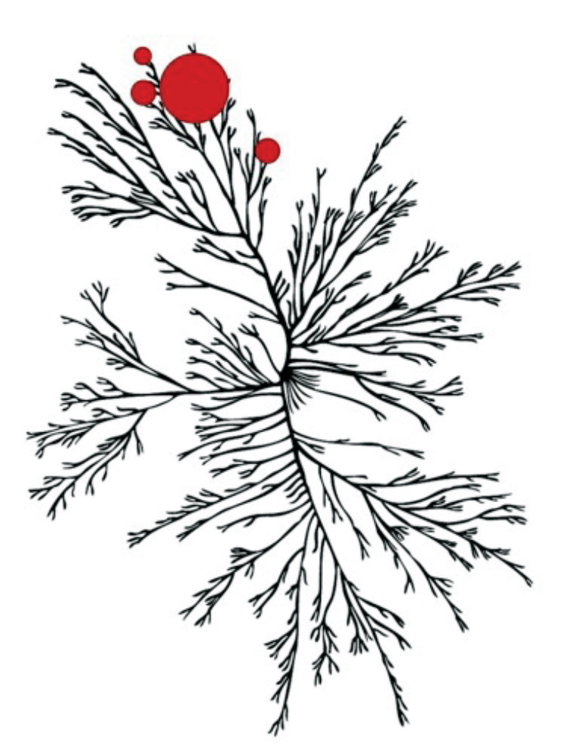

	BLU-667 IC_{50}	Cabozantinib IC_{50}	Vandetanib IC_{50}
Wild-type RET	0.4	11	4
RET V804L Gatekeeper resistance	0.3	45	3597
RET V804M Gatekeeper resistance	0.4	162	726
RET M918T Mutation	0.4	8	7
CCDC6-RET Fusion	0.4	34	20
VEGFR2 Anti-target	35	2	4

BLU-667 vs. pharmacologically relevant kinases:

- BLU-667 is ~90-fold more selective for RET than VEGFR2
- BLU-667 is 20-fold more selective for RET than JAK1

IC_{50}, half maximal inhibitory concentration; MKI, multikinase inhibitor.
[a]Kinome illustration reproduced courtesy of Cell Signaling Technology, Inc. (www.cellsignal.com) (CSTI). The foregoing website is maintained by CSTI, and Blueprint Medicines is not responsible for its content.

METHODS

ARROW: BLU-667 Dose-Escalation/Expansion Study

Part 1: Dose-Escalation (Complete; N=62)[4] → **Part 2: Expansion Cohorts (Ongoing)**

RET-altered advanced solid tumors

BLU-667 30–600 mg PO daily (QD or BID)

Phase 2 dose determined (400 mg QD)

ARROW is registered with clinicaltrials.gov (NCT03037385)

BLU-667 400 mg QD
- Unresectable, advanced solid tumor
- RET alteration status by local tumor testing
- No additional driver mutation
- ECOG PS 0–1
- Progressive disease or intolerant to SOC therapy, or not a candidate

Primary objectives:
- Overall response rate (RECIST 1.1)
- Safety

Expansion cohorts:
- MTC, prior cabozantinib or vandetanib (n=60)
- MTC, no prior cabozantinib or vandetanib (n=40)
- RET fusion+ tumors, including PTC (n=40)
- RET fusion+ NSCLC, prior platinum (n=80)
- RET fusion+ NSCLC, platinum naïve (n=40)
- Other RET-mutated tumors (n=20)
- RET-altered, prior selective RET inhibitor (n=20)

BID, twice daily dosing; ECOG PS, Eastern Cooperative Oncology Group performance status; PO, orally; QD, once daily dosing; RECIST, response evaluation criteria in solid tumors; SOC, standard of care.

Data are preliminary and based on a data cut-off date of April 28, 2019. BLU-667 is an investigational agent discovered and currently in development by Blueprint Medicines Corporation (Blueprint Medicines).

QR Code Disclaimer

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

References

1. Romei, et al. *Oncotarget*, 2018;9:9875-9884.
2. Santoro, et al. *J Clin Invest*. 1992;89(5):1517-1522.
3. Subbiah, et al. *Cancer Discov*. 2018;8(7):836-849.
4. Subbiah, et al. *Cancer Res*. 2018;78(13 Suppl):Abstract CT043.

RESULTS: ADVANCED RET-MUTATED MTC

Patient Baseline Characteristics

Characteristic	RET-mutated MTC (400 mg QD Starting Dose) All (N=64)	Prior Cabo or Vand (N=43)
Age (years), median (range)	59 (19–81)	57 (25–81)
Male, n (%)	42 (66)	27 (63)
ECOG PS, n (%)		
0	21 (33)	9 (21)
1–2	43 (66)	33 (79)
Metastatic disease, n (%)	64 (100)	43 (100)
Prior systemic regimens, median (range)	1 (0–10)	2 (1–10)
Any prior anticancer treatment	50 (78)	43 (100)
Cabozantinib or vandetanib, n (%)	43 (67)	43 (100)
Cabozantinib and vandetanib, n (%)	13 (20)	13 (30)
RET mutation, n (%)		
M918T	36 (56)	26 (61)
C634R/S/W	10 (16)	7 (16)
V804M	3 (5)	2 (5)
Other	15 (23)	8 (19)

Cabo, cabozantinib; Vand, vandetanib.

Tolerability

Among 64 patients with RET-mutated MTC receiving BLU-667 starting dose of 400 mg QD:
- Treatment-related toxicity is generally low-grade and reversible
- No patients discontinued BLU-667 due to treatment-related toxicity (4% across the entire study)

Adverse Event Term	Treatment-Emergent (≥15% overall) n (%) All	Treatment-Emergent Grade ≥3	Treatment-Related n (%) All	Treatment-Related Grade ≥3
Hypertension	26 (41)	15 (23)	19 (30)	10 (16)
Constipation	21 (33)	1 (2)	12 (19)	1 (2)
Neutropenia[a]	17 (27)	7 (11)	15 (23)	7 (11)
Anemia	14 (22)	3 (5)	6 (9)	1 (2)
Aspartate aminotransferase increased	14 (22)	-	9 (14)	-
Leukopenia[b]	14 (22)	1 (2)	11 (17)	-
Alanine aminotransferase increased	13 (20)	-	8 (13)	-
Diarrhea	13 (20)	3 (5)	6 (9)	1 (2)
Headache	12 (19)	-	5 (8)	-
Blood creatinine increased	11 (17)	-	7 (11)	-
Fatigue	11 (17)	-	6 (9)	-
Hypocalcemia	11 (17)	4 (6)	4 (6)	1 (2)

RET-mutated MTC (400 mg QD Starting Dose; N=64)

Additional grade ≥3 treatment-related AEs (≥2%): blood creatine phosphokinase increased (5%).
[a]Combined term including decreased neutrophil count. [b]Combined term including decreased white blood cell count.

Reduction of RET Mutant ctDNA, Calcitonin, and CEA

RET-mutated MTC (400 mg QD Starting Dose)

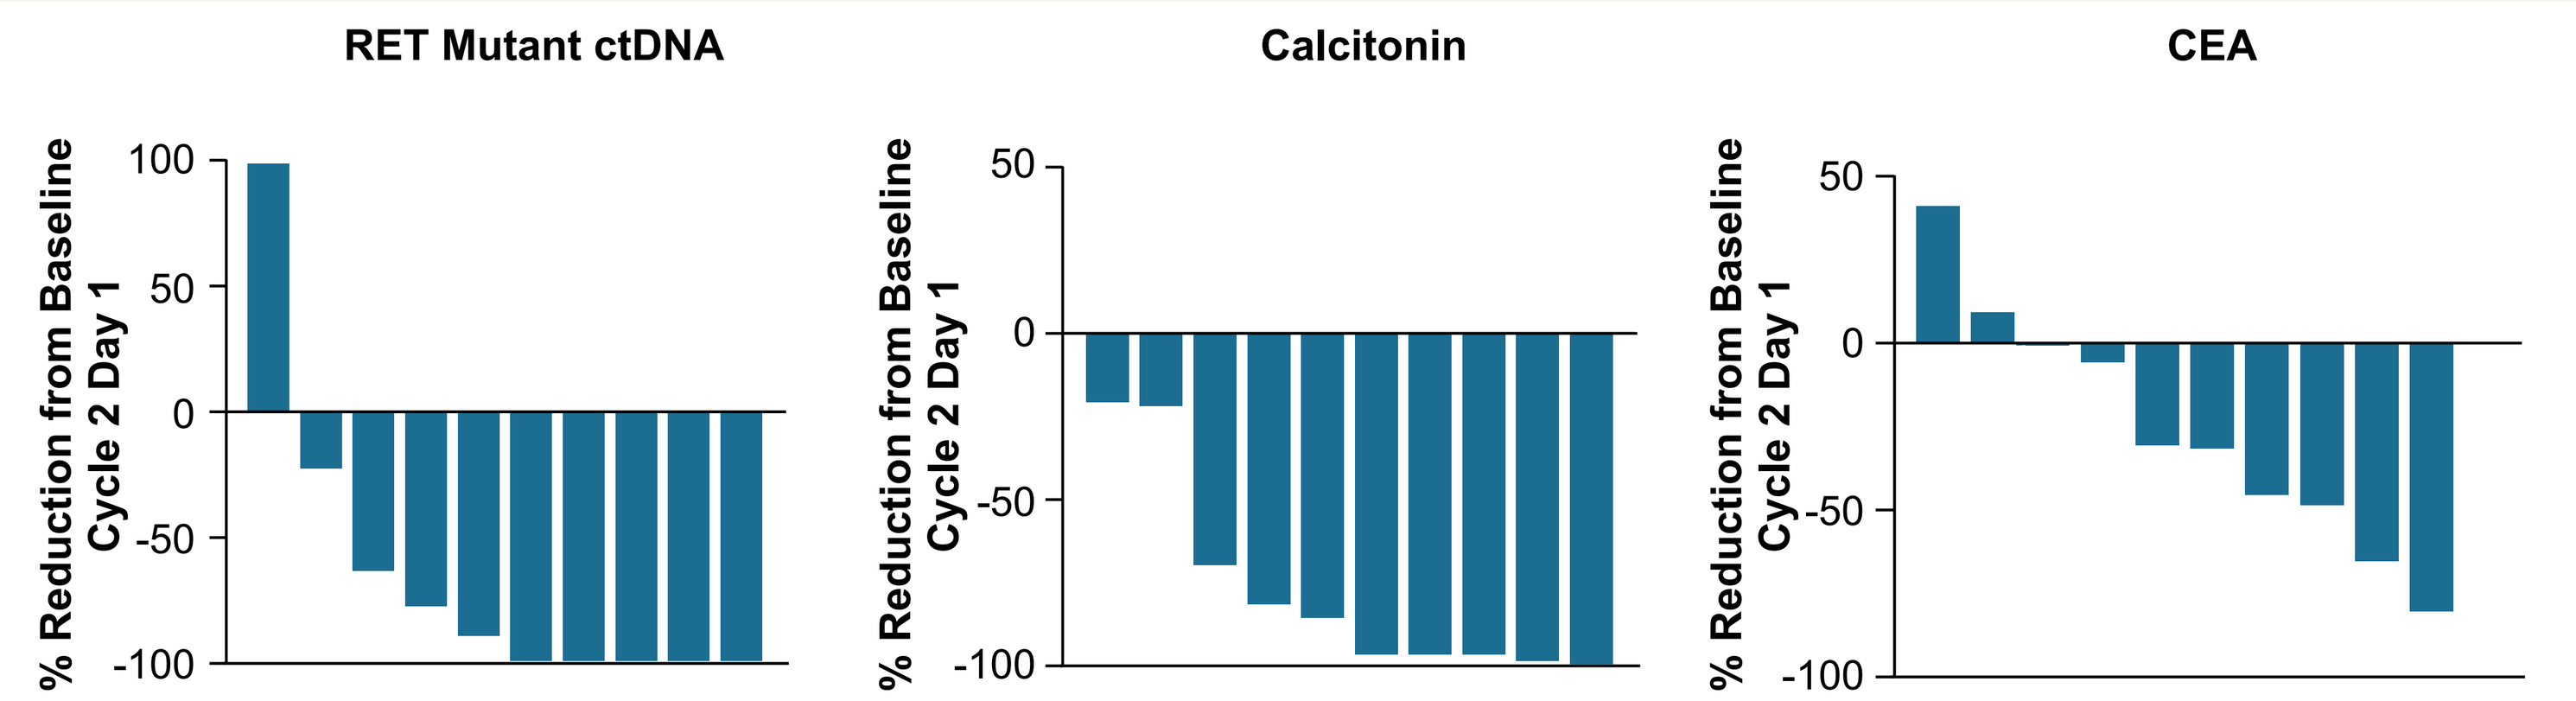
RET Mutant ctDNA
Calcitonin
CEA
% Reduction from Baseline Cycle 2 Day 1
100
50
0
-50
-100

CEA, carcinoembryonic antigen; ctDNA, circulating tumor deoxyribonucleic acid.

Antitumor Activity

Tumor Response
RET-mutated MTC (400 mg QD starting dose)[a]

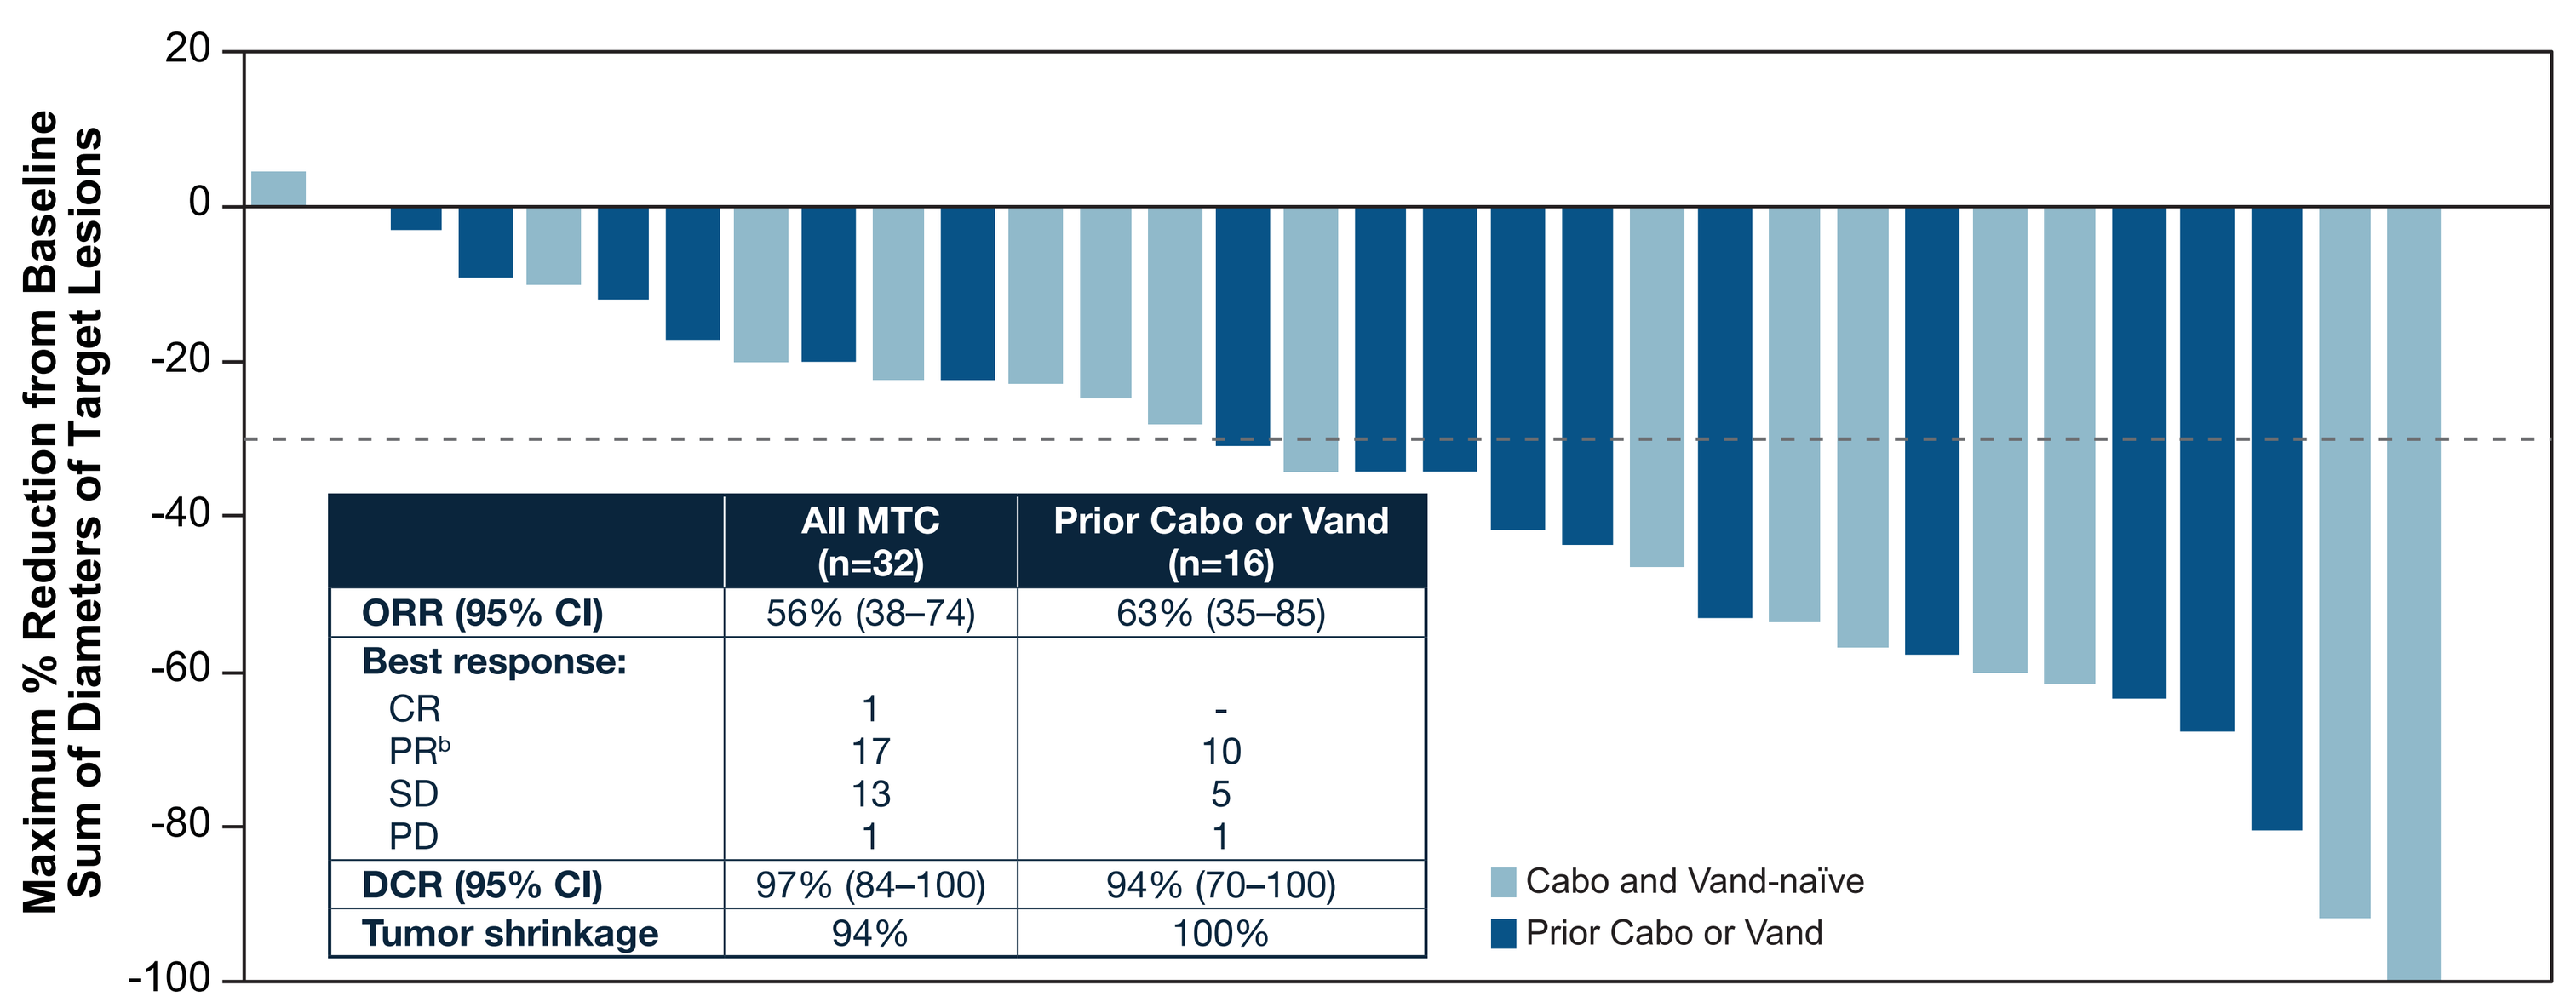
Maximum % Reduction from Baseline Sum of Diameters of Target Lesions
20
0
-20
-40
-60
-80
-100
Cabo and Vand-naïve
Prior Cabo or Vand

	All MTC (n=32)	Prior Cabo or Vand (n=16)
ORR (95% CI)	56% (38–74)	63% (35–85)
Best response:		
CR	1	-
PR[b]	17	10
SD	13	5
PD	1	1
DCR (95% CI)	97% (84–100)	94% (70–100)
Tumor shrinkage	94%	100%

DCR, disease control rate (best response of SD or better); ORR, overall response rate.
[a]Data for response-evaluable patients enrolled by 14 Nov 2018. Response-evaluable population includes patients with measurable disease and ≥1 evaluable post-treatment disease assessment. [b]Two patients (one previously received vand, one cabo/vand-naïve) are pending confirmation of response.

Treatment and Response Duration
RET-mutated MTC (400 mg QD starting dose)[a]

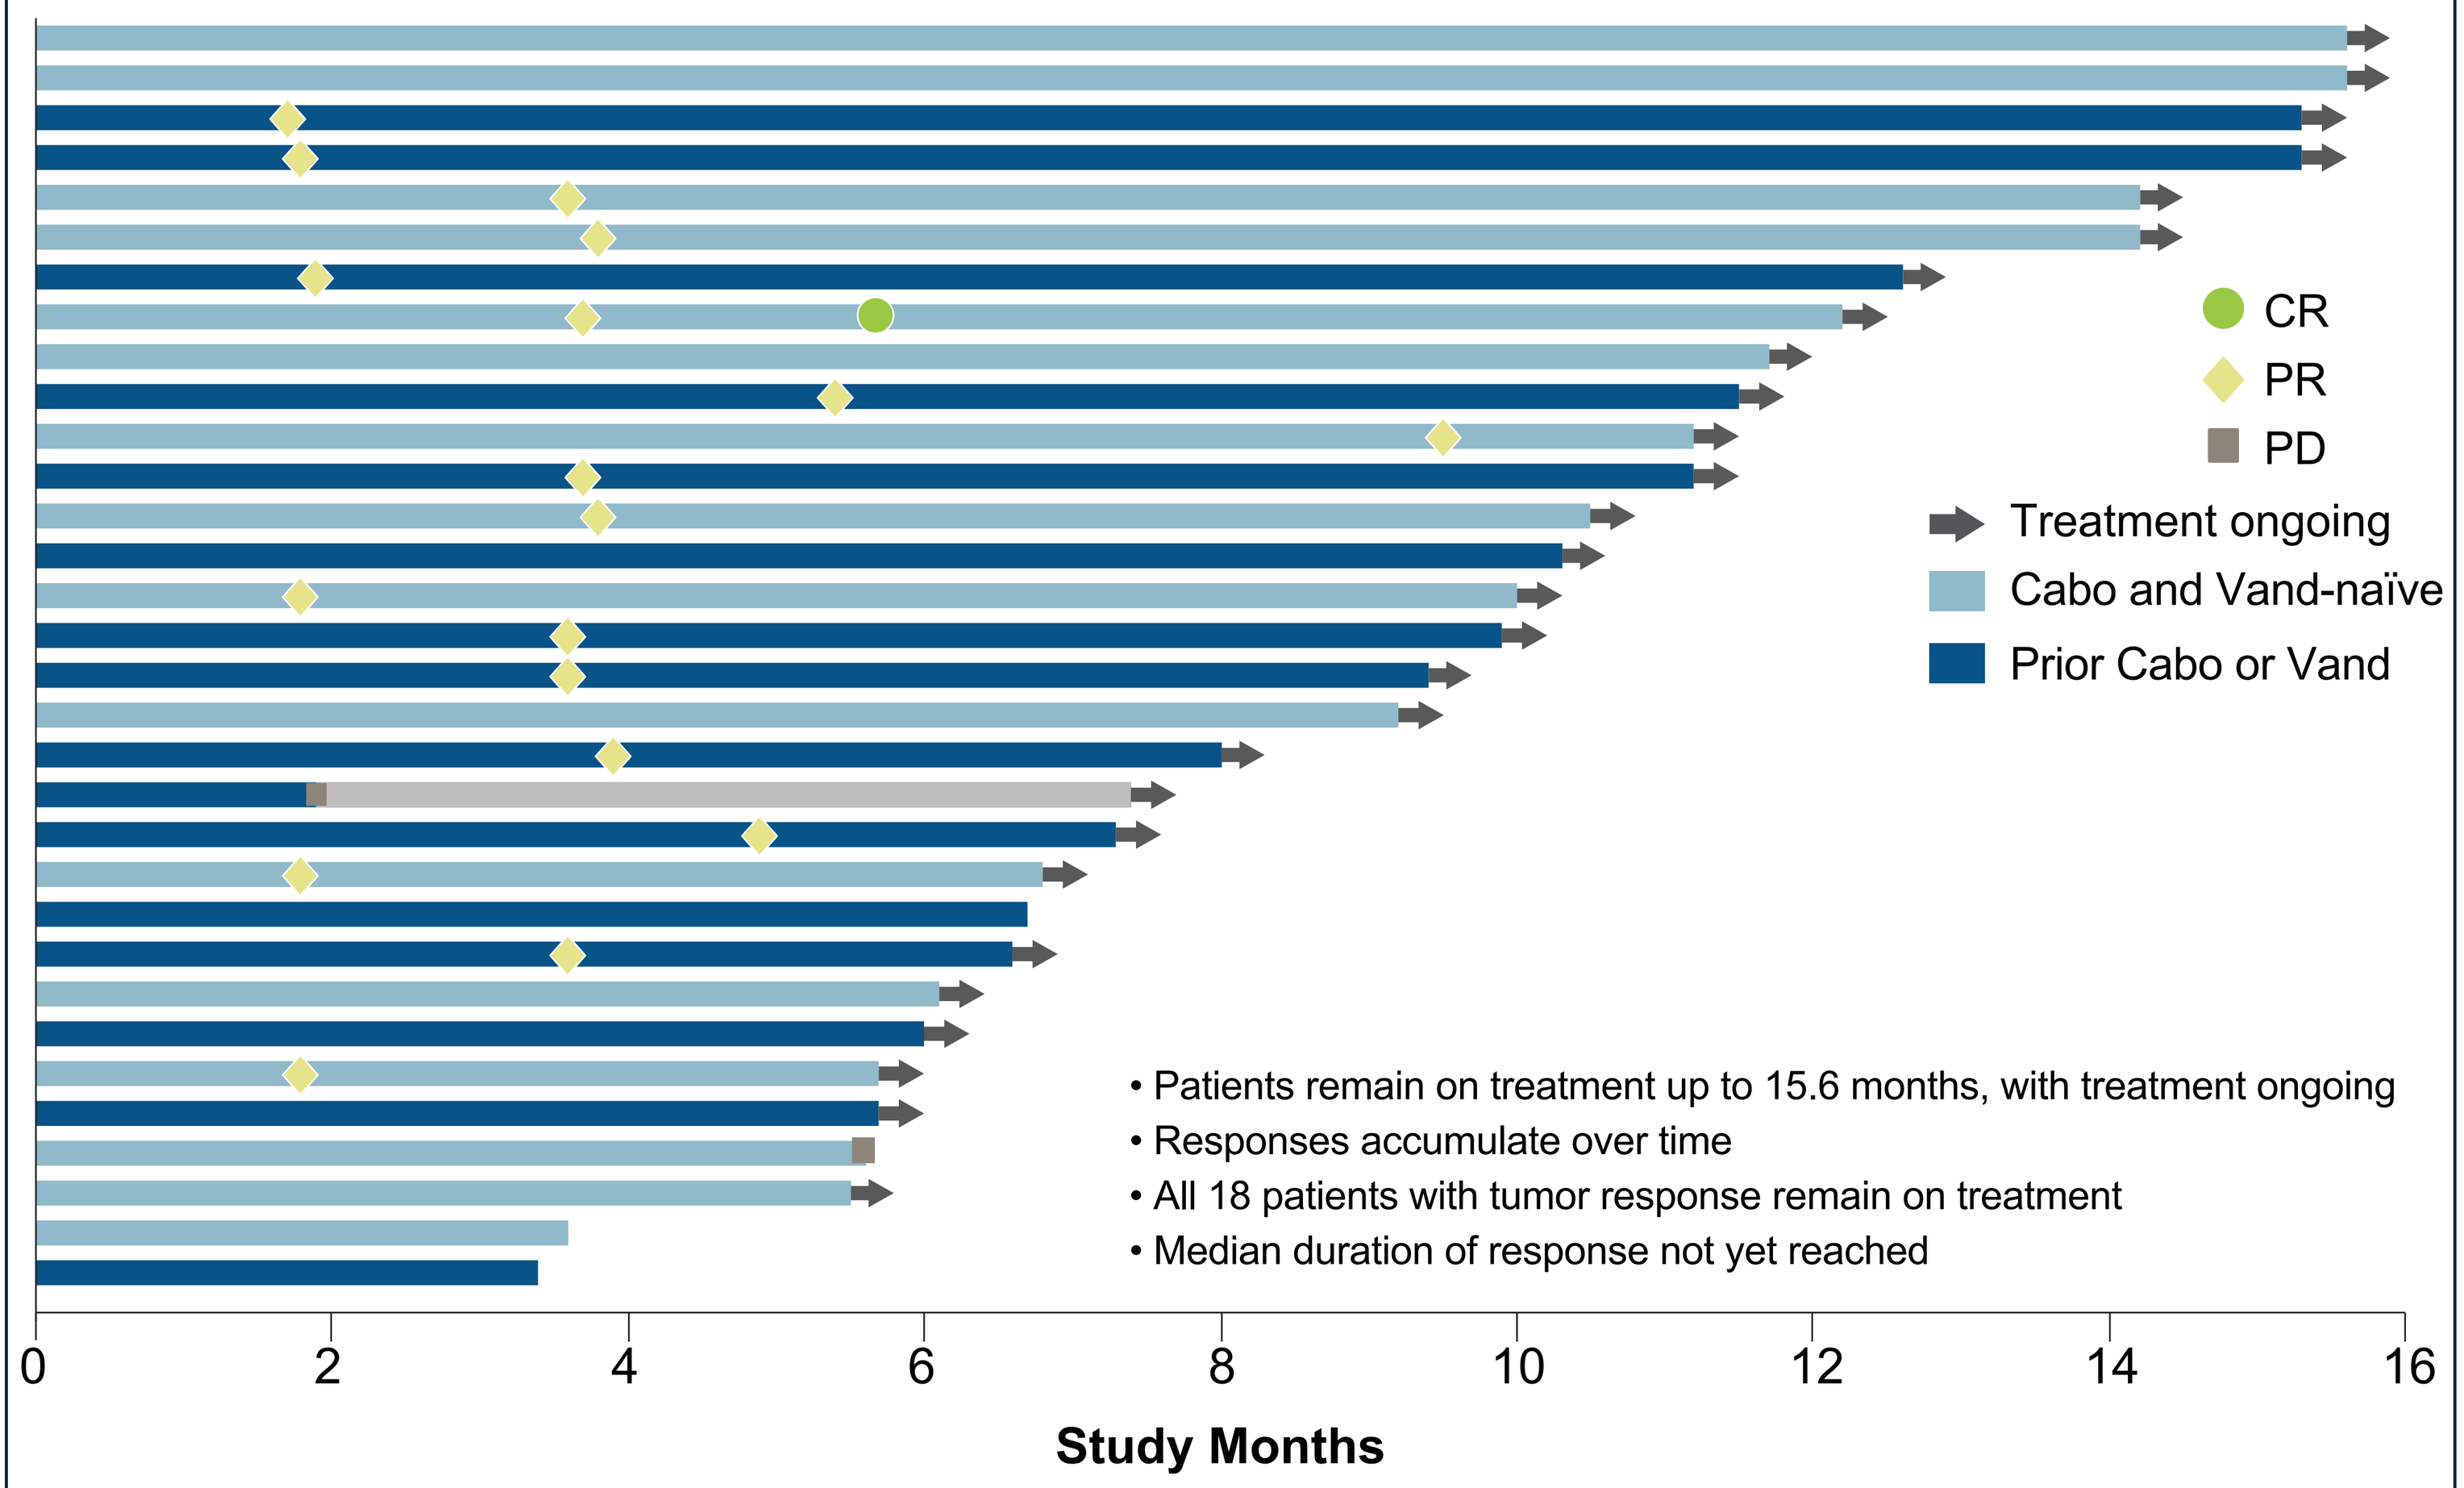
CR
PR
PD
Treatment ongoing
Cabo and Vand-naïve
Prior Cabo or Vand
0
2
4
6
8
10
12
14
16
Study Months

- Patients remain on treatment up to 15.6 months, with treatment ongoing
- Responses accumulate over time
- All 18 patients with tumor response remain on treatment
- Median duration of response not yet reached

[a]Data for response-evaluable patients enrolled by 14 Nov 2018. Response-evaluable population includes patients with measurable disease and ≥1 evaluable post-treatment disease assessment.

RESULTS: ADVANCED RET FUSION+ PTC

Patient Baseline Characteristics

Characteristic	RET fusion+ PTC (All Starting Doses; N=9[a])
Age (years), median (range)	66 (23–70)
Male, n (%)	5 (56)
ECOG PS, n (%)	
0	4 (44)
1–2	5 (56)
Metastatic disease, n (%)	9 (100)
Prior systemic regimens, median (range)	2 (0–8)
Any prior anticancer treatment	8 (89)
Sorafenib or lenvatinib, n (%)	3 (33)
Radioactive iodine, n (%)	8 (89)
RET fusion partner	
CCDC6	4 (44)
NCOA4	4 (44)
Other (SNRNP70)	1 (11)

[a]Includes 2 patients treated with starting doses of 200 mg and 300 mg QD in the dose-escalation.

Tolerability

Among 9 patients with RET fusion+ PTC (regardless of starting dose):
- Safety profile similar to MTC
- No patients discontinued BLU-667 treatment due to treatment-related toxicity

Antitumor Activity
RET fusion+ PTC (All starting doses)

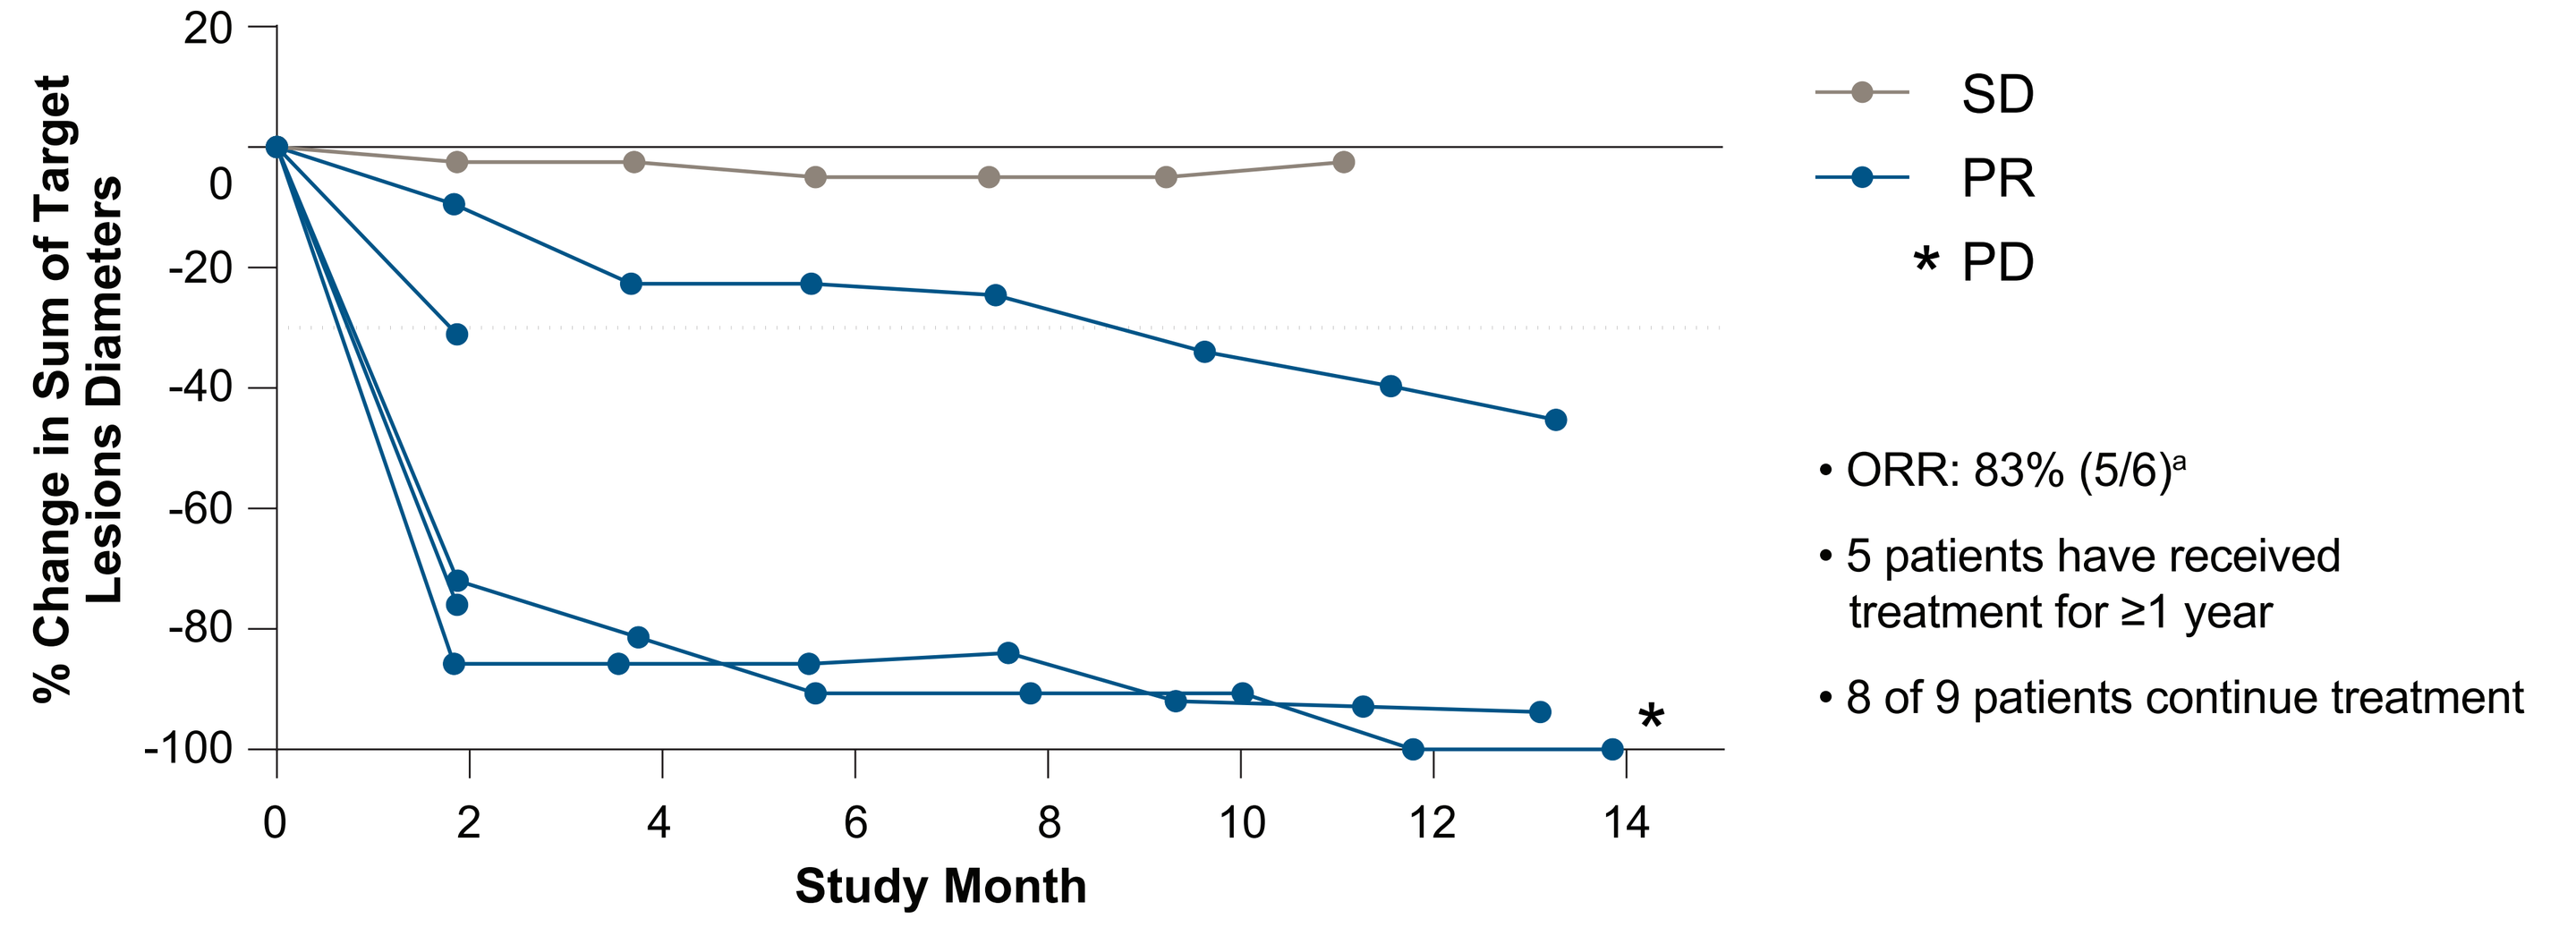
% Change in Sum of Target Lesions Diameters
20
0
-20
-40
-60
-80
-100
0
2
4
6
8
10
12
14
Study Month
SD
PR
* PD

- ORR: 83% (5/6)[a]
- 5 patients have received treatment for ≥1 year
- 8 of 9 patients continue treatment

[a]Confirmation of response is pending for two patients. Three patients are not response-evaluable due to absence of measurable disease at baseline (n=1) and pending response assessments (n=2).

CONCLUSIONS

- BLU-667 demonstrates broad and durable antitumor activity in patients with advanced, RET-altered MTC and PTC
 - 63% ORR and 94% DCR in RET-mutated MTC previously treated with cabozantinib or vandetanib; 83% ORR in PTC
 - Reponses observed regardless of treatment history or RET mutation genotype (including gatekeeper mutation V804M)
 - Well-tolerated at 400 mg QD; all responding patients with MTC remain on treatment
- Breakthrough therapy designation granted for RET-mutated MTC requiring systemic treatment and for which there are no acceptable alternative treatments
- Additional cohorts continue to assess benefit of BLU-667 in multiple other RET-mutated and RET fusion+ solid tumors

Acknowledgments

We thank the participating patients, their families, all study co-investigators, research coordinators and data managers who contributed to this study. Third-party writing assistance was provided by Ashfield Healthcare and funded by Blueprint Medicines.

Presented at the Annual Congress of the American Society of Clinical Oncology Annual Meeting, June 1, 2019, Chicago, IL